Adenosine and AMP Gene Expression Profiles Predict Response to Adenosine Pathway Therapies and Indicate a Need for Dual Blockade of CD73 and A2AR with CD73 Inhibitors

Willingham S, Hotson A, Hsieh J, Munneke B, Kwei L, Mobasher M, Buggy J, Miller R

Corvus Pharmaceuticals, Burlingame CA, USA

CORVUS PHARMACEUTICALS

ADENOSINE INHIBITS ANTI-TUMOR IMMUNITY

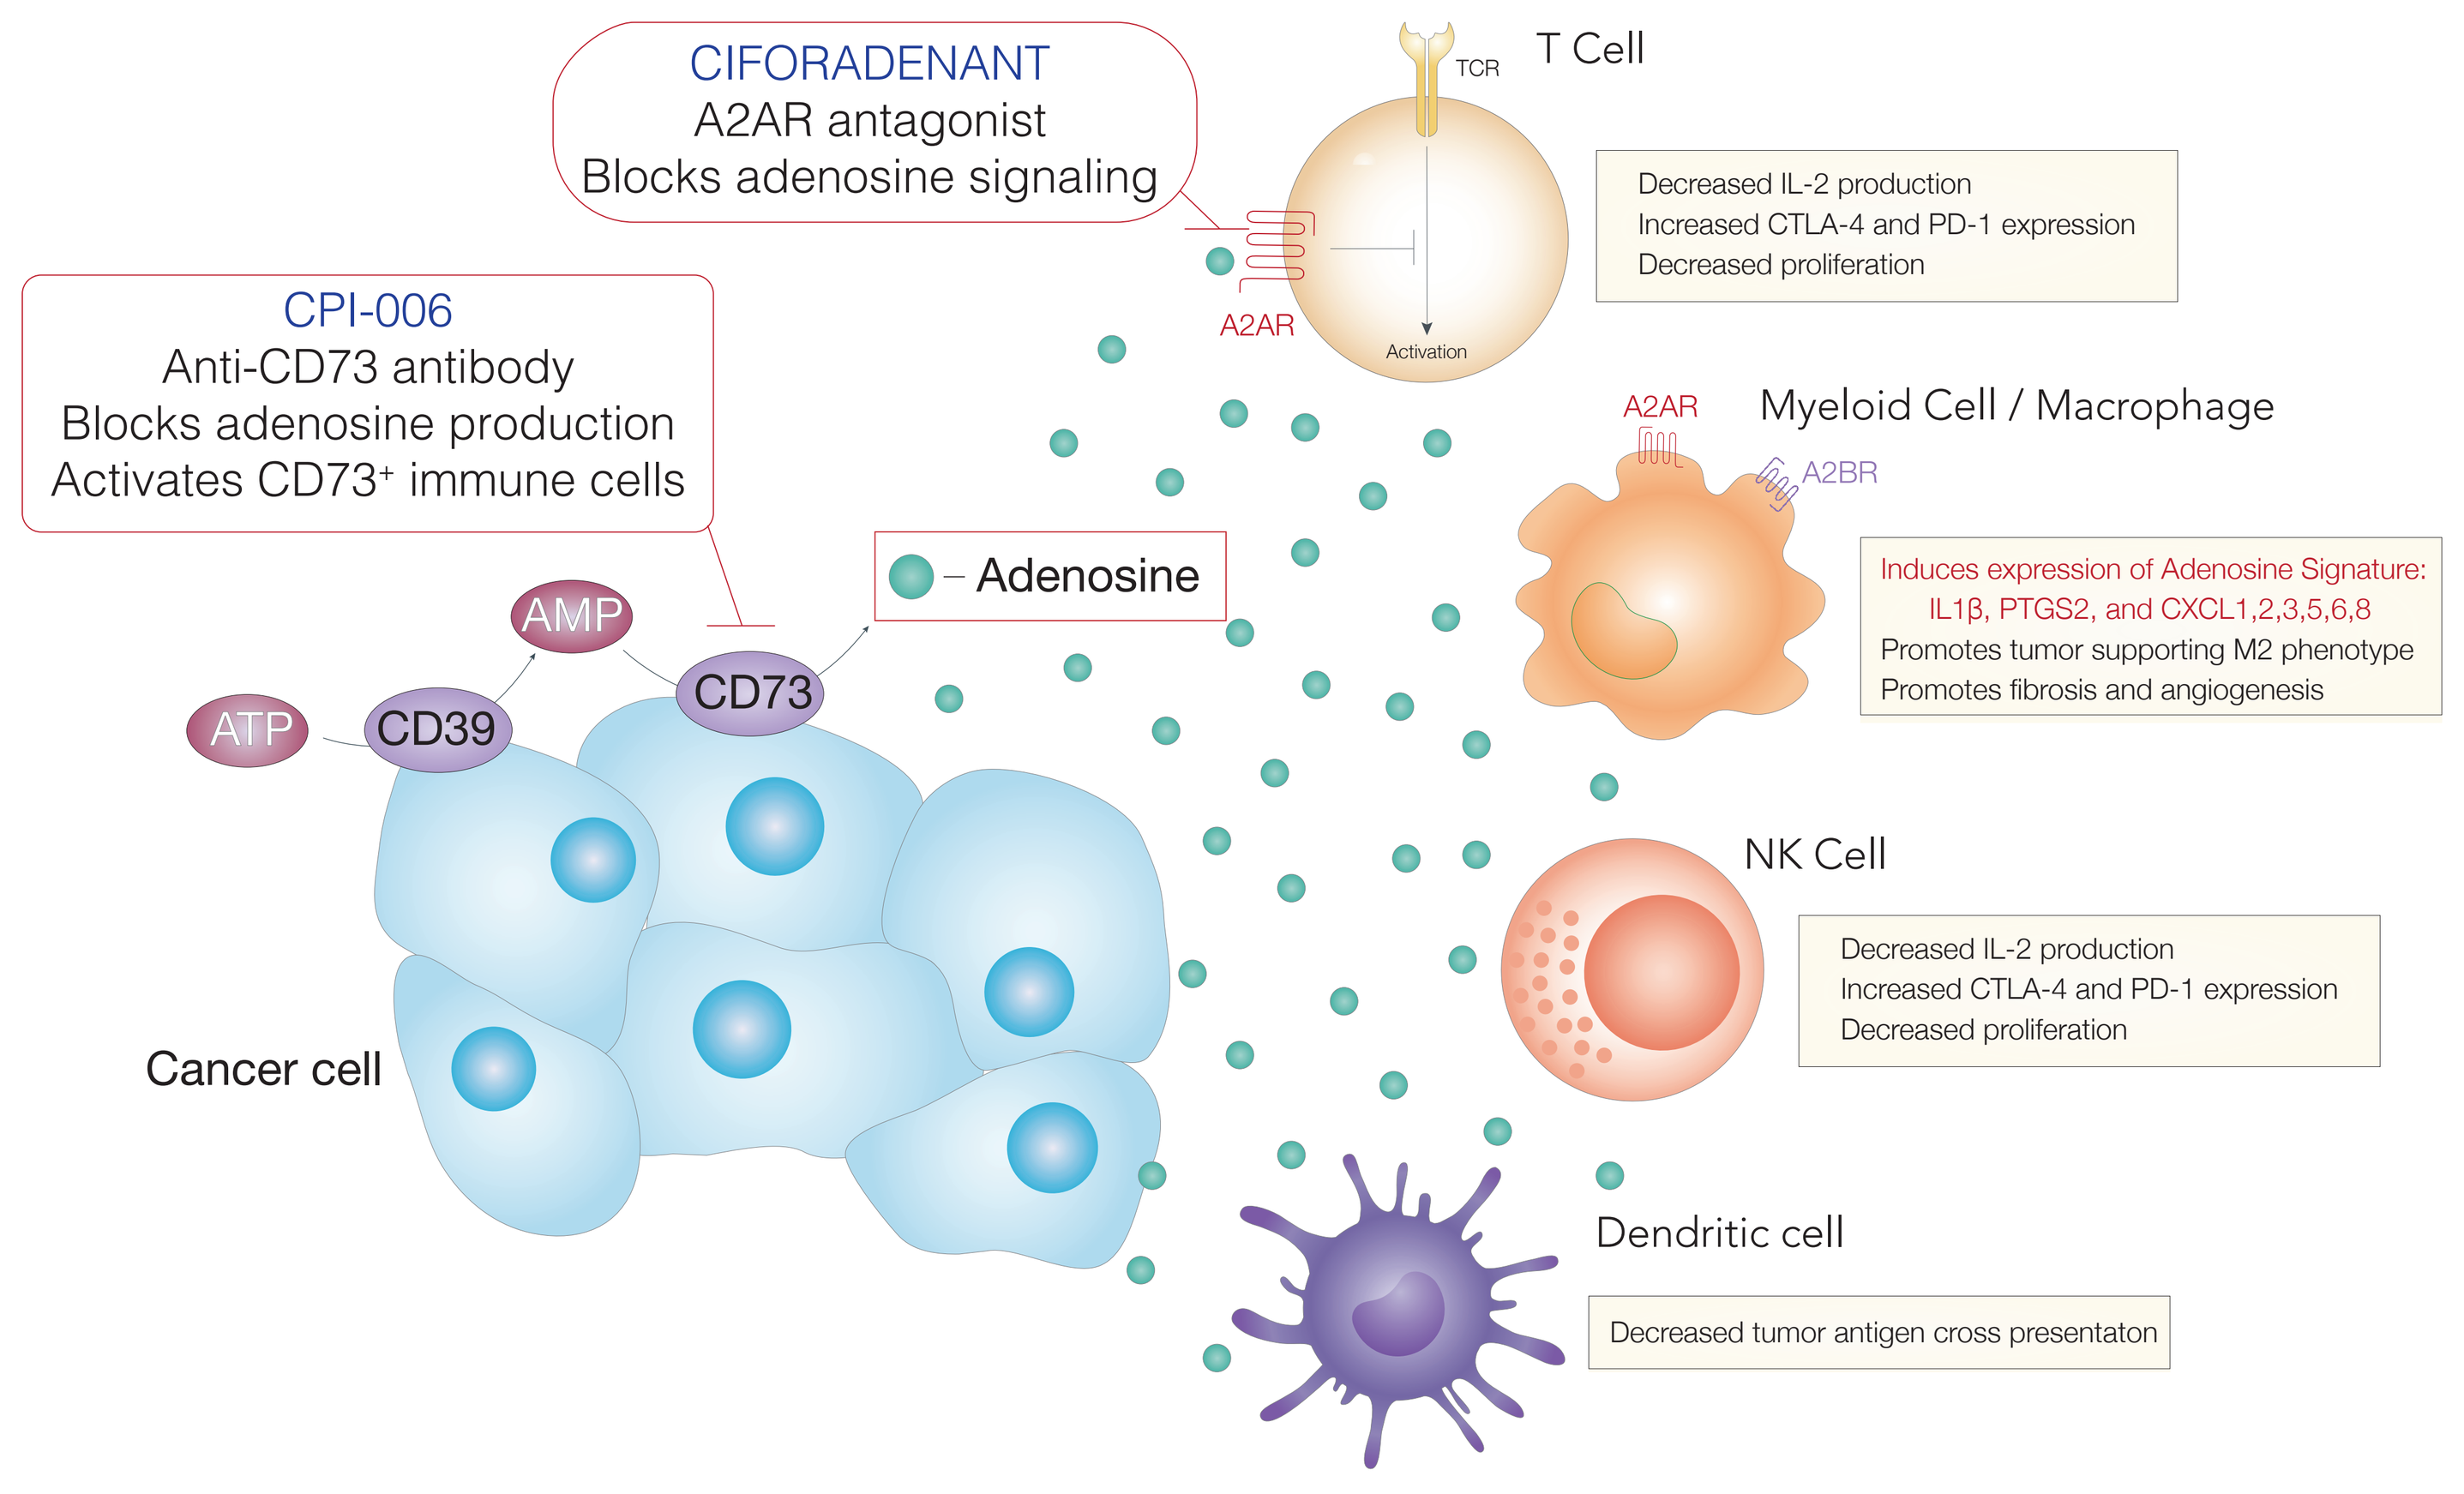
CIFORADENANT
A2AR antagonist
Blocks adenosine signaling
CPI-006
Anti-CD73 antibody
Blocks adenosine production
Activates CD73+ immune cells
T Cell
TCR
A2AR
Activation
Decreased IL-2 production
Increased CTLA-4 and PD-1 expression
Decreased proliferation
Adenosine
A2AR
A2BR
Myeloid Cell / Macrophage
Induces expression of Adenosine Signature:
IL1β, PTGS2, and CXCL1,2,3,5,6,8
Promotes tumor supporting M2 phenotype
Promotes fibrosis and angiogenesis
AMP
ATP
CD39
CD73
Cancer cell
NK Cell
Decreased IL-2 production
Increased CTLA-4 and PD-1 expression
Decreased proliferation
Dendritic cell
Decreased tumor antigen cross presentaton

- Extracellular adenosine has a short half life and it is not feasible to routinely measure in human tumors
- Determine genes/proteins modulated by adenosine as a surrogate to identify patients with adenosine rich tumors

ADENOSINE INDUCES A SPECIFIC GENE SIGNATURE

HUMAN PBMCs STIMULATED WITH NECA, A STABLE ADENOSINE ANALOG

Experiment Setup — Human PBMC — + NECA — 1 hour — + ACTIVATION anti-CD3/CD28 — 48 hours — NanoString

ADENOSINE SIGNATURE - NANOSTRING

	Adjusted p Value	Function
IL23A	1.44E-04	Increases angiogenesis and reduces CD8+ T-cell infiltration
SLC11A1	1.27E-03	Natural resistance-associated macrophage protein 1
CXCL2	1.27E-03	MIP2a: macrophage inflammatory protein 2, alpha
CXCL7	1.27E-03	PPBP. Pro-Platelet Basic Protein
CXCL6	1.40E-03	GCP2: Granulocyte chemotactic protein 2
CXCL3	1.40E-03	Controls migration and adhesion of monocytes
IL-6	1.48E-03	Pro- and anti-inflammatory cytokine
IL-1α	1.73E-03	Inflammation
CXCL8	1.98E-03	IL-8. Nutrophil chemotactic factor
CXCL5	1.98E-03	Attracts and activates neutrophils
THBS1	2.28E-03	Multiple functions. Inhibits angiogenesis & immune regulation
IL-1β	2.38E-03	Inflammation
PTGS2	2.65E-03	COX-2. Elevated during inflammation and cancer
IL-24	2.70E-03	Cell survival and proliferation. Activates STAT1/3
CXCL1	3.92E-03	Neutrophil chemotractant
CD86	5.31E-03	B7-2: Costimulatory signal for T cell activation and survival
CLEC5A	5.82E-03	Interacts with DAP-12 and may play a role in cell activation
CD14	6.24E-03	Expressed by myeloid cells

Human PBMCS were isolated from buffy coat samples by density centrifugation with Histopaque 1077 (400*g, 30 min). Cells were washed and resuspended at a density $2*10^6$ cells/ml in RPMI + 10% human serum. PBMCs were stimulated with DMSO or 5'-N-Ethylcarboxamidoadenosine (NECA) at 0.1, 1, or 10 μM for one hour. T cells were then activated with anti-CD3 + anti-CD28 antibodies and incubated for 48 hrs. Purified RNA was collected using a Qiagen RNAEasy Kit and gene expression analysis was performed using the NanoString PanCancer Immune Panel with PLUS codeset. Normalized counts were obtained using NanoString nSolver Software. Log2 transformed expression data were fit to a linear model comprised of donor and treatment effects. Genes which showed a statistically significant treatment effect were identified in 3 initial donors. Adjusted p-values were used to correct for multiple hypothesis testing using the Benjamini-Hochberg procedure.

SIGNATURE CORRELATES WITH TUMOR RESPONSE

Adenosine Signature High (PR=17%)

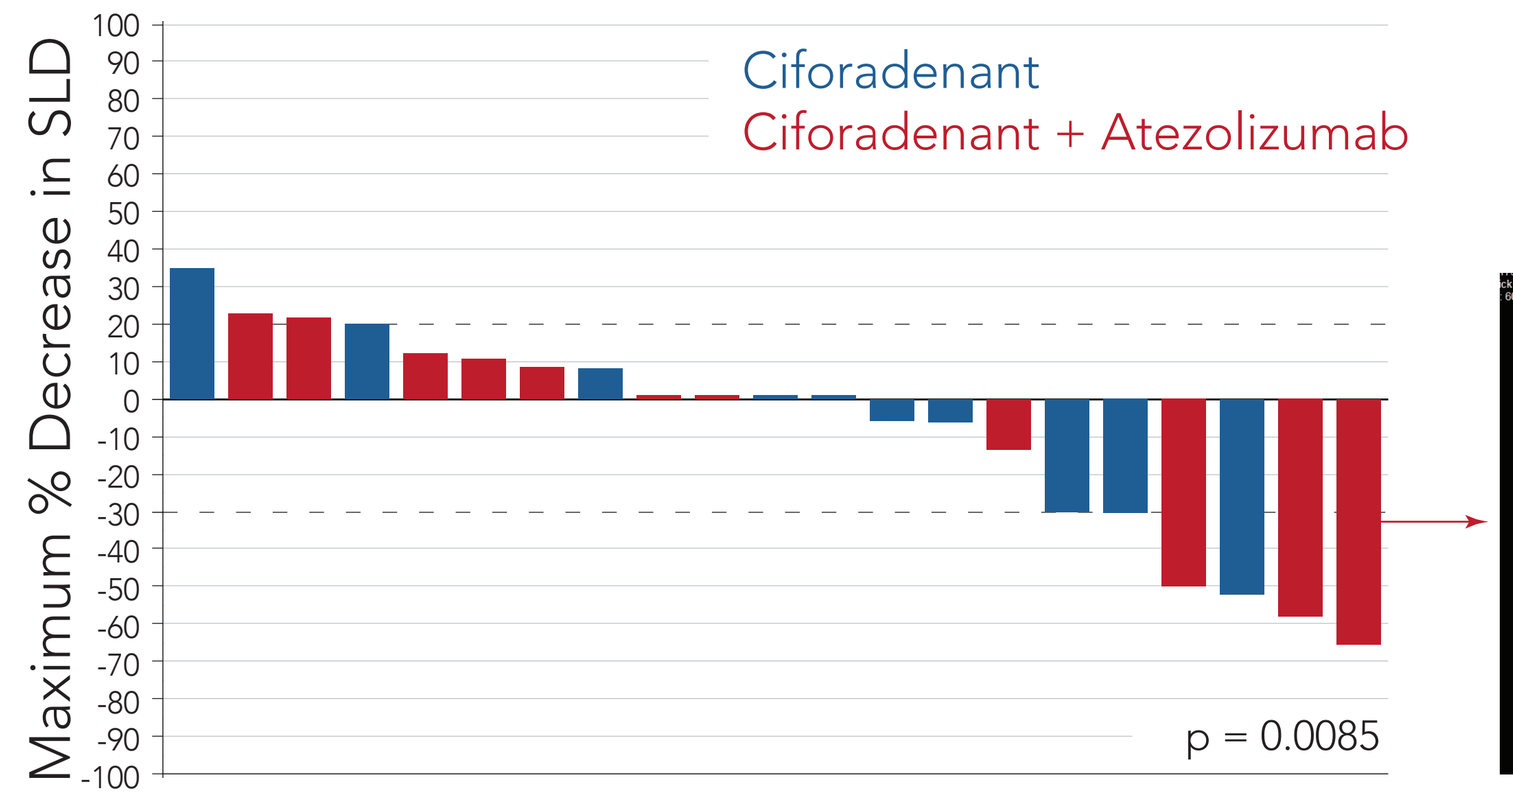
Maximum % Decrease in SLD
Ciforadenant
Ciforadenant + Atezolizumab
p = 0.0085

Adenosine Signature High Patient

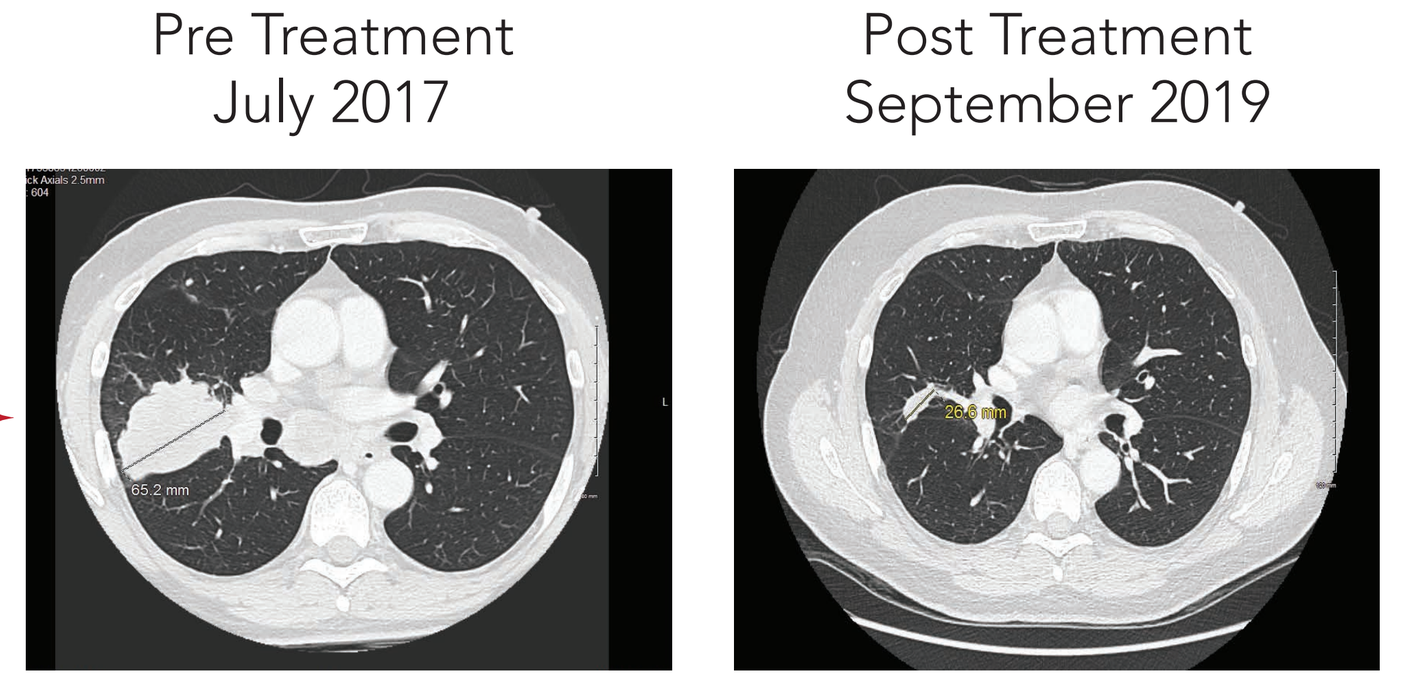
Pre Treatment
July 2017
Post Treatment
September 2019
65.2 mm

Adenosine Signature Low (PR=0%)

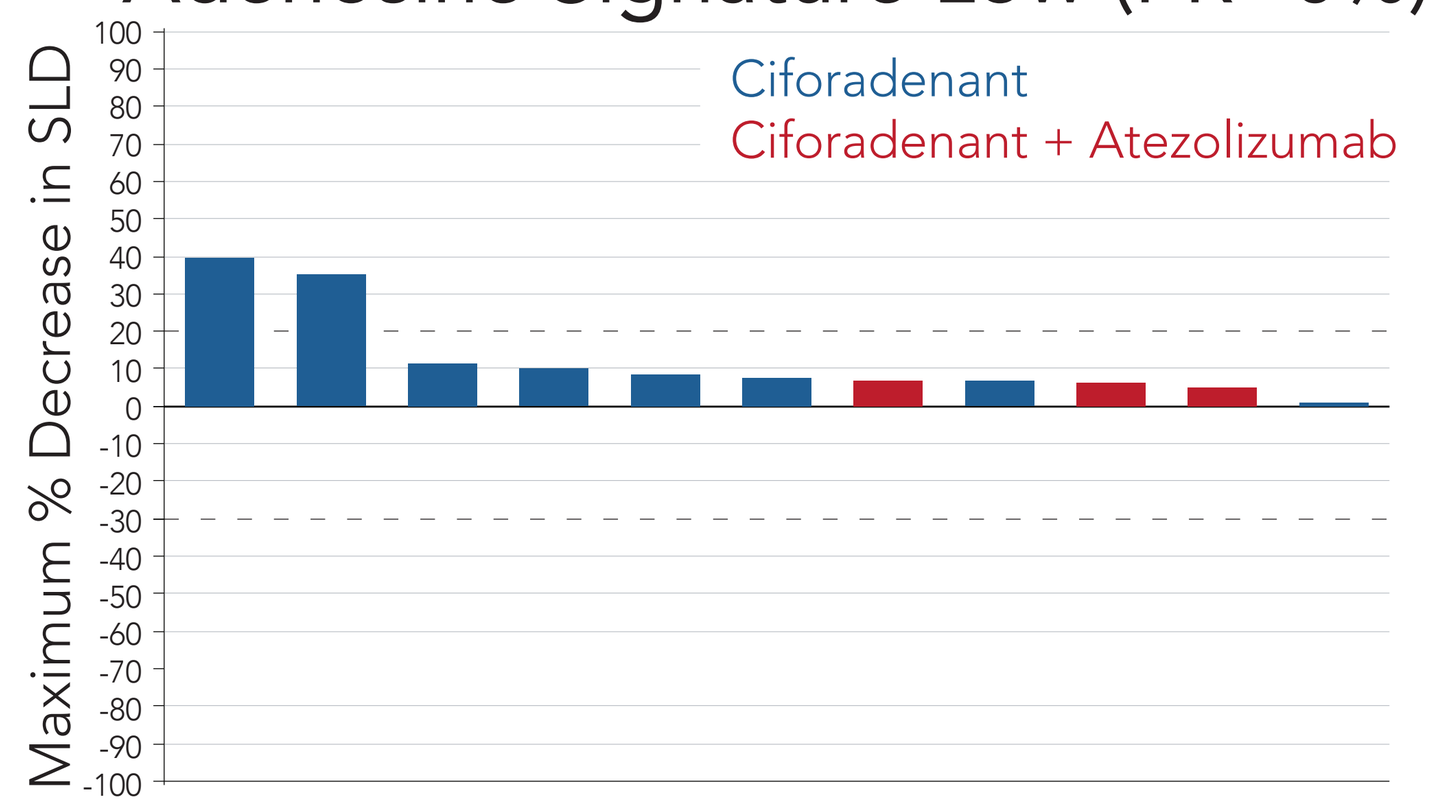
Maximum % Decrease in SLD
Ciforadenant
Ciforadenant + Atezolizumab

Clinical Trial Design:

- Metastatic renal cell cancer
 - Must have progressed on prior therapy
- Ciforadenant monotherapy
 - 100 mg BID 28d cycle
- Ciforadenant + atezolizumab
 - 100 mg BID 28d cycle + 840 mg, IV, 2QW
- Median 3 prior treatments (range 0-5)
- 85% of patients were resistant/refractory to prior IO

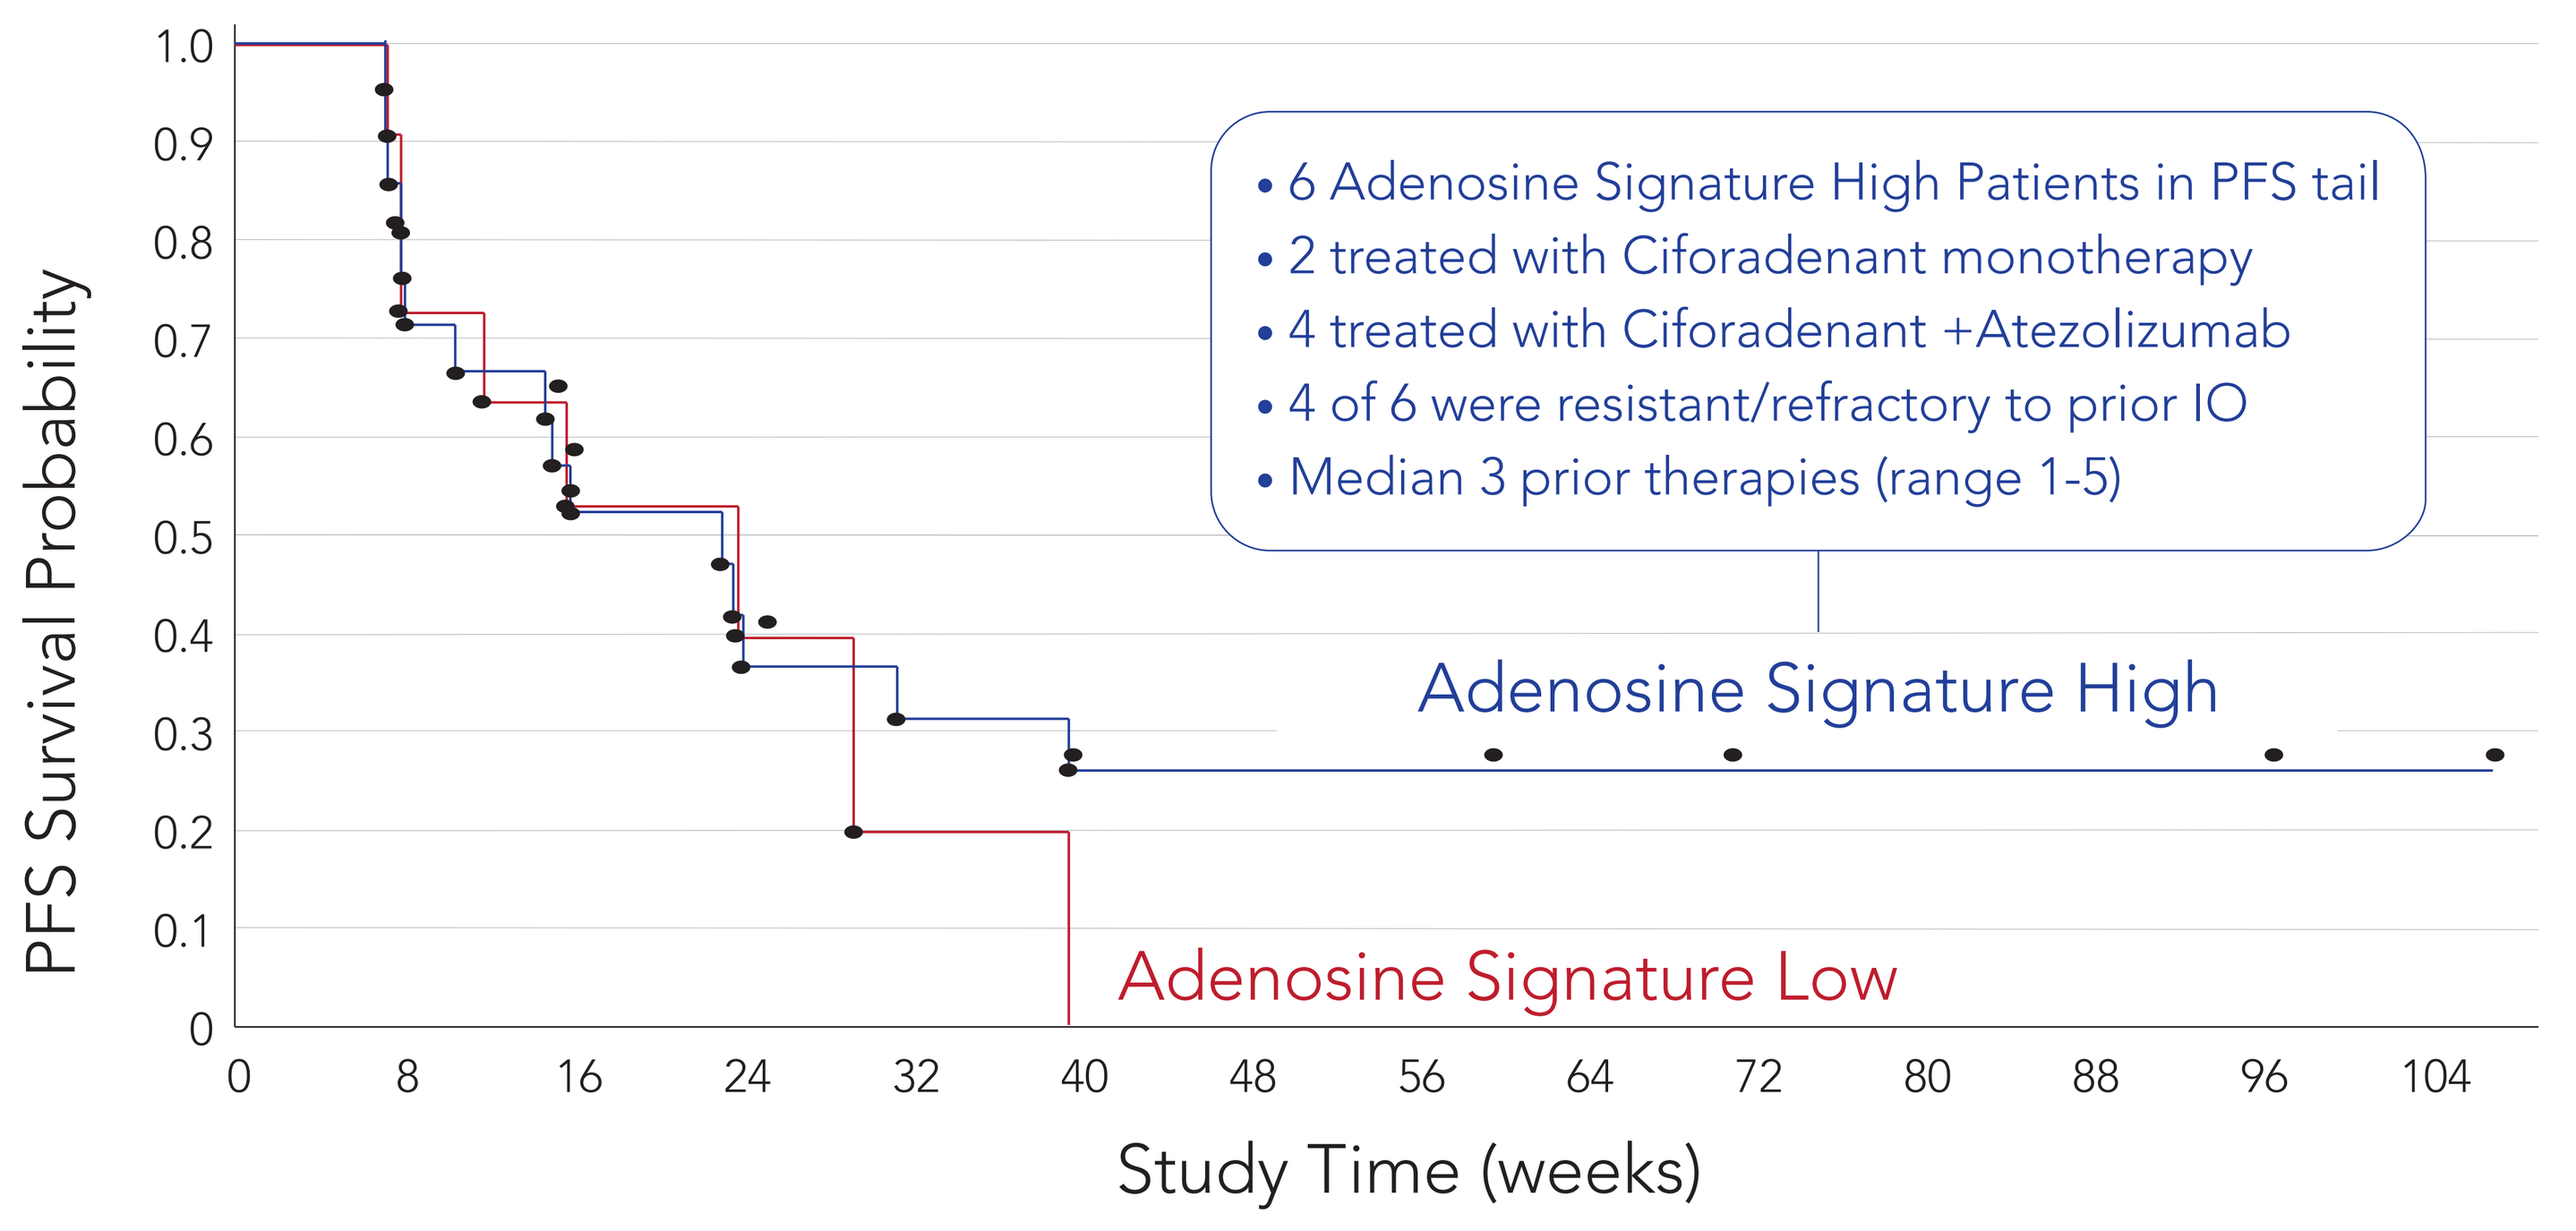
PFS Survival Probability
Study Time (weeks)
• 6 Adenosine Signature High Patients in PFS tail
• 2 treated with Ciforadenant monotherapy
• 4 treated with Ciforadenant +Atezolizumab
• 4 of 6 were resistant/refractory to prior IO
• Median 3 prior therapies (range 1-5)
Adenosine Signature High
Adenosine Signature Low

PRIOR TREATMENTS IN ADENOSINE SIGNATURE HIGH PATIENTS WITH PROLONGED PFS

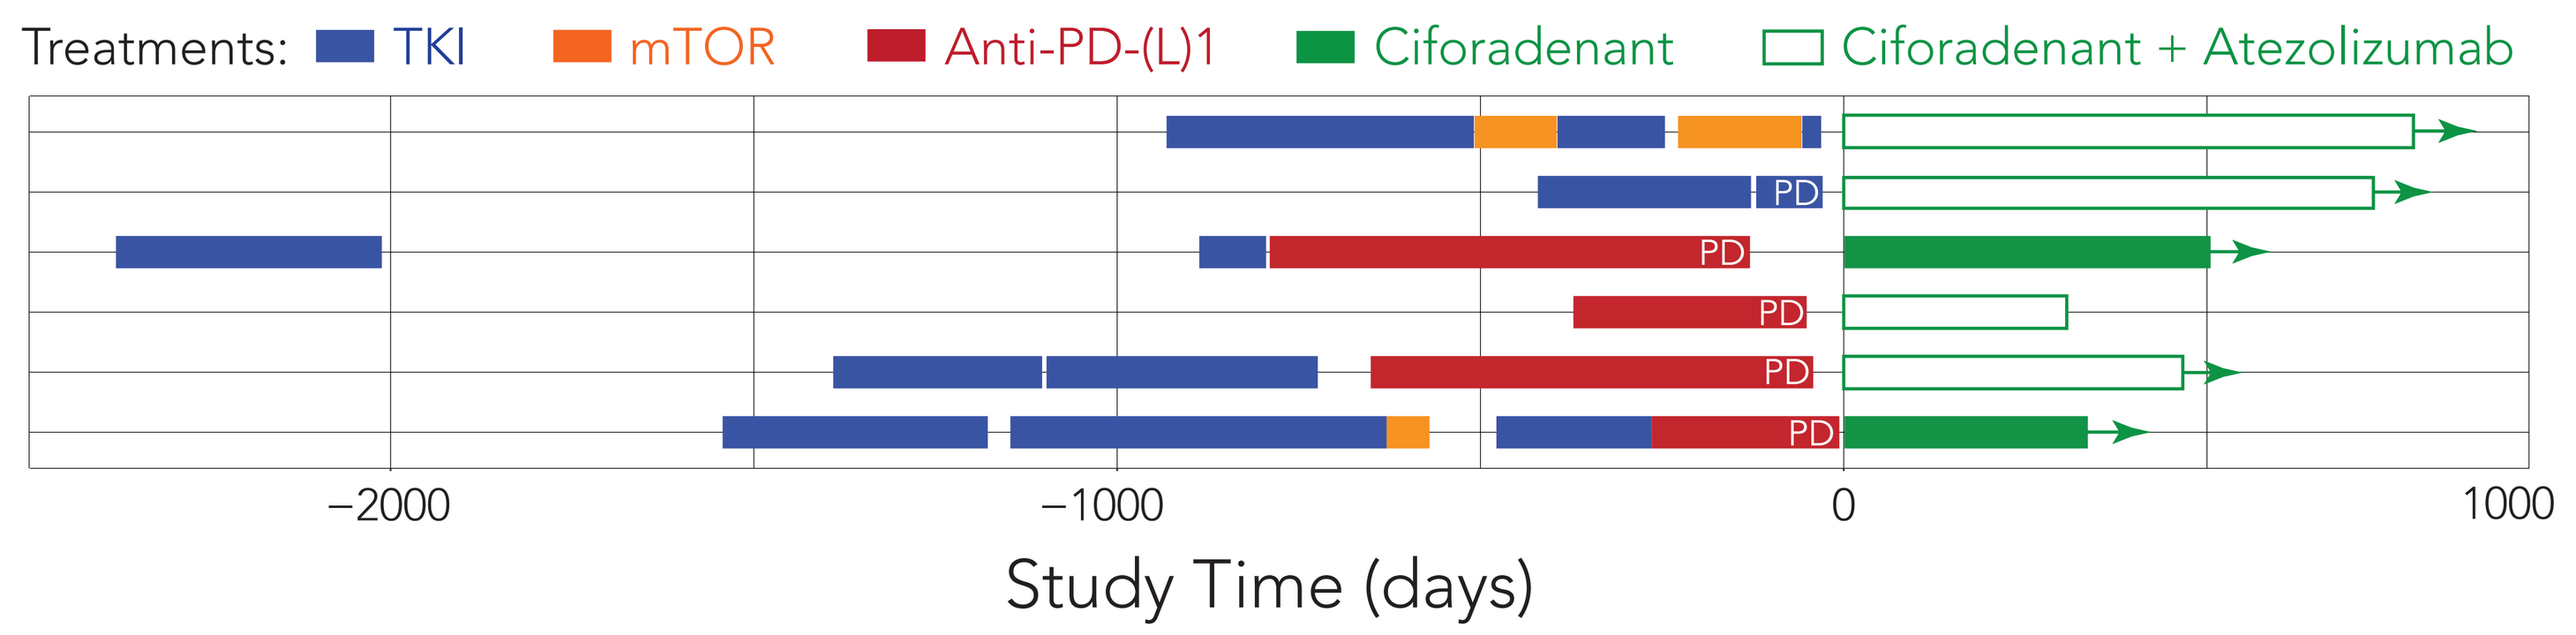
Treatments: TKI mTOR Anti-PD-(L)1 Ciforadenant Ciforadenant + Atezolizumab
PD
−2000
−1000
0
1000
Study Time (days)

Adenosine Signature levels were determined in pre-treatment biopsy tissue. In brief, RNA was extracted from tumor tissue macrodissected from patient biopsy specimens and analyzed using the NanoString PanCancer Immune Panel. Cutoffs for determining Adenosine Signature high and low tumors were based on the mean of the Log2 NanoString counts of select Adenosine Signature genes for all subjects evaluated. The t-test for comparing the maximum percentage decrease in SLD between the adenosine positive and negative signatures is 2-sided p-value = 0.0085, using a 5% level two-sided test.

AMP INDUCES ADENOSINE-LIKE GENE SIGNATURE

AMP INDUCED GENE SIGNATURE IS NEARLY IDENTICAL TO ADENOSINE SIGNATURE

AMPαS IS A NON-HYDROLIZABLE FORM OF AMP - IT DOES NOT FORM ADENOSINE

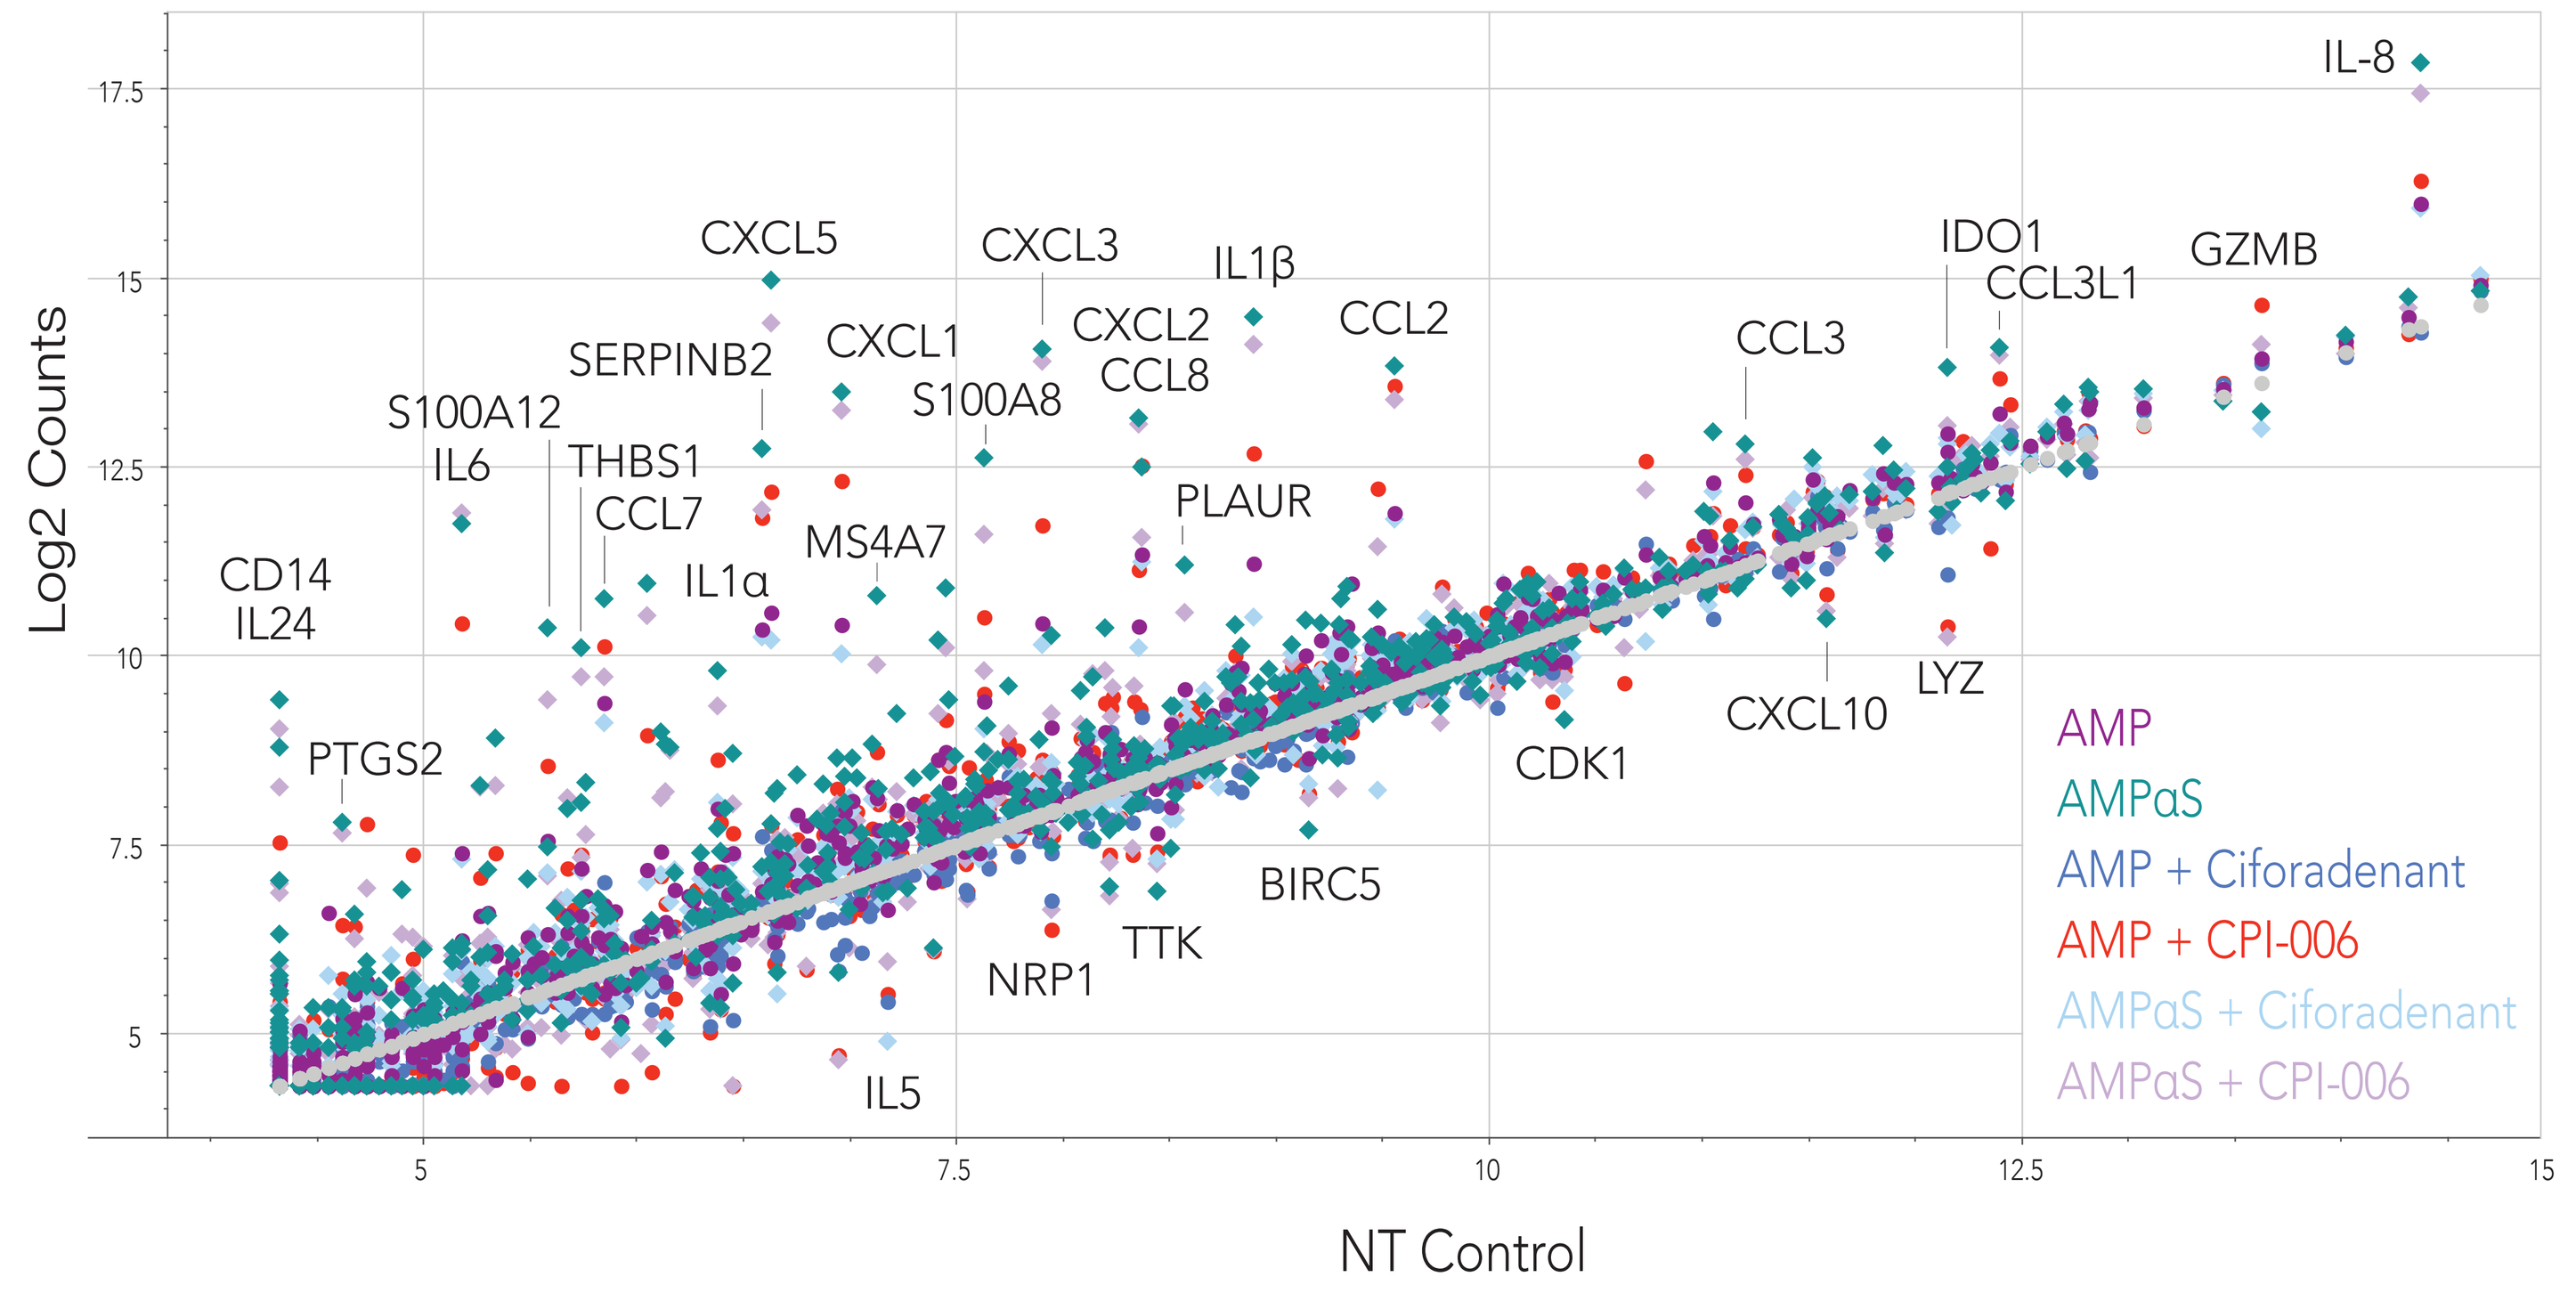
Log2 Counts
NT Control
IL-8
IDO1
CCL3L1
GZMB
CXCL5
CXCL3
IL1β
CCL2
CCL3
SERPINB2
CXCL1
CXCL2
CCL8
S100A8
S100A12
IL6
THBS1
CCL7
IL1α
MS4A7
PLAUR
CD14
IL24
PTGS2
CDK1
CXCL10
LYZ
BIRC5
TTK
NRP1
IL5
AMP
AMPαS
AMP + Ciforadenant
AMP + CPI-006
AMPαS + Ciforadenant
AMPαS + CPI-006

Human peripheral blood mononuclear cells (PBMCs) were isolated from buffy coat samples by density centrifugation with Histopaque 1077 (400*g, 30 min). Cells were washed and resuspended at a density $2*10^6$ cells/ml in RPMI + 10% human serum. PBMCs were co-cultured with ciforadenant (7 μM) or CPI-006 (10 μg/mL) for 1 hour before adding AMP (150 μM) or AMPαS (150 μM). PBMCs were then stimulated after 1 hour with anti-CD3 (clone HIT3a, 1 μg/ml) and anti-CD28 (clone CD28.2, 1 μg/ml) antibodies and incubated for 48 hours at 37 degrees. Purified RNA was collected using a Qiagen RNAEasy Kit according to manufacturer's protocol. NanoString analysis was performed according to manufacturer's protocol on a NanoString Sprint instrument using the NanoString PanCancer Immune Panel with PLUS codeset. Normalized counts were obtained using NanoString nSolver Software. Representative data from one of >3 patients is shown above.

CD73 ANTAGONISTS AMPLIFY AMP SIGNATURE

CD73 ANTAGONISTS INHIBIT ADENOSINE FORMATION, BUT CONSEQUENTLY PRESERVE AMP

PRESERVED AMP AMPLIFIES ADENOSINE-LIKE GENE SIGNATURE SUGGESTING AMP SIGNALS THROUGH A2AR

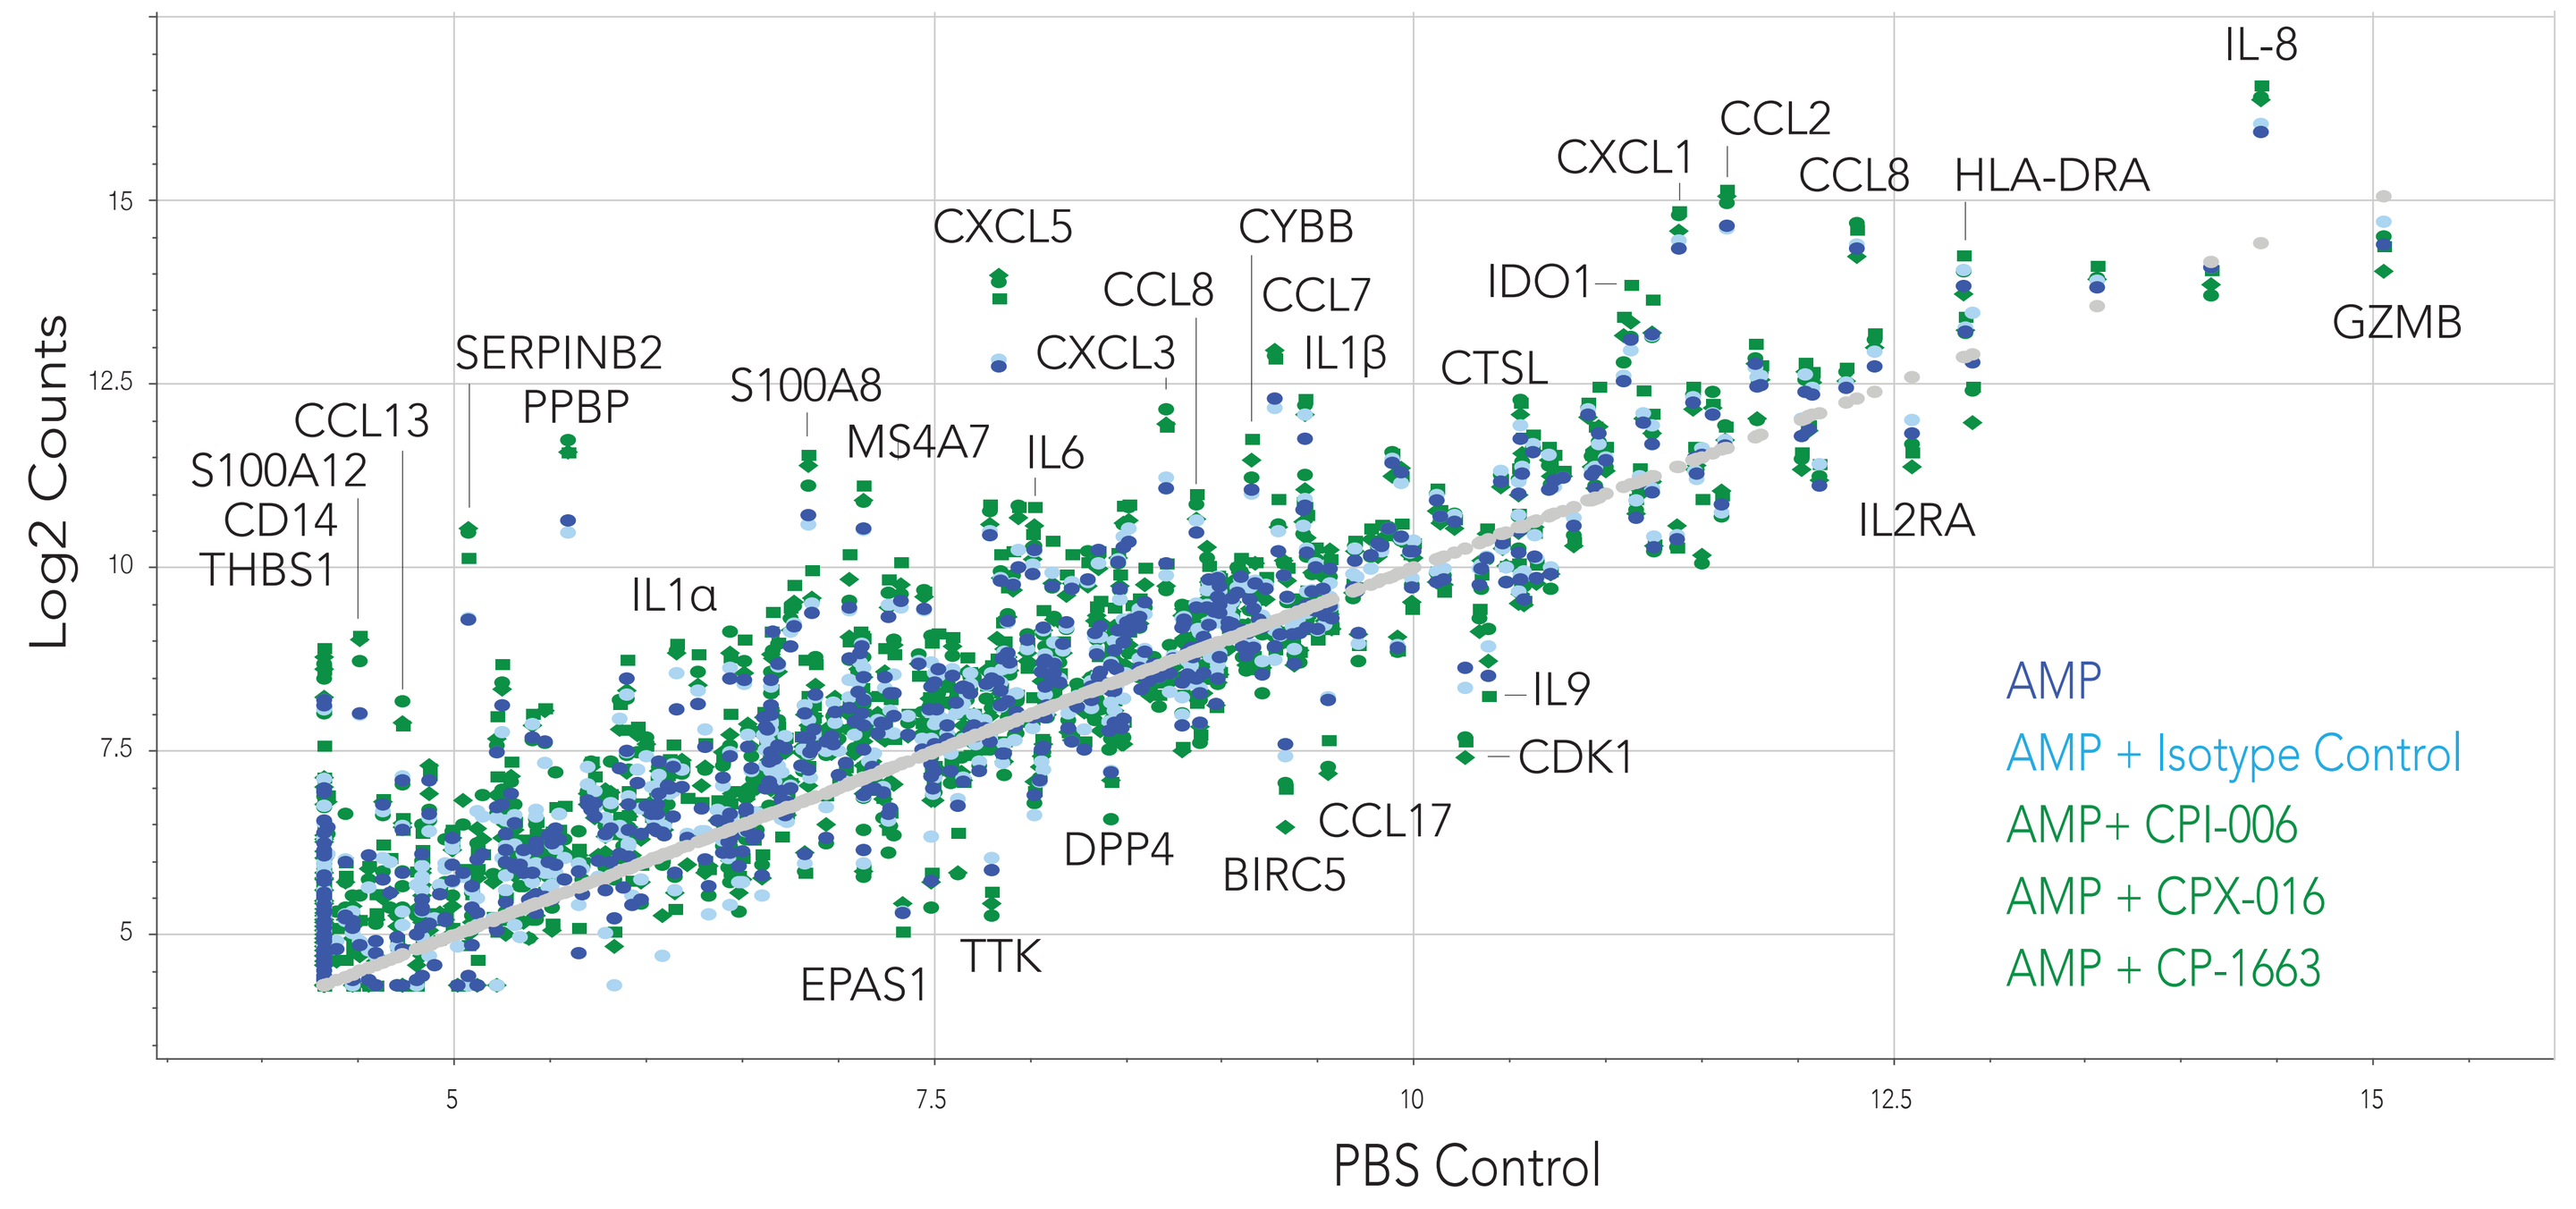
Log2 Counts
PBS Control
IL-8
CXCL1
CCL2
CCL8
HLA-DRA
GZMB
CXCL5
CYBB
CCL8
CCL7
CXCL3
IL1β
IDO1
CTSL
SERPINB2
PPBP
S100A8
MS4A7
IL6
CCL13
S100A12
CD14
THBS1
IL1α
IL2RA
IL9
CDK1
CCL17
DPP4
BIRC5
EPAS1
TTK
AMP
AMP + Isotype Control
AMP+ CPI-006
AMP + CPX-016
AMP + CP-1663

CPI-006 AND CPX-016 ARE ANTI-CD73 ANTIBODIES. CP-1663 IS A CD73 SMALL MOLECULE INHIBITOR

Purified human PBMCs from healthy donors were co-cultured with CD73 antagonists including CPI-006 (Corvus anti-CD73 mAb, 10 μg/mL), CPX-016 (competitor anti-CD73 mAb, 10 μg/mL), an isotype control antibody, or CP-1663 (1 μM)) for 1 hour before adding AMP (150 μM). PBMCs were then stimulated after 1 hour with anti-human CD3 and CD28 antibodies. RNA was purified from cells collected after incubation for 48 hours. Gene expression changes were evaluated using the NanoString PanCancer Immune Panel. Representative data from one of >3 patients is shown above.

AMP IS AN ADENOSINE RECEPTOR AGONIST

AMP AND AMPαS AGONIZE A2AR, BUT ARE WEAKER THAN ADENOSINE

A2AR	EC50 (nM)	n
NECA	0.17	n = 2
ADENOSINE	2.7	n = 4
AMP	235.2	n = 4
AMPαS	228.6	n = 4

A2BR	EC50 (nM)	n
NECA	1.3	n = 2
ADENOSINE	10.8	n = 4
AMP	227.5	n = 4
AMPαS	427.3	n = 4

A1	EC50 (nM)	n
NECA	2.8	n = 2
ADENOSINE	11.9	n = 4
AMP	237.5	n = 4
AMPαS	418.2	n = 4

A3	EC50 (nM)	n
NECA	1.7	n = 2
ADENOSINE	12.9	n = 4
AMP	199.0	n = 4
AMPαS	240.1	n = 4

cAMP assays were performed at Pharmaron using PerkinElmer CHO-K1 or HEK293 cells designed to stably overexpress human adenosine receptors, including hA1, hA2AR, hA2BR, and hA3. 3-fold dilutions of test compounds were evaluated, starting at a maximum concentration of 1 μM (NECA) or 10 μM (adenosine, AMP, or AMPαS). EC50 concentrations are shown along with the number of experiment replicates.

CIFORADENANT NEUTRALIZES AMP & AMPαS

CIFORADENANT BLOCKS AMP AND AMPαS SIGNALING AT A2AR

A2AR	IC50 (nM)	n
ADENOSINE	4.0	n = 4
AMP	4.7	n = 2
AMPαS	7.1	n = 2

A2BR	IC50 (nM)	n
ADENOSINE	171.2	n = 4
AMP	204.8	n = 2
AMPαS	270.2	n = 2

A1	IC50 (nM)	n
ADENOSINE	200.9	n = 4
AMP	111.4	n = 2
AMPαS	62.0	n = 2

A3	IC50 (nM)	n
ADENOSINE	516.0	n = 4
AMP	231.7	n = 2
AMPαS	315.1	n = 2

cAMP assays were performed at Pharmaron using PerkinElmer CHO-K1 or HEK293 cells designed to stably overexpress human adenosine receptors, including hA1, hA2AR, hA2BR, and hA3. Cells were stimultated with EC80 concentrations of indicated test compounds. Ciforadenant was added at a maximum concentration of 10 μM. IC50 values for ciforadenant inhibition of test compounds are shown, along with the number of experiment replicates.

CONCLUSIONS

- A2AR agonists induce a specific gene signature in human immune cells. This "Adenosine Signature" is dominated by inflammatory myeloid cytokines and chemokines.
- **Updated clinical data confirms original reports that expression of the Adenosine Signature correlates with tumor regression in an ongoing Ph 1/1b trial in patients with advanced/refractory RCC.**
- **High Adenosine Signature expression has a statistically significant correlation with tumor response.**
- The Adenosine Signature may be used as a predictive biomarker to select patients most likely to respond to therapy with agents that antagonize adenosine production or signaling.
- AMP induces gene expression changes nearly identical to the Adenosine Signature in human immune cells, suggesting AMP can activate and signal through adenosine receptors. CD73 antagonists further amplify the AMP-associated gene expression changes as a consequence of preserving AMP.
- AMP is an agonist of adenosine receptors. AMP agonism of A2AR and A2BR can be blocked by ciforadenant.
- These data suggest treatment with CD73 antagonists will benefit from concominant blockade of A2AR to neutralize the resultant induction of AMP mediated gene expression changes.