Publication Number: 1442 / Submission ID: 174421 / 2023ASH Annual Meeting / Session 605 Molecular Pharmacology and Drug Resistance, Lymphoid Neoplasms / Saturday, December 9, 2023 5:30 PM - 7:30 PM / San Diego Convention Center, Halls G-H

Dynamic Single-Cell Profiling Reveals Novel Immune Regulatory Mechanism of ITKi Soquelitinib in Refractory T Cell Lymphoma

Ning Ding[1*], Yan Xie[1*], Yong Guo[2*], Kecheng Li[2*], Tingting DU[1*], Zhenjun Li[1*], Yuejiao Li[1*], Jiaowu Cao[1*], Dingyao Hu[1*], Lih-Yun Hsu[3*], Richard Miller[3*], Jun Zhu[1] and Yuqin Song, MD[1]

[1]Department of Lymphoma, Key Laboratory of Carcinogenesis and Translational Research (Ministry of Education), Peking University Cancer Hospital and Institute, Beijing, China;

[2]Angel Pharmaceuticals Co., Ltd., Jiaxing, China;

[3]Corvus Pharmaceuticals Inc, Burlingame, United States of America.

1. INTRODUCTION

Interleukin 2 inducible T cell kinase (ITK) plays an important role in the T cell receptor (TCR) signaling pathway.

The highly selective covalent ITK inhibitor, soquelitinib (SQL, formerly known as CPI-818), is being evaluated as a monotherapy in an ongoing Phase 1/1b trial in patients (pts) with refractory T-cell lymphoma (TCL) (NCT03952078).

Single cell RNA-sequencing and flow cytometry were performed on patient samples to identify potential prognostic biomarkers and to study immune regulatory mechanisms.

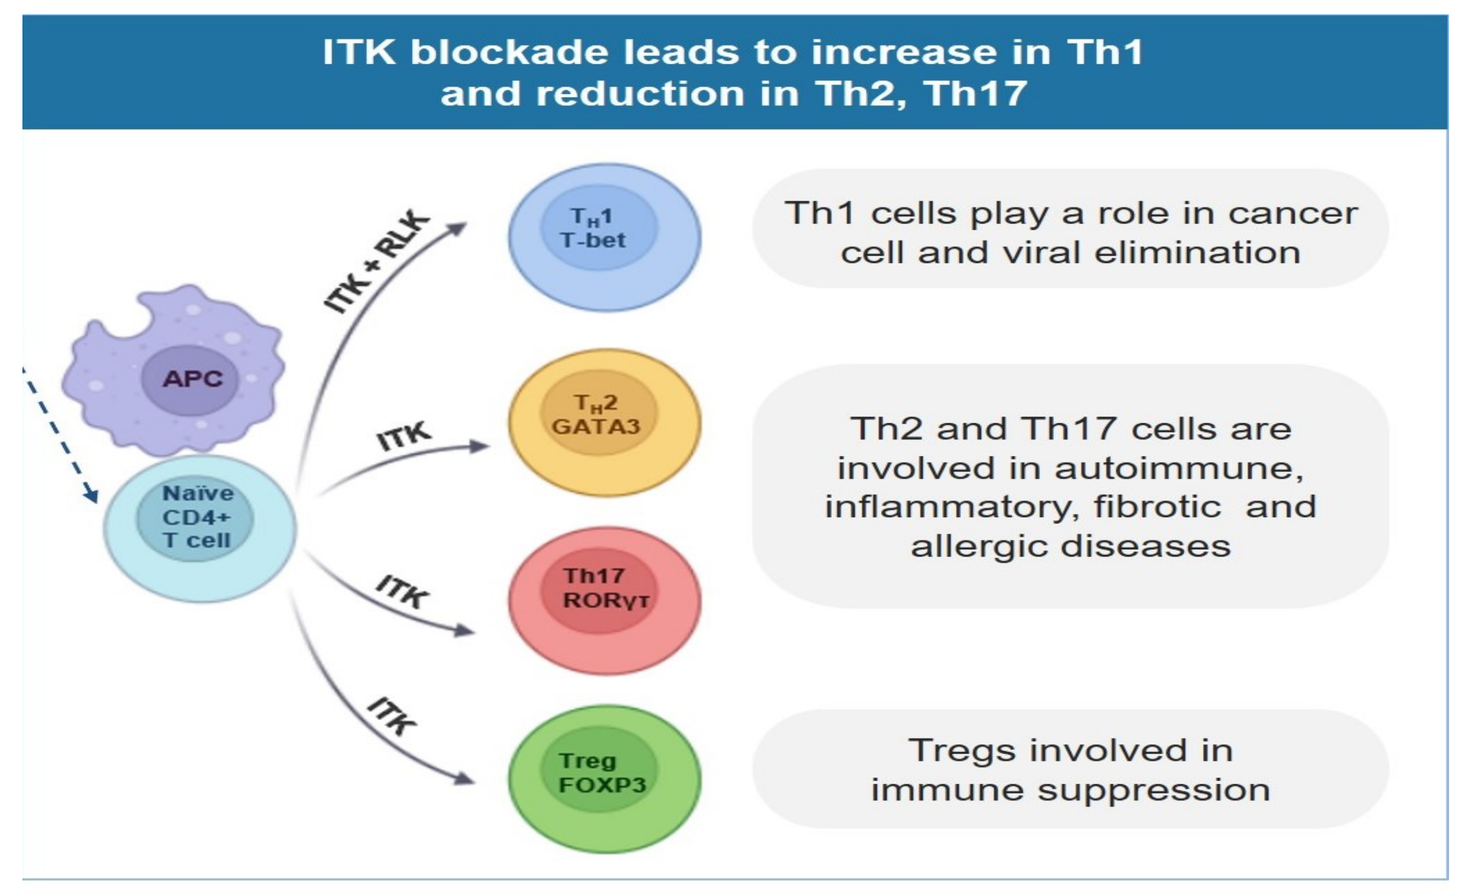
ITK blockade leads to increase in Th1 and reduction in Th2, Th17
APC
Naive CD4+ T cell
ITK + RLK
ITK
ITK
ITK
TH1 T-bet
TH2 GATA3
Th17 RORγt
Treg FOXP3
Th1 cells play a role in cancer cell and viral elimination
Th2 and Th17 cells are involved in autoimmune, inflammatory, fibrotic and allergic diseases
Tregs involved in immune suppression

ITK blockade leads to increase in Th1 and reduction in Th2, Th17

2. METHODS

Paired peripheral blood and tumor tissue samples were collected from 8 TCL patients enrolled in the trial. .

The single cell RNA-seq and flow cytometry were used to dynamically analyze the changes of gene expression profiling, followed by signature genes enrichment to identify ITK related molecular and immune regulatory mechanisms in TCL patients treated with soquelitinib.

3. PTS, SAFETY & EFFICACY

Patients for the assessment

Patient Characteristics	N=8
Age (yrs.), median (range)	57 (29, 76)
Gender, male N (%)	5 (62.5)
No. of prior therapies, median (range)	3 (1, 4)
HISTOLOGIES	
PTCL-NOS	4
AITL	2
ALCL	1
NKTCL	1

Summary of safety

Most common AEs (>1 patient) all causality were: anemia, bilirubin increased, neutrophil count decreased, WBC decreased, and pruritus. Grade 3+ AEs were reported in 1 patient: neutrophil count decreased and WBC decreased.

Target lesion best % changes from baseline

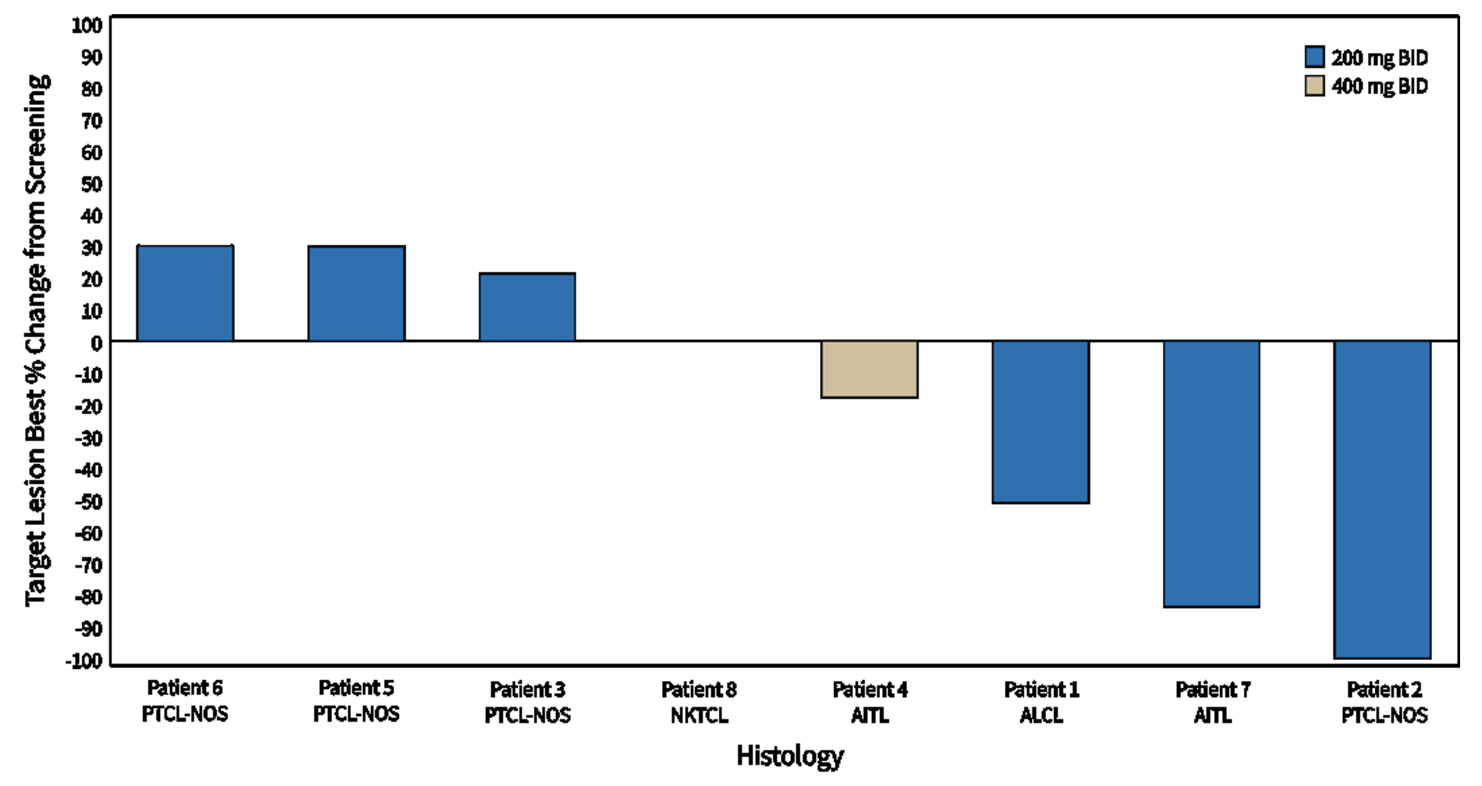
Target Lesion Best % Change from Screening
200 mg BID
400 mg BID
Patient 6 PTCL-NOS
Patient 5 PTCL-NOS
Patient 3 PTCL-NOS
Patient 8 NKTCL
Patient 4 AITL
Patient 1 ALCL
Patient 7 AITL
Patient 2 PTCL-NOS
Histology

4. RESULTS

2 responding pts: The percentage of normal CD4+ Th1 cells and CD8+ TEMRA cells post-SQL treatment

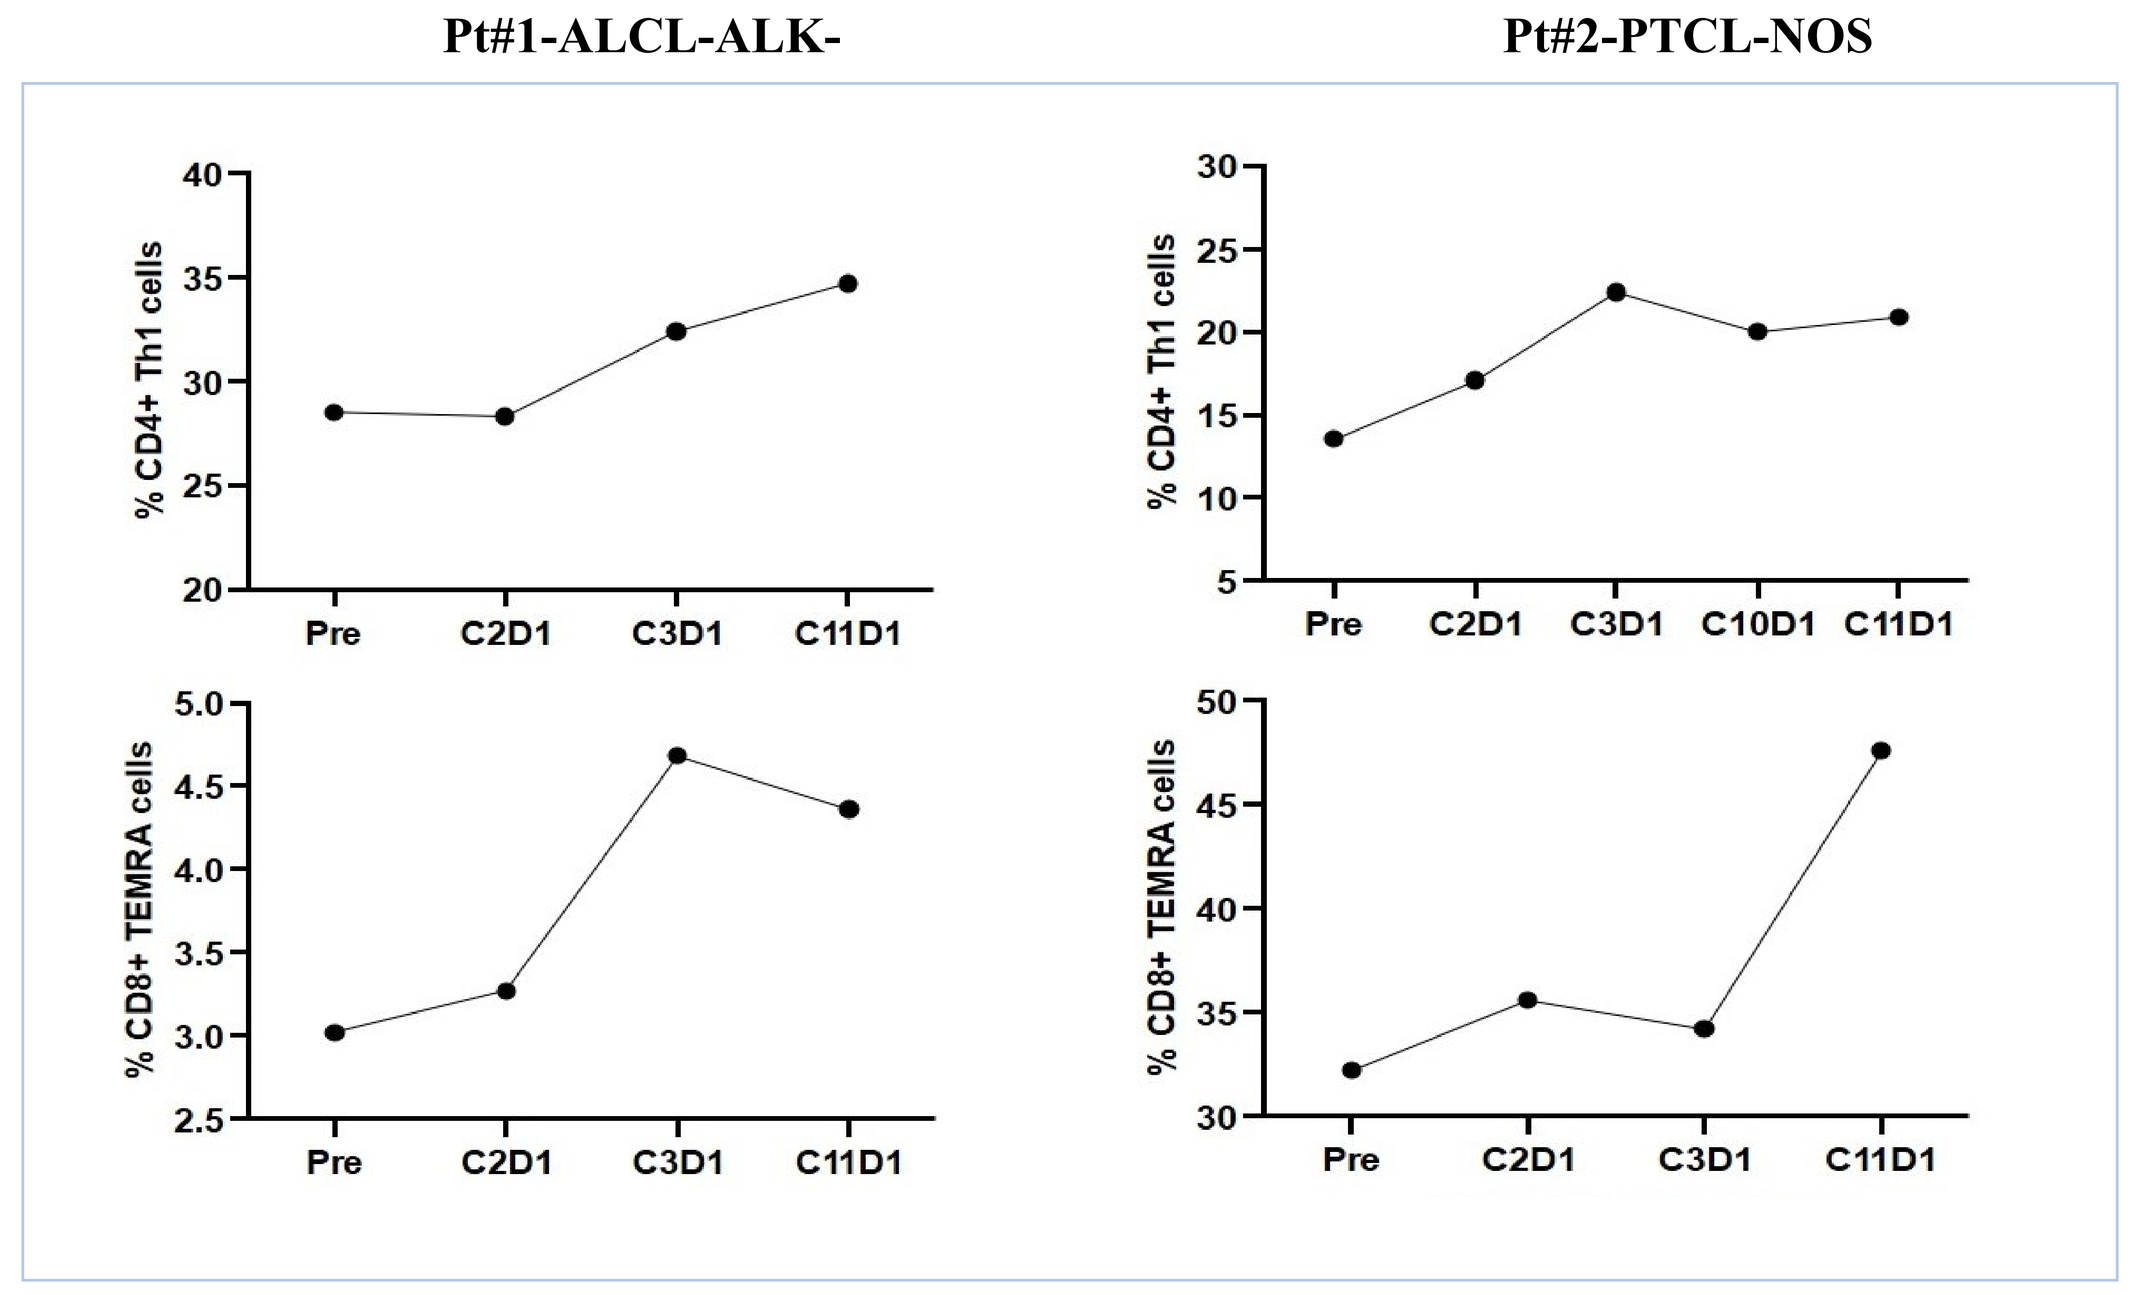
Pt#1-ALCL-ALK-
Pt#2-PTCL-NOS
% CD4+ Th1 cells
% CD8+ TEMRA cells
Pre
C2D1
C3D1
C10D1
C11D1

Figure 1. In the 2 patients with objective response (2 partial responses), the percentage of normal CD4+ Th1 cells and CD8+ TEMRA cells was increased and sustained after soquelitinib treatment. Increased levels of normal CD4+ Th1 cells and CD8+ TEMRA cells correlated with the clinical responses.

3 PD pts: The percentage of normal CD4+ Th1 cells and CD8+ TEMRA cells post-SQL treatment

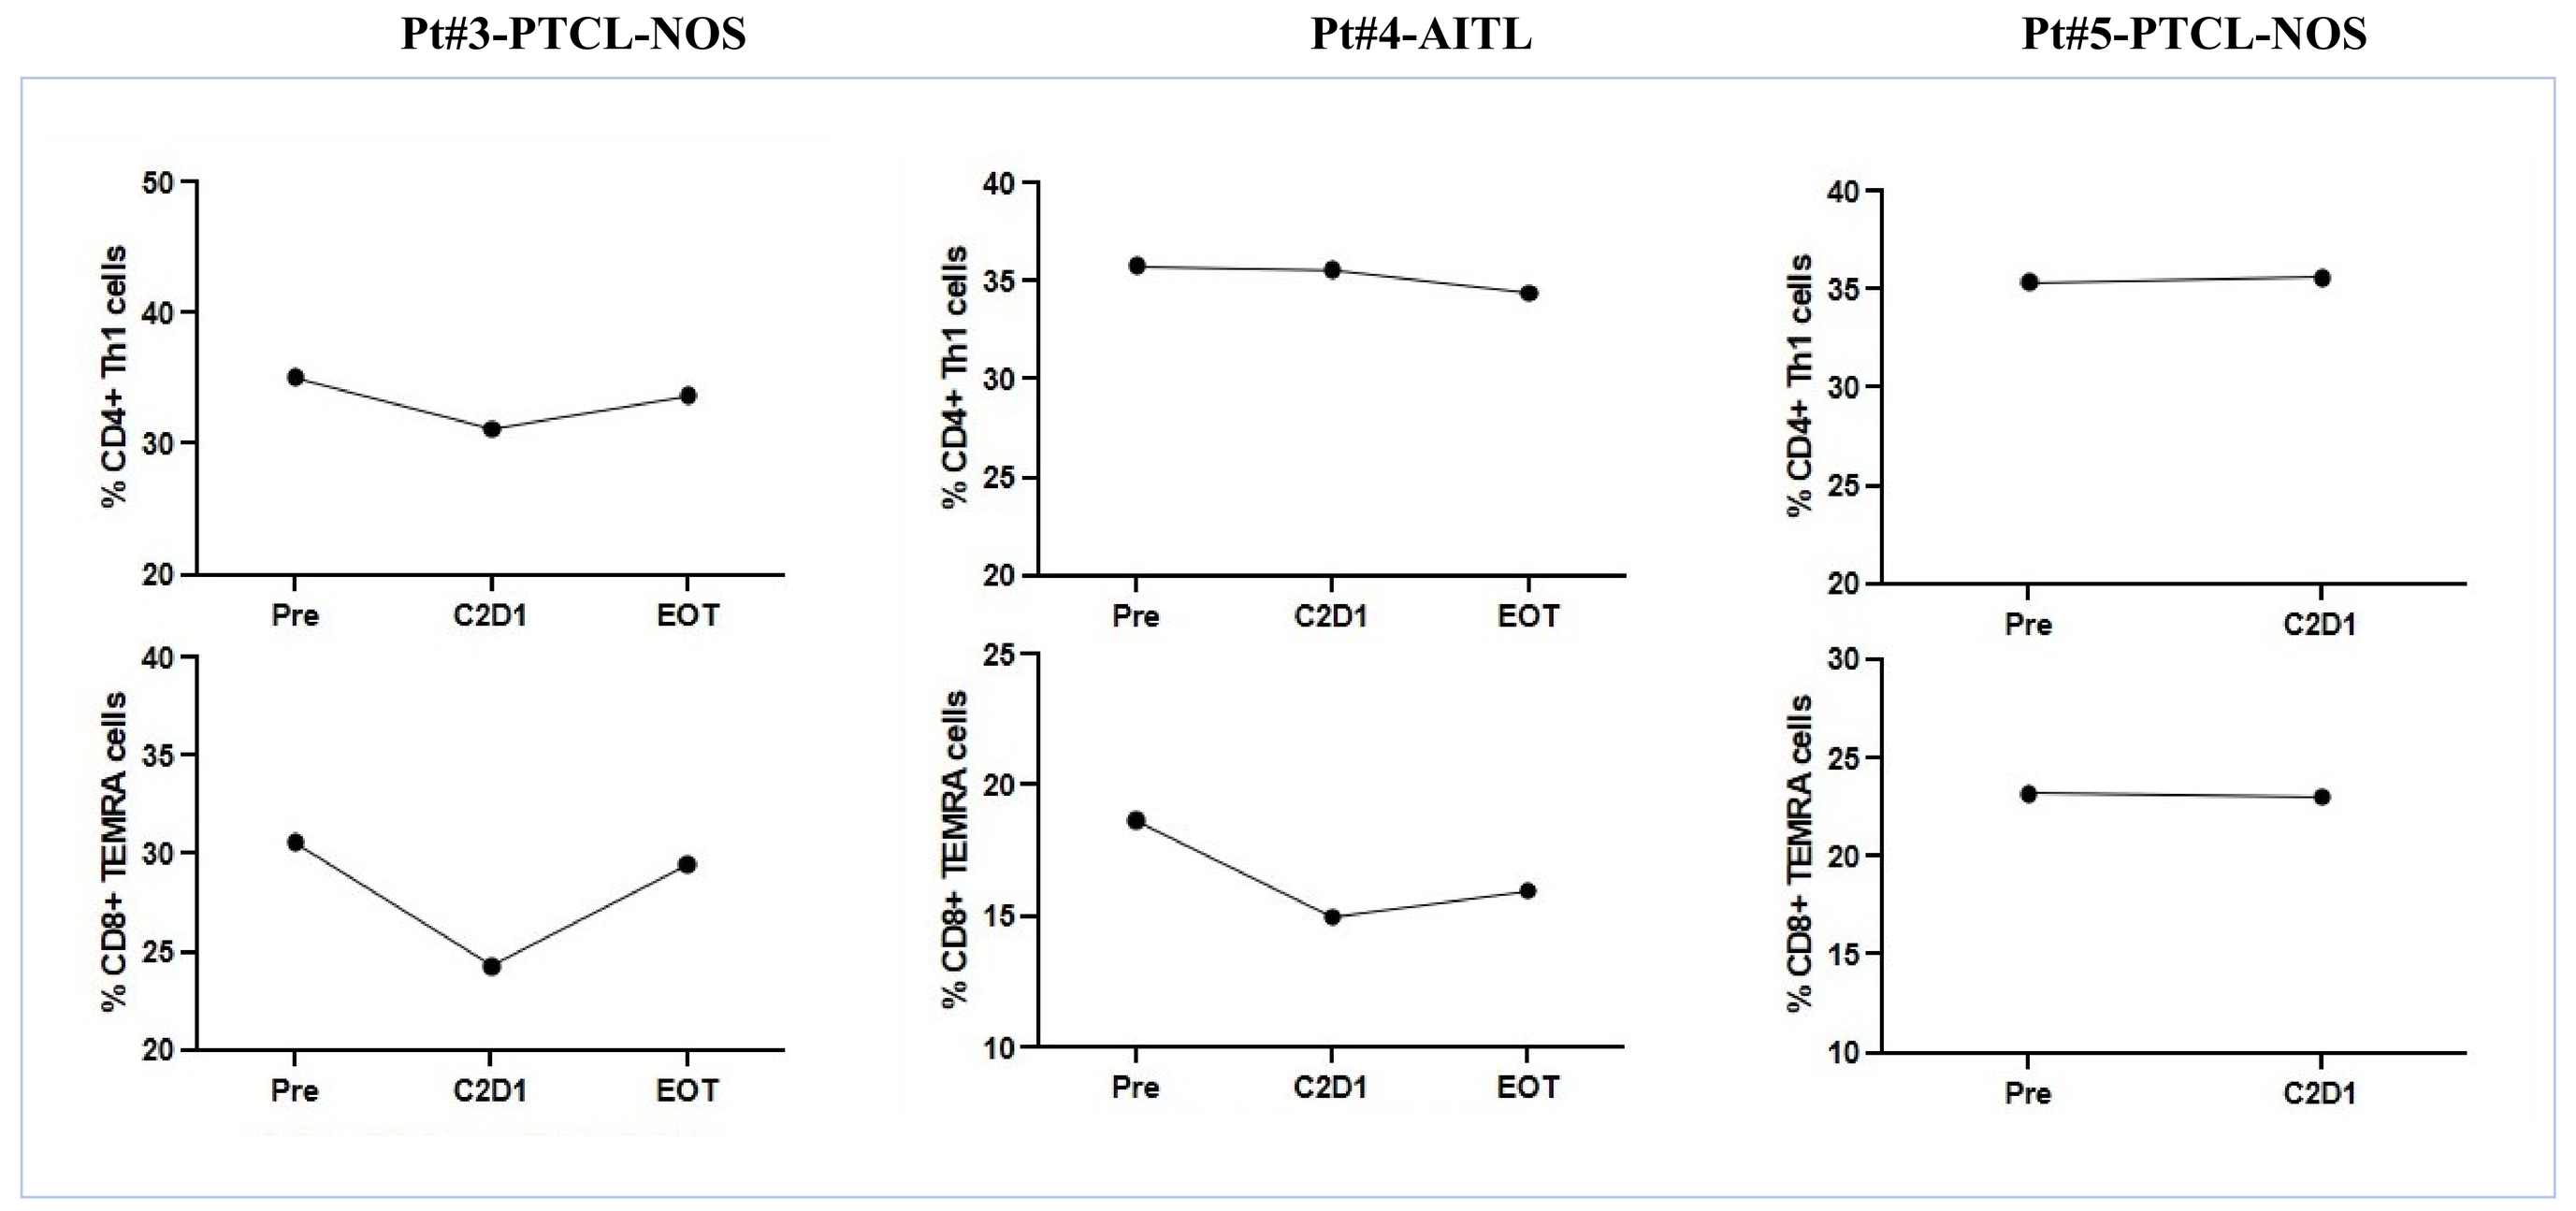
Pt#3-PTCL-NOS
Pt#4-AITL
Pt#5-PTCL-NOS
% CD4+ Th1 cells
% CD8+ TEMRA cells
Pre
C2D1
EOT

Figure 2. In the 3 patients with progressive disease (PD), the percentage of normal CD4+ Th1 cells and CD8+ TEMRA cells was not increased after soquelitinib treatment.
Peripheral blood from PD patients were collected at various timepoints and T cell subsets evaluated by flow cytometry. Plots show percent change from baseline for CD4+ Th1 and for CD8+ TEMRA cells. EOT = end of treatment.

4 SD pts: The percentage of normal CD4+ Th1 cells and CD8+ TEMRA cells post-SQL treatment

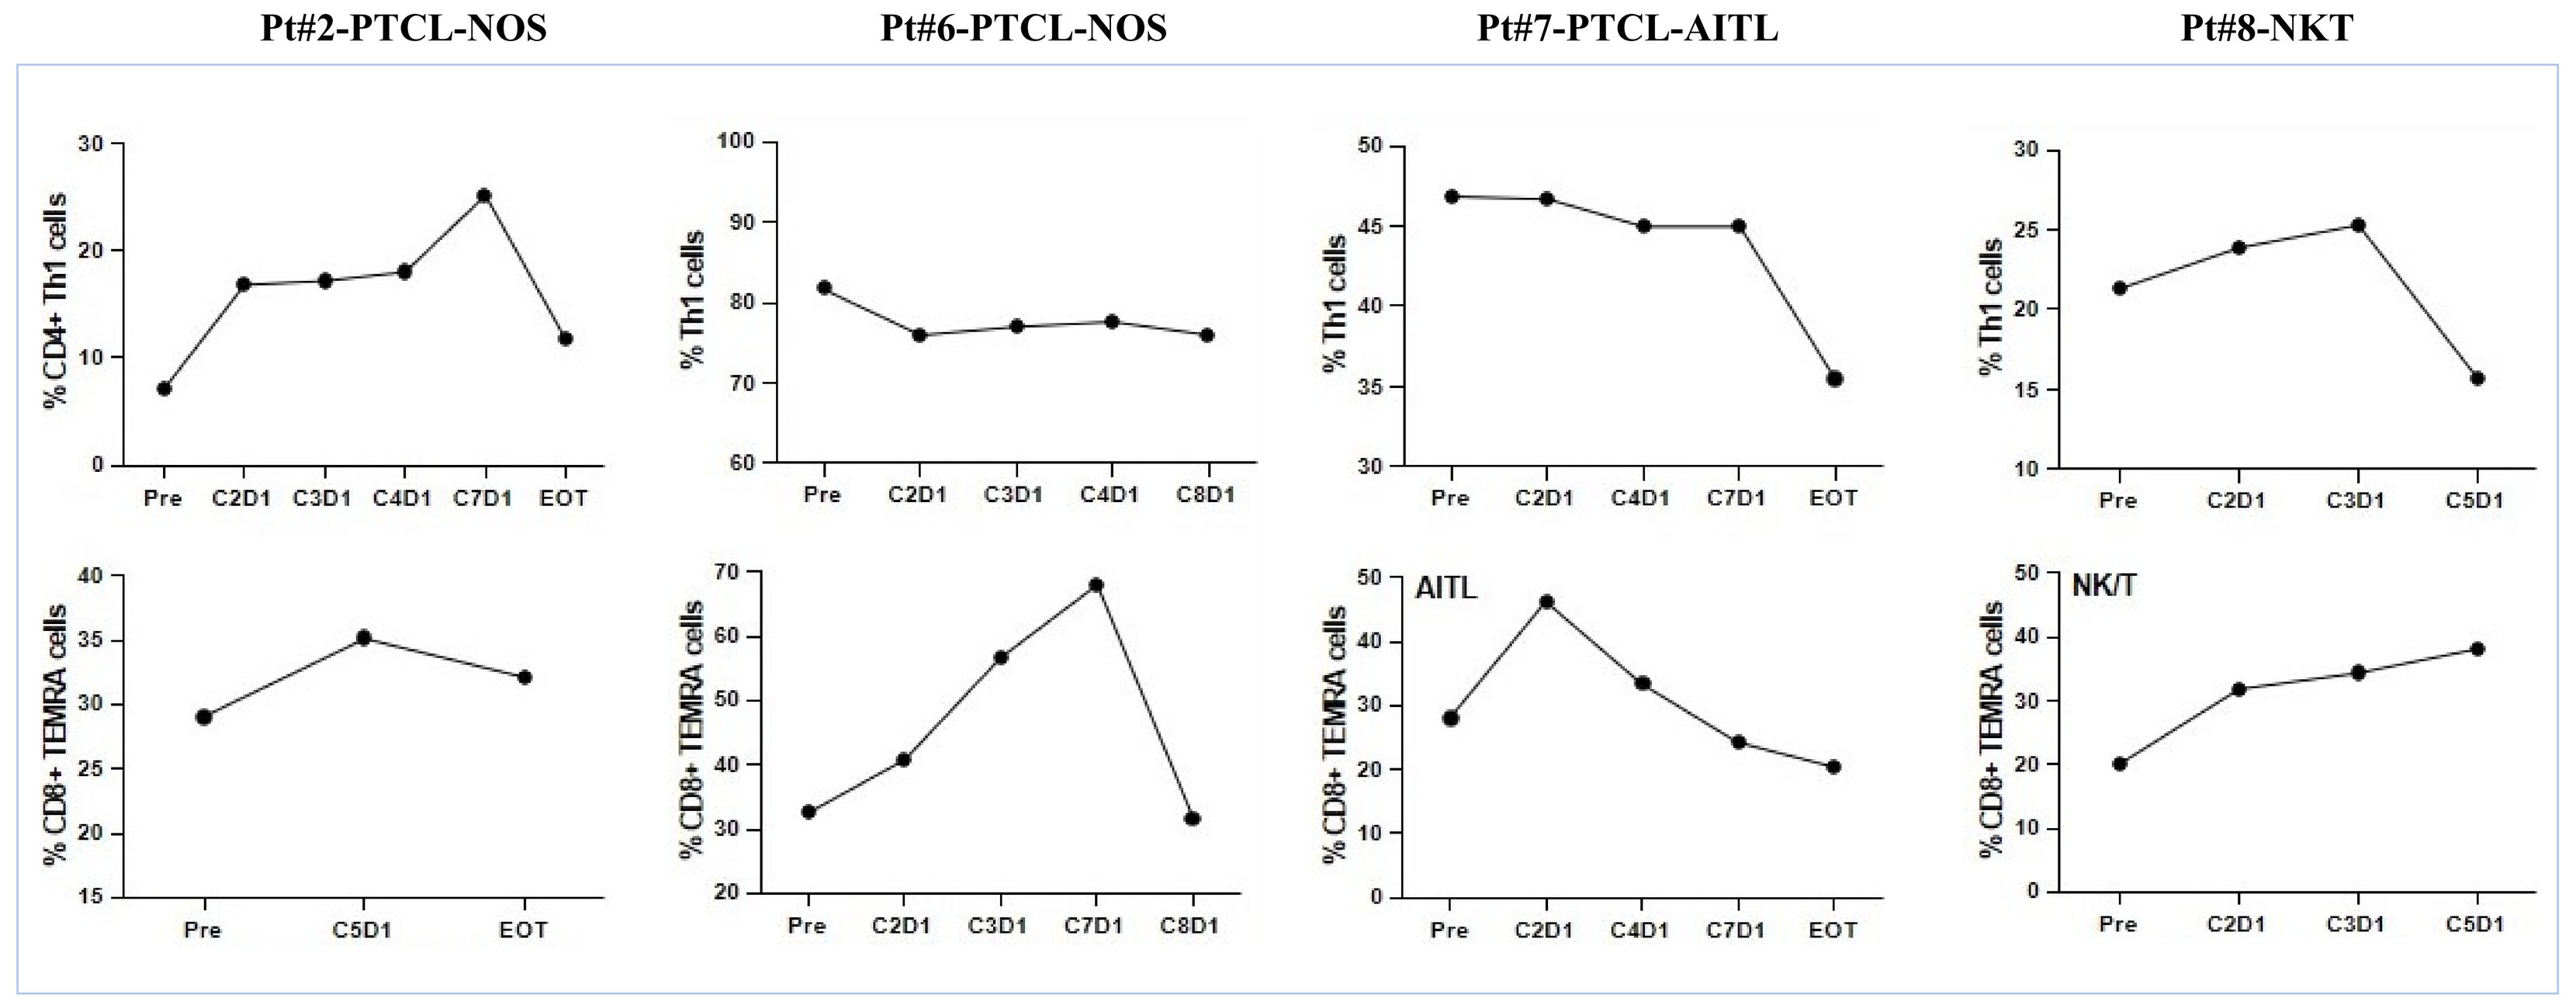
Pt#2-PTCL-NOS
Pt#6-PTCL-NOS
Pt#7-PTCL-AITL
Pt#8-NKT
% CD4+ Th1 cells
% Th1 cells
% CD8+ TEMRA cells
AITL
NK/T
Pre
C2D1
C3D1
C4D1
C5D1
C7D1
C8D1
EOT

Figure 3. In the 4 patients who achieved stable disease (SD), the percentage of normal CD4+ Th1 cells and CD8+ TEMRA cells was increased at the very beginning and decreased in the late phase of soquelitinib treatment.
Peripheral blood from SD patients was collected at various timepoints and T cell subsets evaluated by flow cytometry. Plots show percent change from baseline for CD4+ Th1 and for CD8+ TEMRA cells. Patient #2 sample obtained during period of stable disease.

SQL induced cytotoxic gene expression - the dynamic single-cell profiling

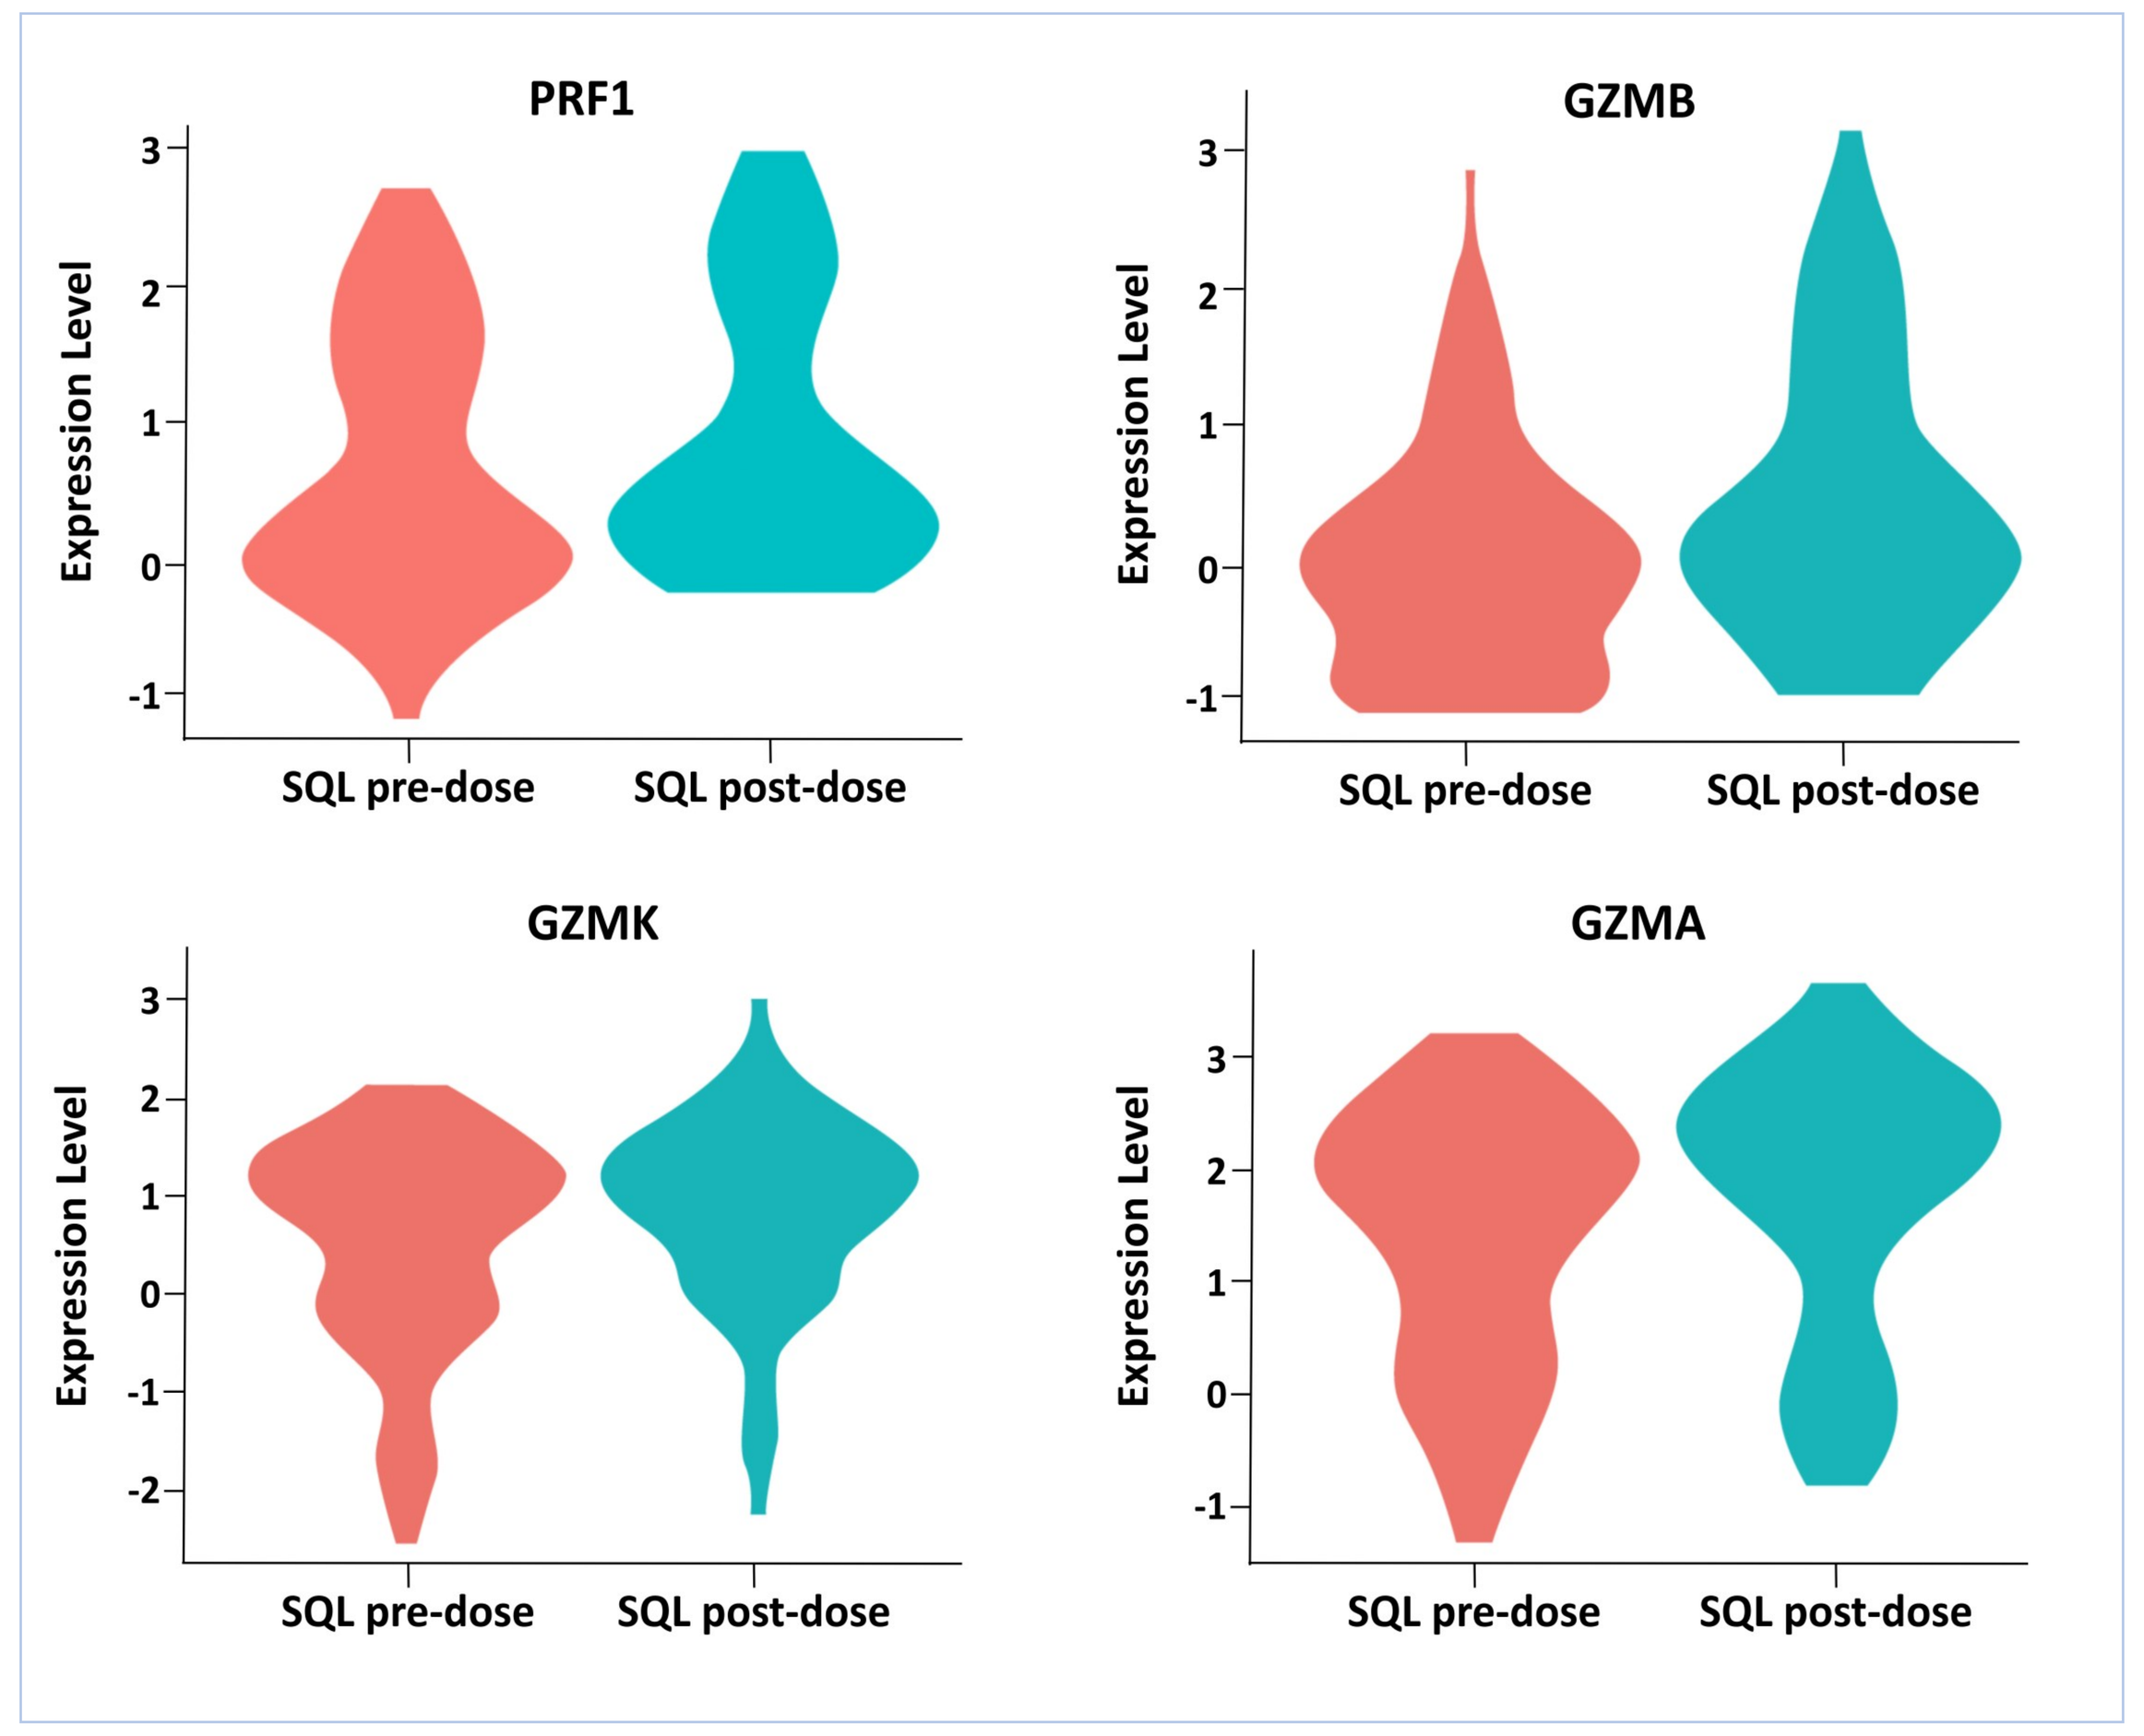
PRF1
GZMB
GZMK
GZMA
Expression Level
SQL pre-dose
SQL post-dose

Figure 4. Tumor tissue from patients was collected at different time points and assayed by single-cell RNA-seq. Single-cell sequencing studies have shown that ITK inhibitor SQL can increase the expression PRF1 (perforin 1), GZMB (granzyme B), GZMK (granzyme K) and GZMA (granzyme A) mRNA in infiltrating CD8+ cells in tumor tissue. Violin plots showing the distribution of gene expression levels for selected differentially expressed genes over the time course.

SQL decreased exhaust gene expression - the dynamic single-cell profiling

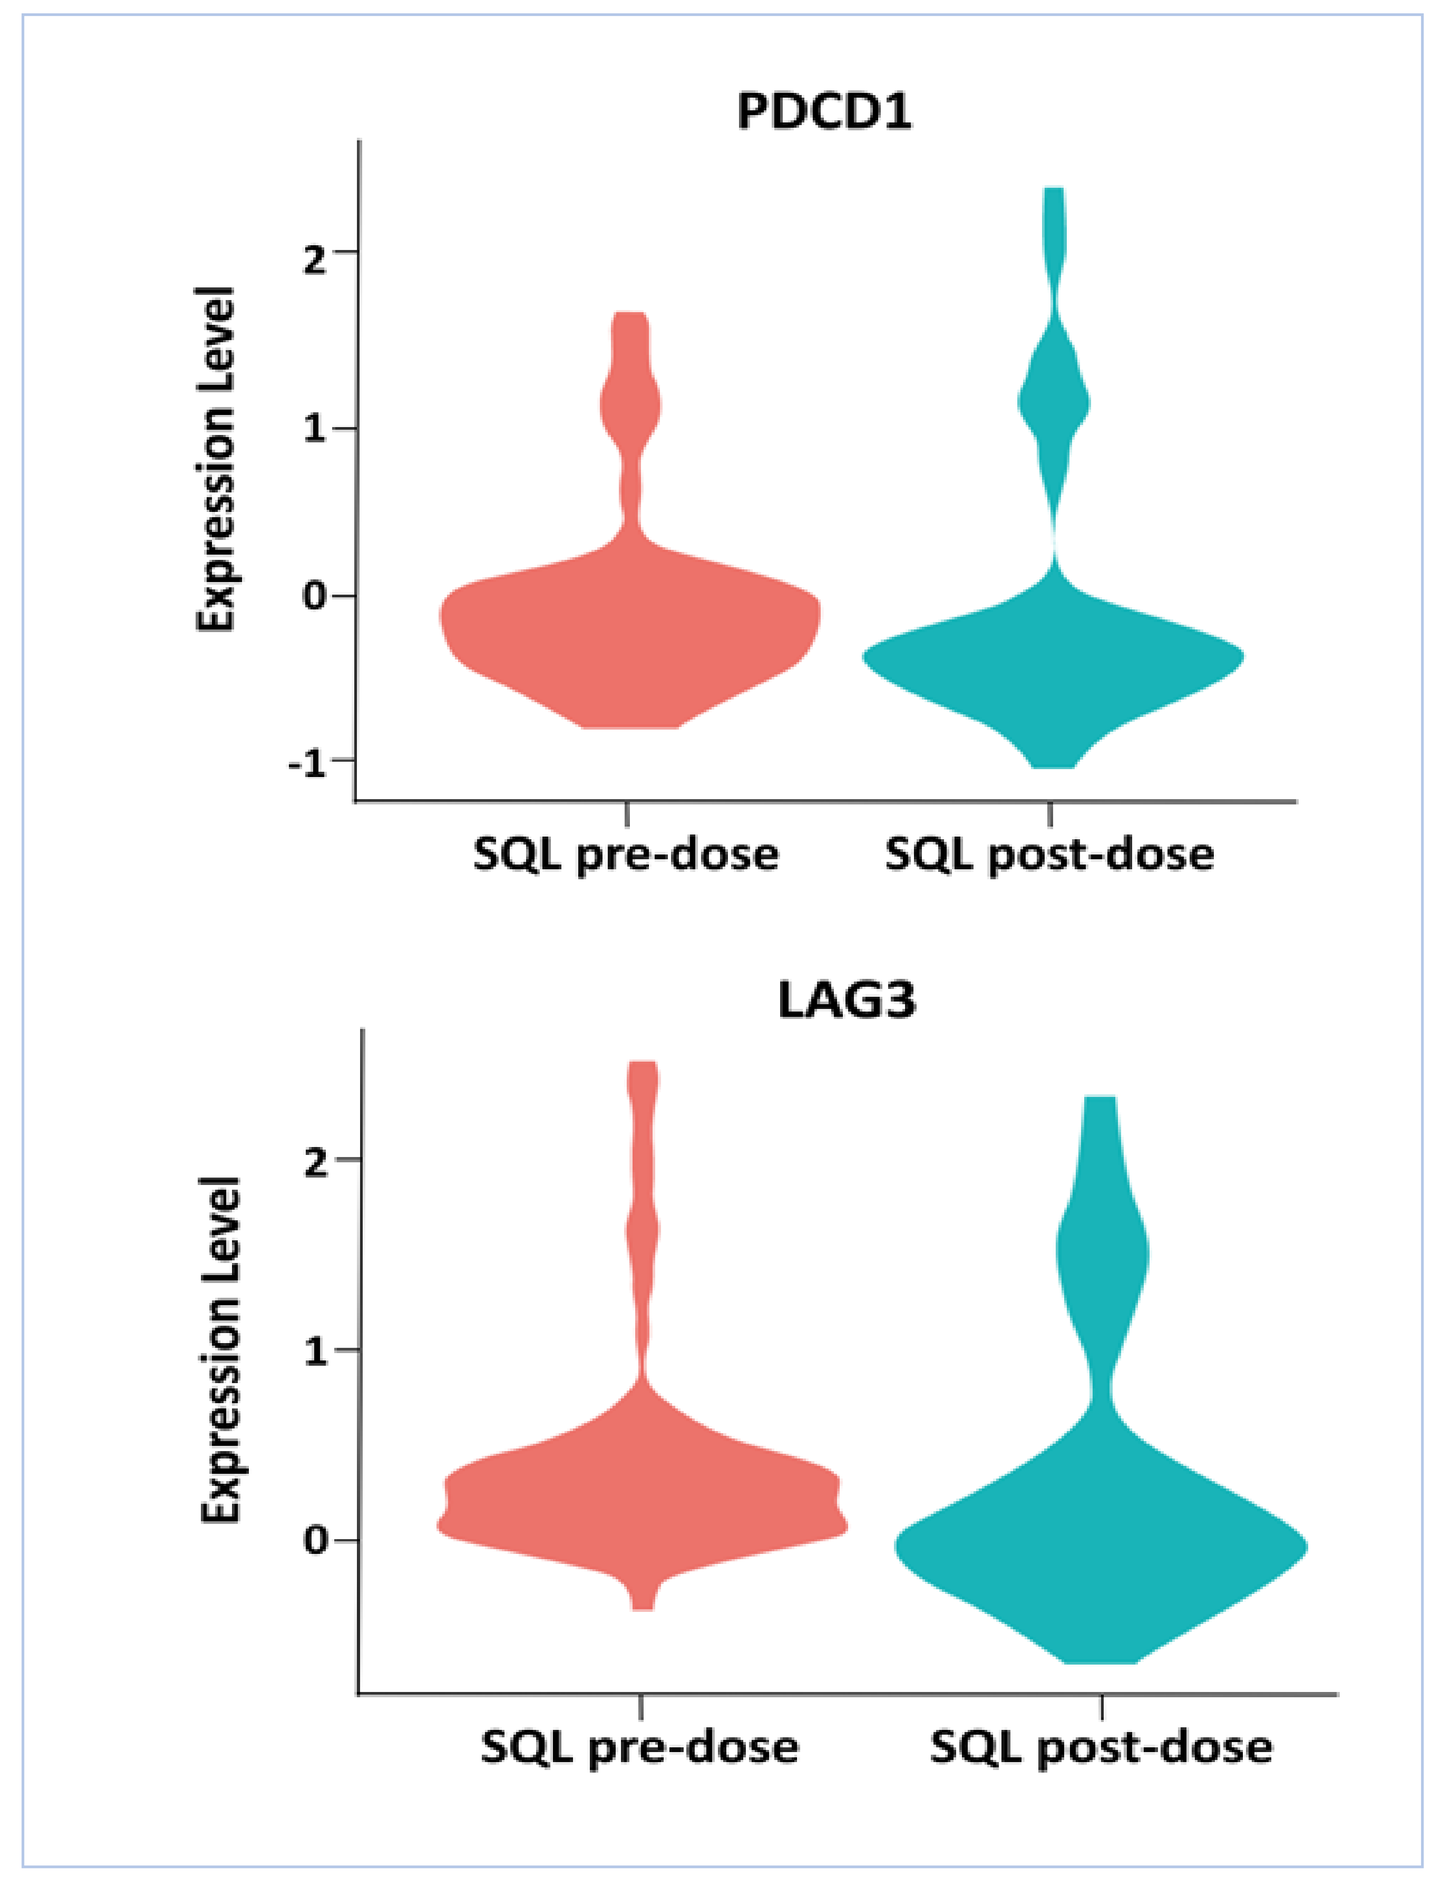
PDCD1
LAG3
Expression Level
SQL pre-dose
SQL post-dose

Figure 5. Tumor tissue from patients was collected at different time points and assayed by single-cell RNA-seq. Single-cell sequencing studies have shown that ITK inhibitors can downregulate the expression of various exhaustion molecules such as PDCD1 and LAG3. Violin plots showing the distribution of gene expression levels for selected differentially expressed genes over the time course.

The protein-protein interaction network analysis reveals that the hub gene TBX21 might play a central regulatory role

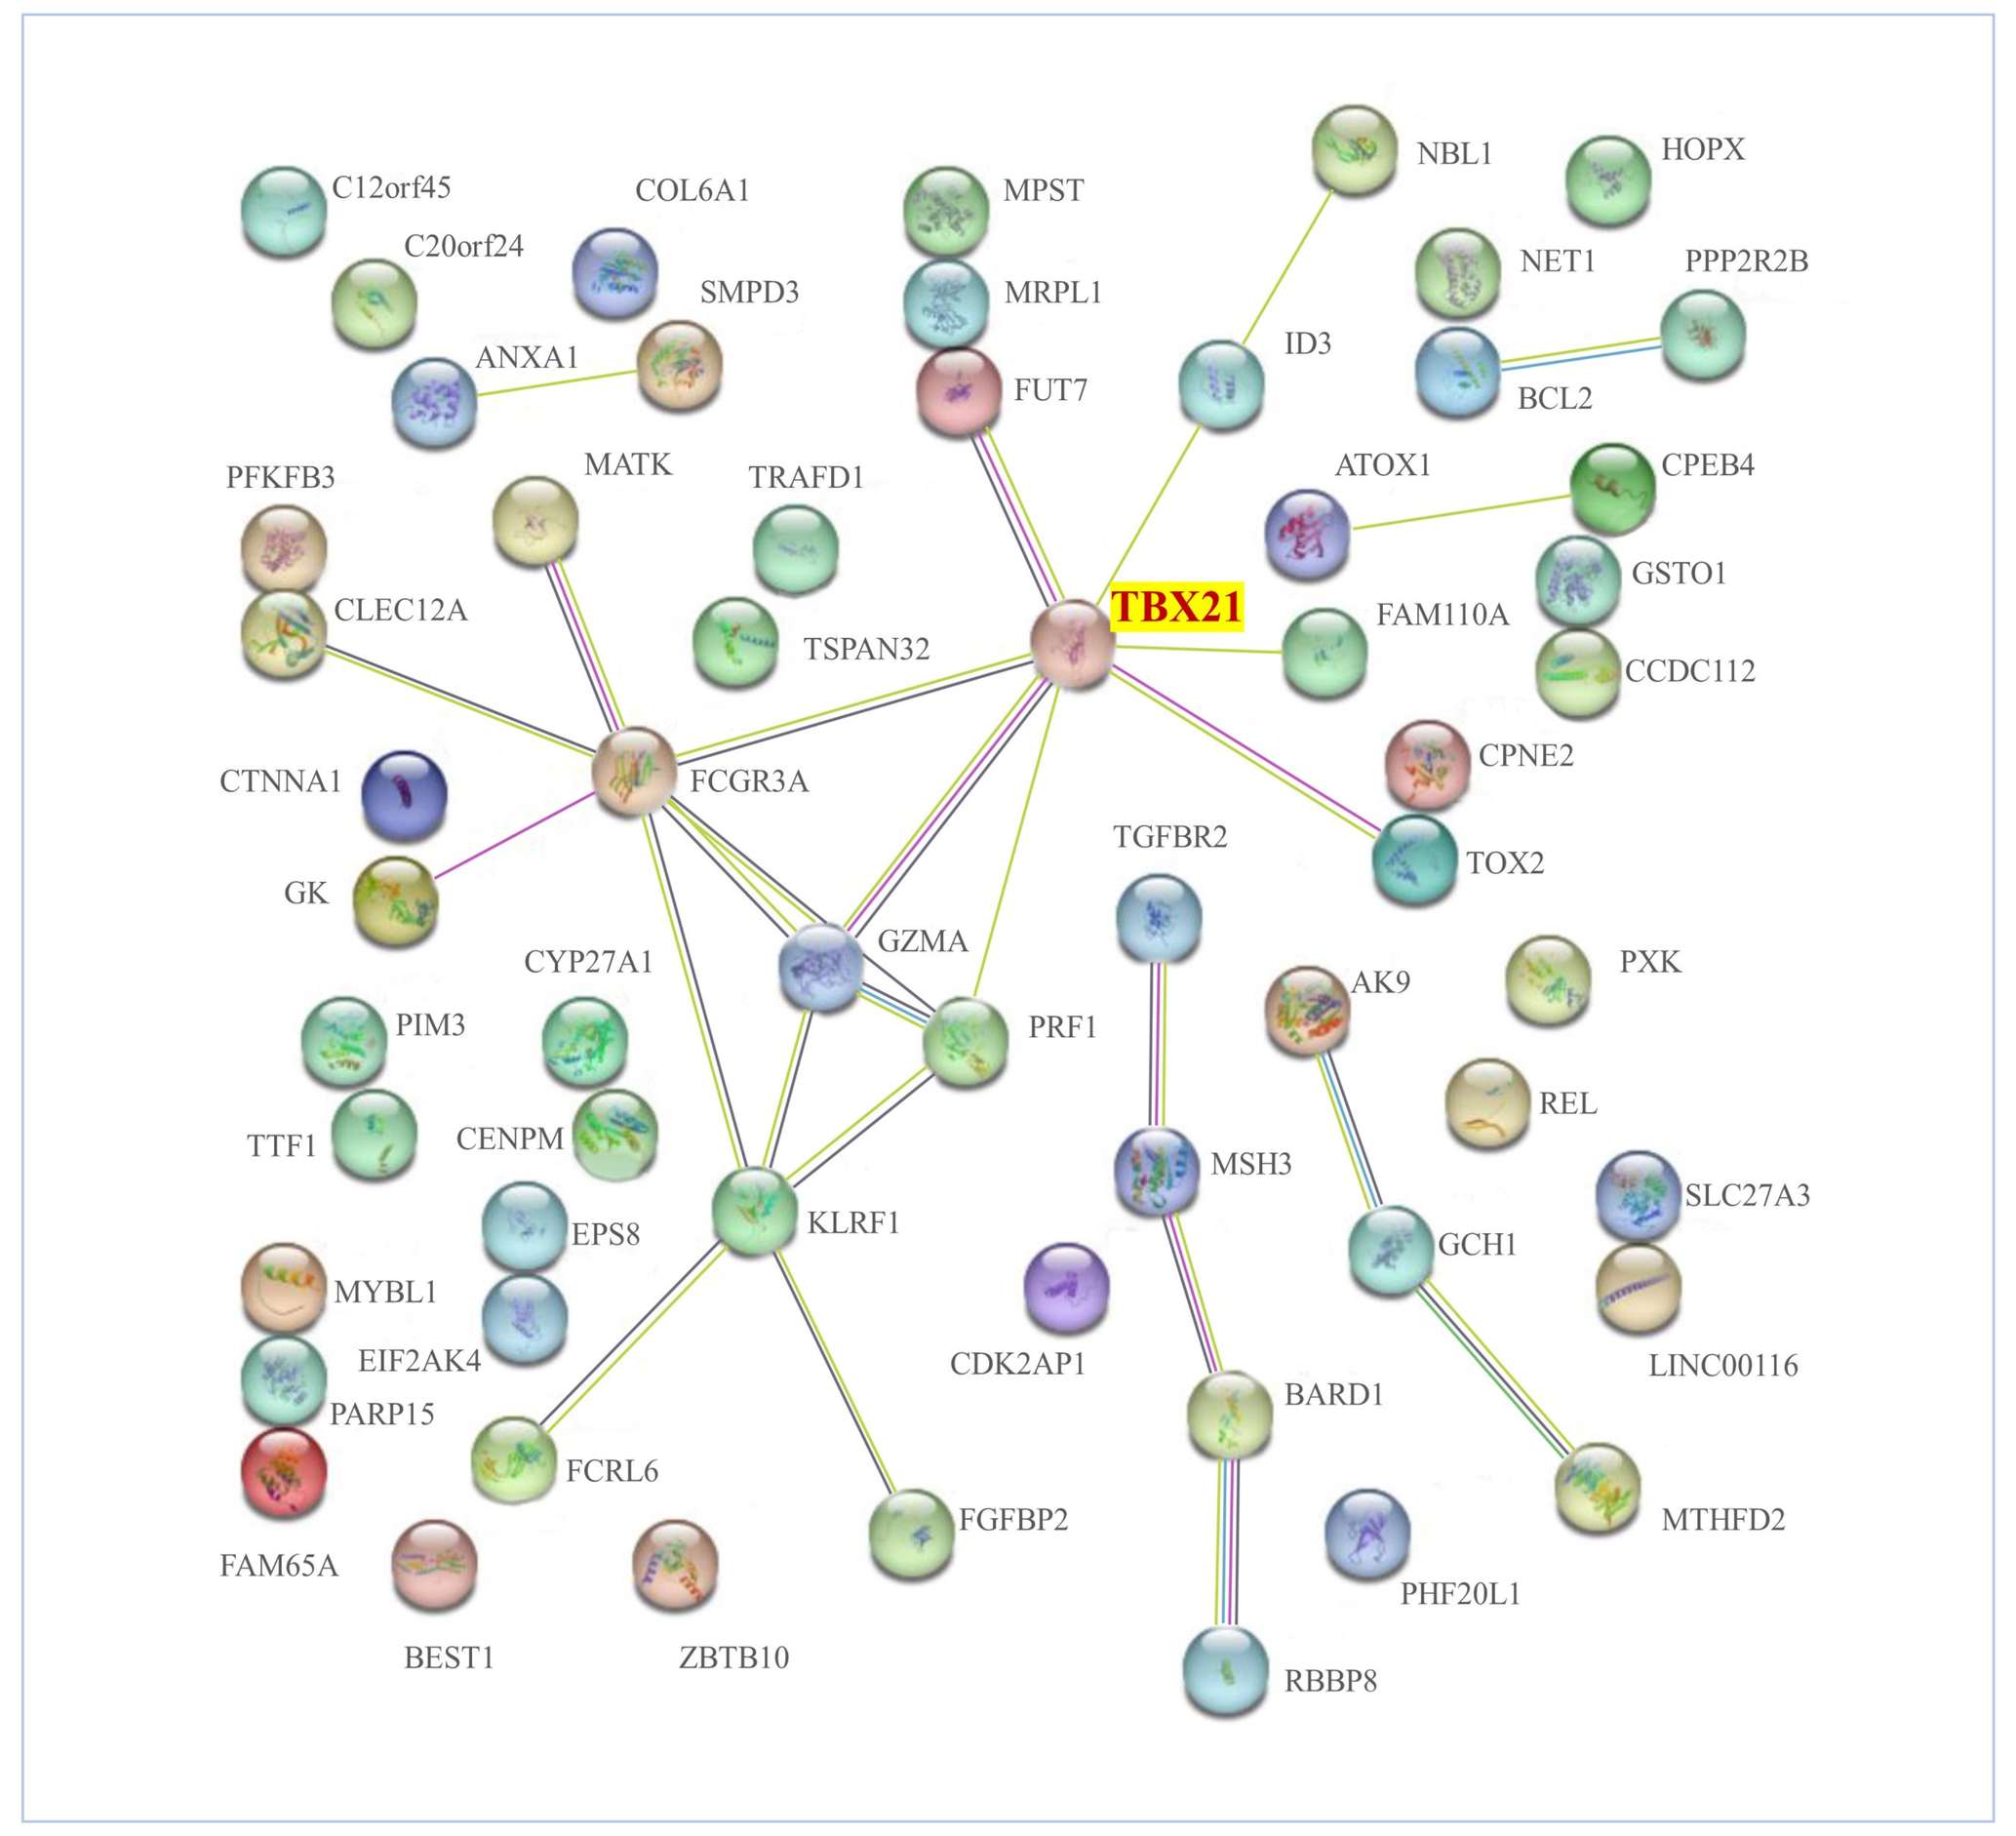
TBX21
C12orf45
C20orf24
COL6A1
SMPD3
ANXA1
MPST
MRPL1
FUT7
NBL1
HOPX
NET1
PPP2R2B
ID3
BCL2
PFKFB3
MATK
TRAFD1
ATOX1
CPEB4
GSTO1
CLEC12A
TSPAN32
FAM110A
CCDC112
CTNNA1
FCGR3A
CPNE2
TGFBR2
TOX2
GK
GZMA
CYP27A1
PIM3
PRF1
AK9
PXK
REL
TTF1
CENPM
MSH3
SLC27A3
EPS8
KLRF1
GCH1
MYBL1
EIF2AK4
PARP15
CDK2AP1
LINC00116
BARD1
FCRL6
FAM65A
FGFBP2
MTHFD2
PHF20L1
BEST1
ZBTB10
RBBP8

Figure 6. Single-cell sequencing studies further confirm that soquelitinib may primarily regulate TBX21 expression in tumor-infiltrating T cells, CD8+ TEMRA cell differentiation, and activation through the modulation of the tumor microenvironment.
Tumor tissues before and after treatment with soquelitinib were collected at different time points and assayed by single-cell RNA-seq. The upregulated and downregulated gene sets were enriched and the selected genes were constructed for protein-protein interaction networks.

SQL induced tumor infiltrated immune cells contribute to the anti-tumor effects of ITK inhibitor

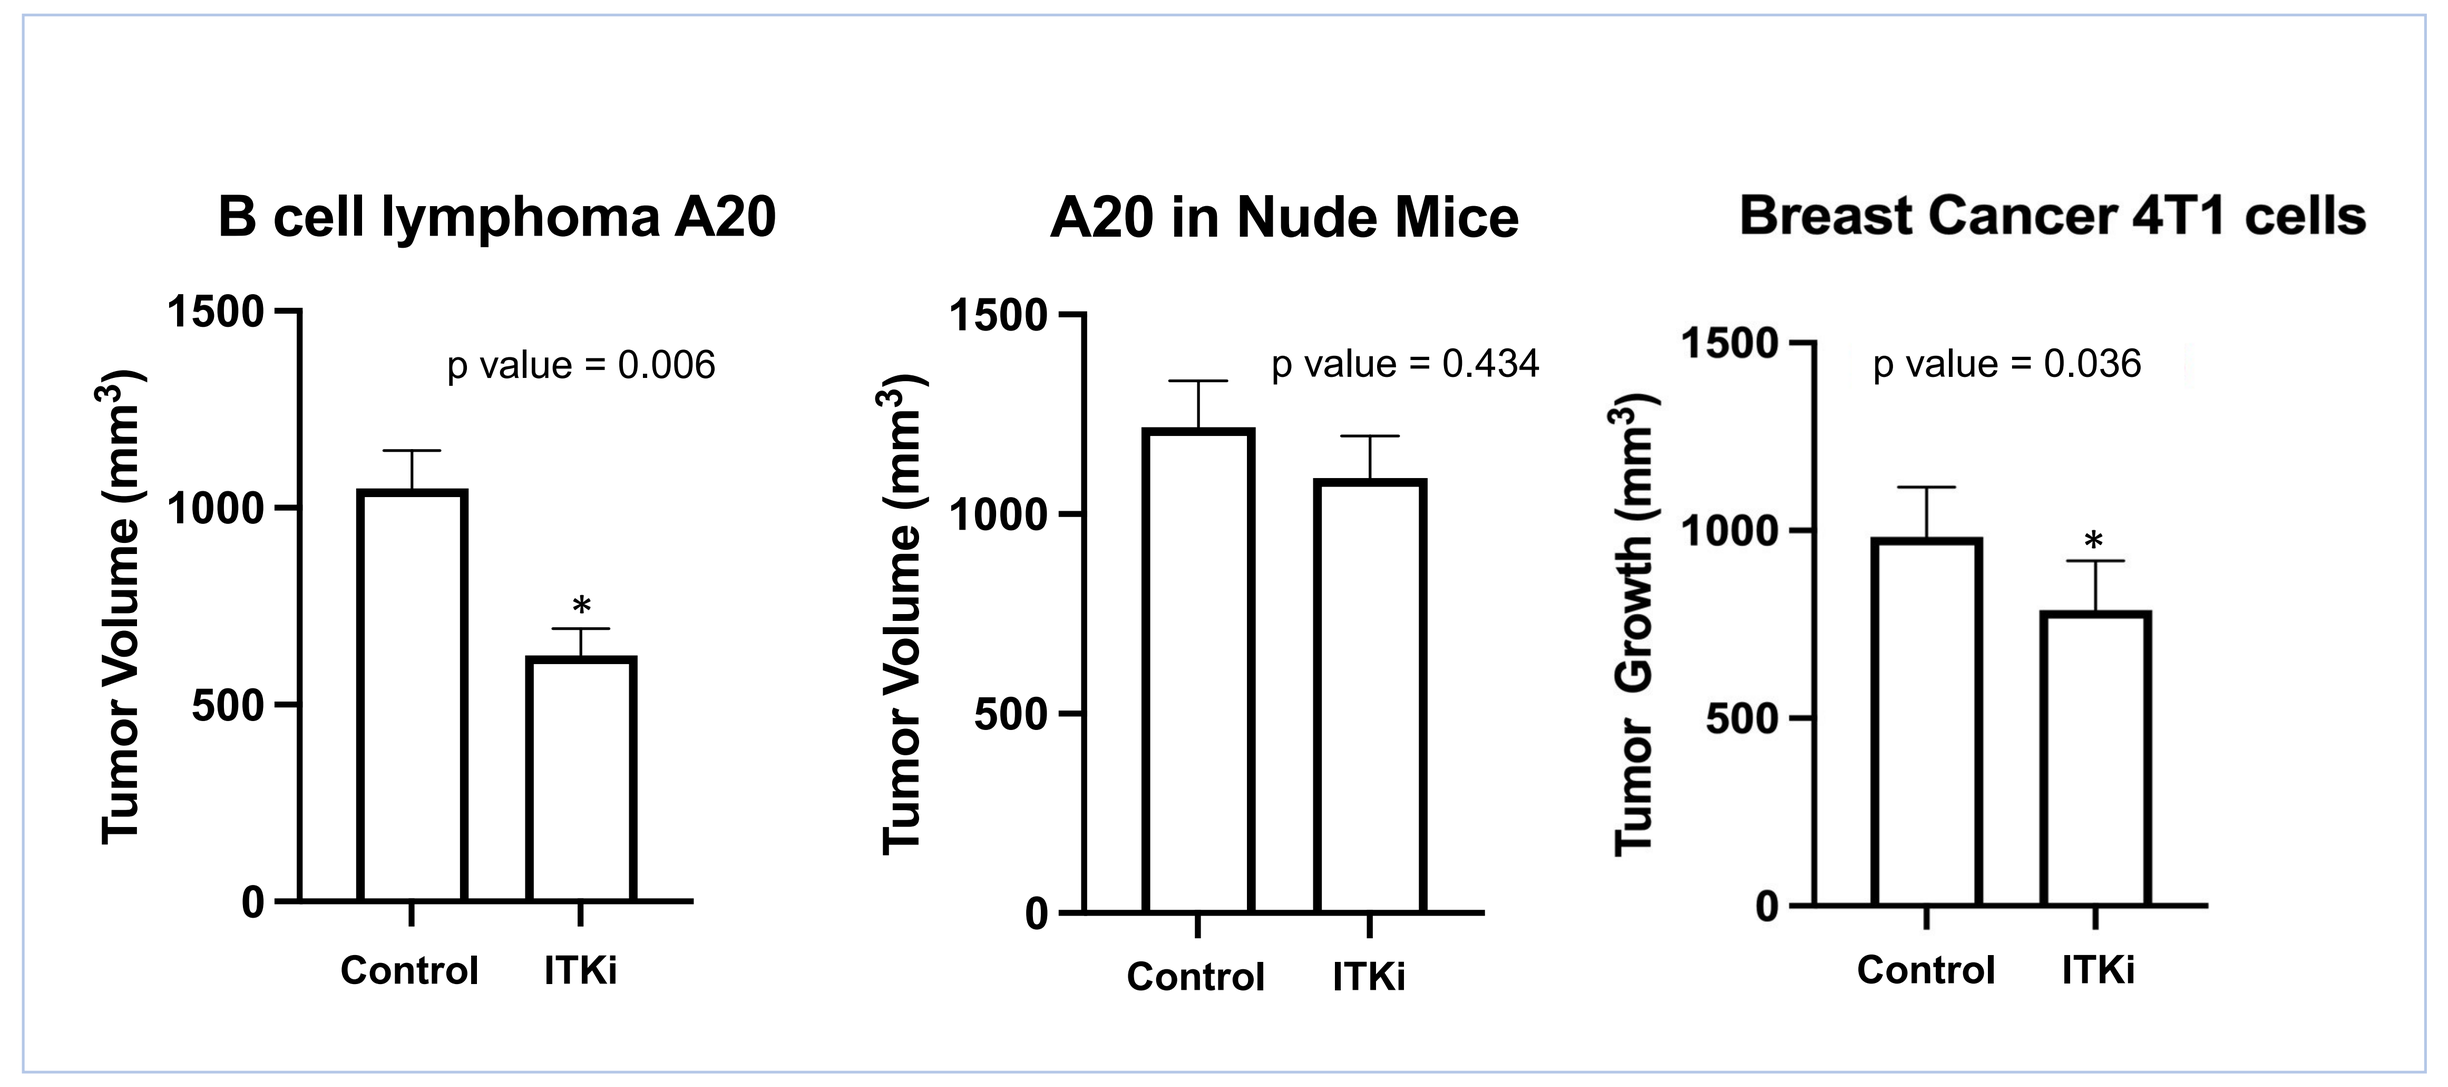
B cell lymphoma A20
A20 in Nude Mice
Breast Cancer 4T1 cells
Tumor Volume (mm3)
Tumor Growth (mm3)
p value = 0.006
p value = 0.434
p value = 0.036
Control
ITKi

Figure 7. Human T cell in vitro analysis and TCL tumor bearing mouse model studies confirmed the immunomodulatory effect of soquelitinib.
Similar results were also observed in the murine syngeneic B cell lymphoma A20, breast cancer 4T1 models. Mice inoculated A20 or 4T1 tumors were treated with ITK inhibitor or vehicle daily for 10 days. Tumor measurements were performed after last dosing. Soquelitinib was not effective in nude mouse model indicating that competent host immune system is required for response.

5. CONCLUSIONS

Our findings reveal a novel immune regulatory mechanism by which the ITK inhibitor soquelitinib induced normal CD4+ Th1 cells and CD8+ TEMRA cells and reduced CD4+ Treg cells in the tumor micro-environment in responding patients. These findings demonstrate the potential of soquelitinib as a novel immunotherapy for the treatment of T-cell lymphomas and solid tumors.

CONTACT INFORMATION: songyuqin622@163.com

American Society of Hematology
Helping hematologists conquer blood diseases worldwide