Selective A2AR Blockade Synergizes with Immune Checkpoint Therapy by Potentiating Proinflammatory Th1 Helper Cell Responses

Dan Li, Poorva Ghosh, Lih-Yun Hsu, Richard A. Miller
Corvus Pharmaceuticals, Inc., Burlingame, California, United States

BACKGROUND

Adenosine Signaling Through A2AR Limits the Efficacy of Immunotherapy

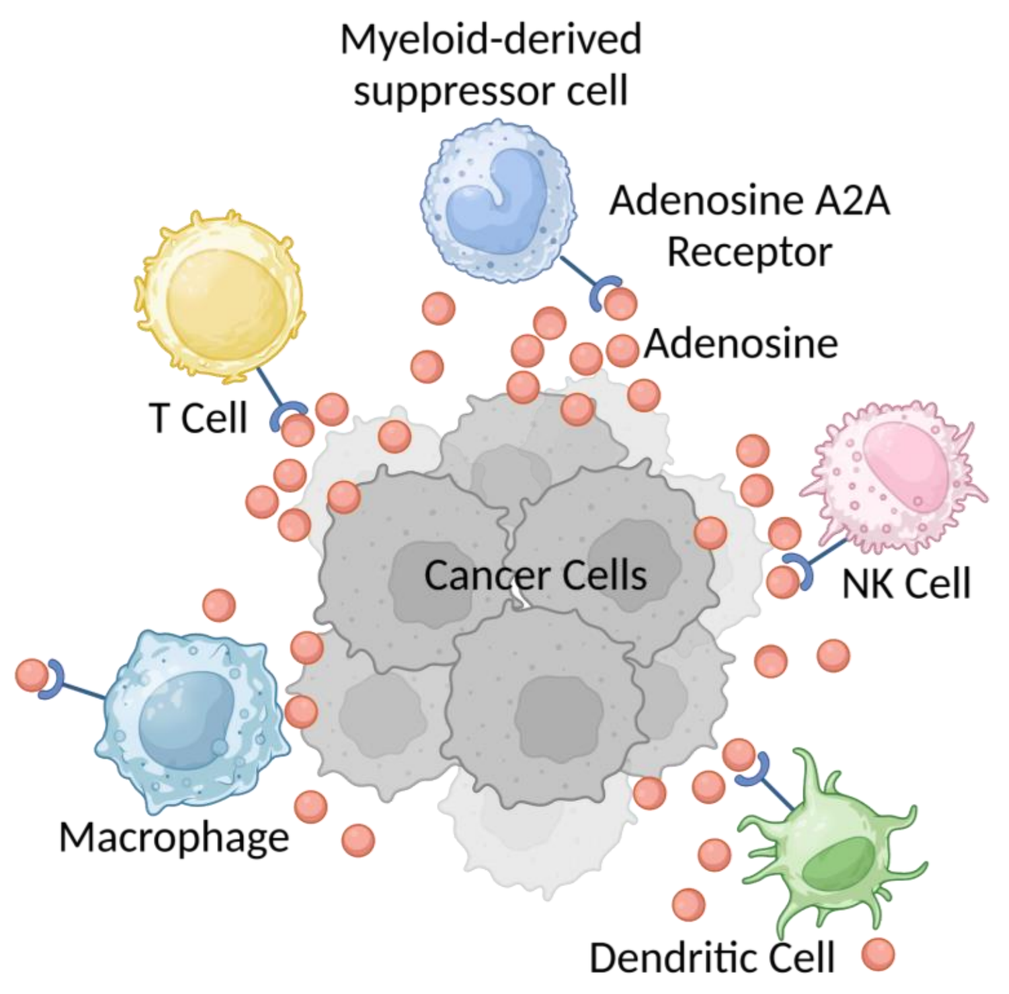
Myeloid-derived suppressor cell
Adenosine A2A Receptor
Adenosine
T Cell
Cancer Cells
NK Cell
Macrophage
Dendritic Cell

Ciforadenant Augments the Efficacy to Immune Checkpoint Blockade (ICB)

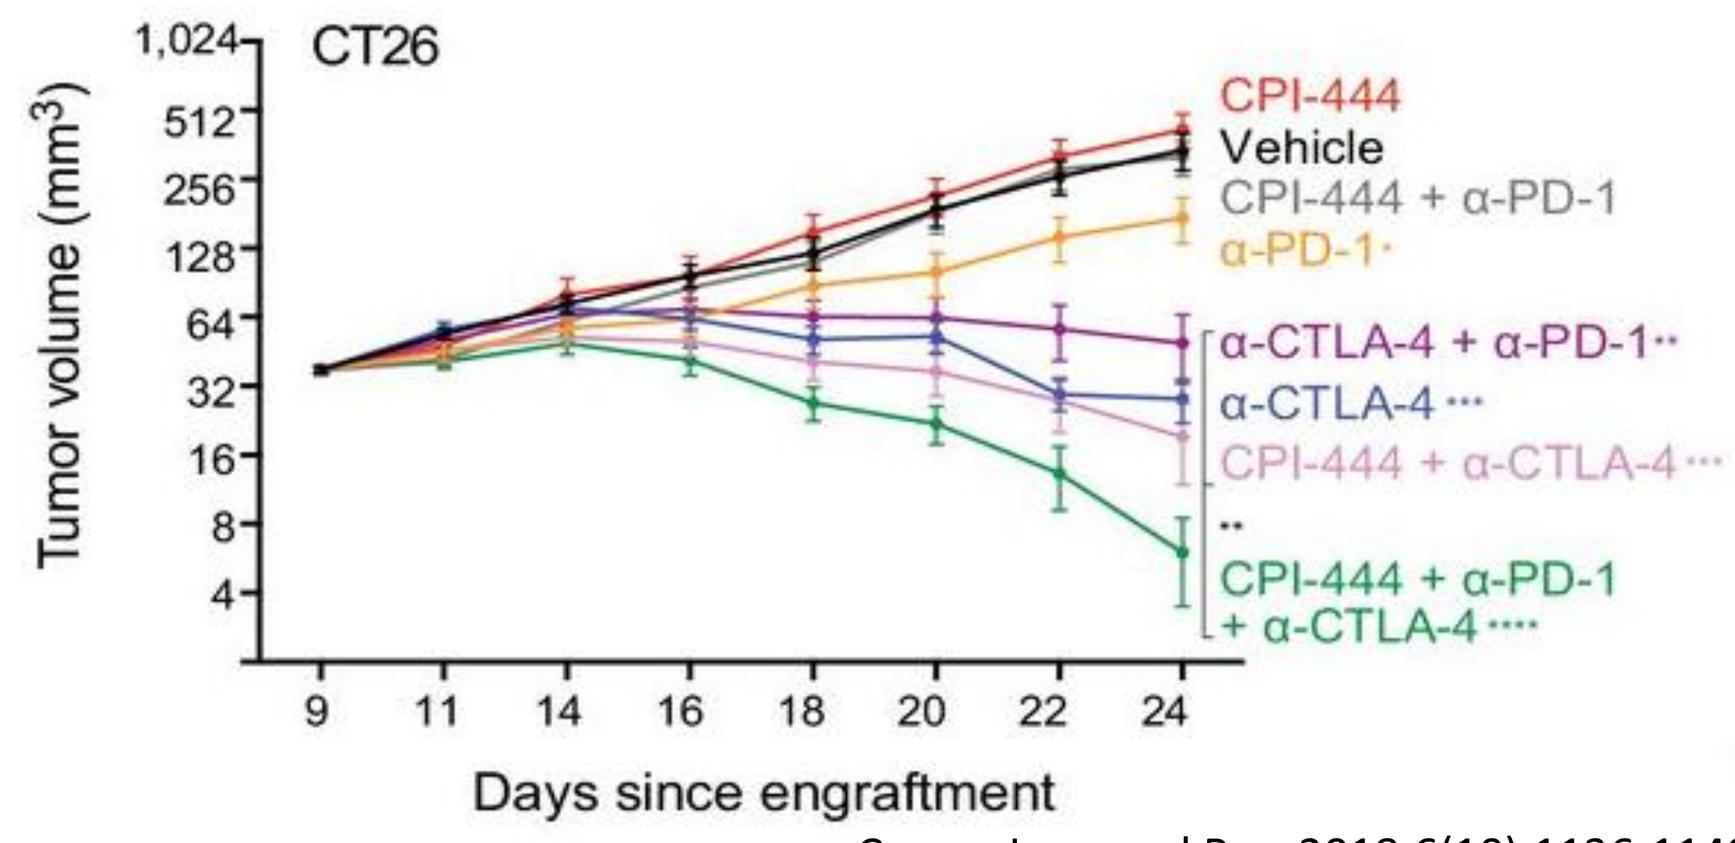
CT26
Tumor volume (mm3)
Days since engraftment
CPI-444
Vehicle
CPI-444 + α-PD-1
α-PD-1
α-CTLA-4 + α-PD-1
α-CTLA-4
CPI-444 + α-CTLA-4
CPI-444 + α-PD-1 + α-CTLA-4

Cancer Immunol Res. 2018;6(10):1136-1149.

Adenosine Signature in Pretreatment Biopsies Predicts Clinical Response to Ciforadenant

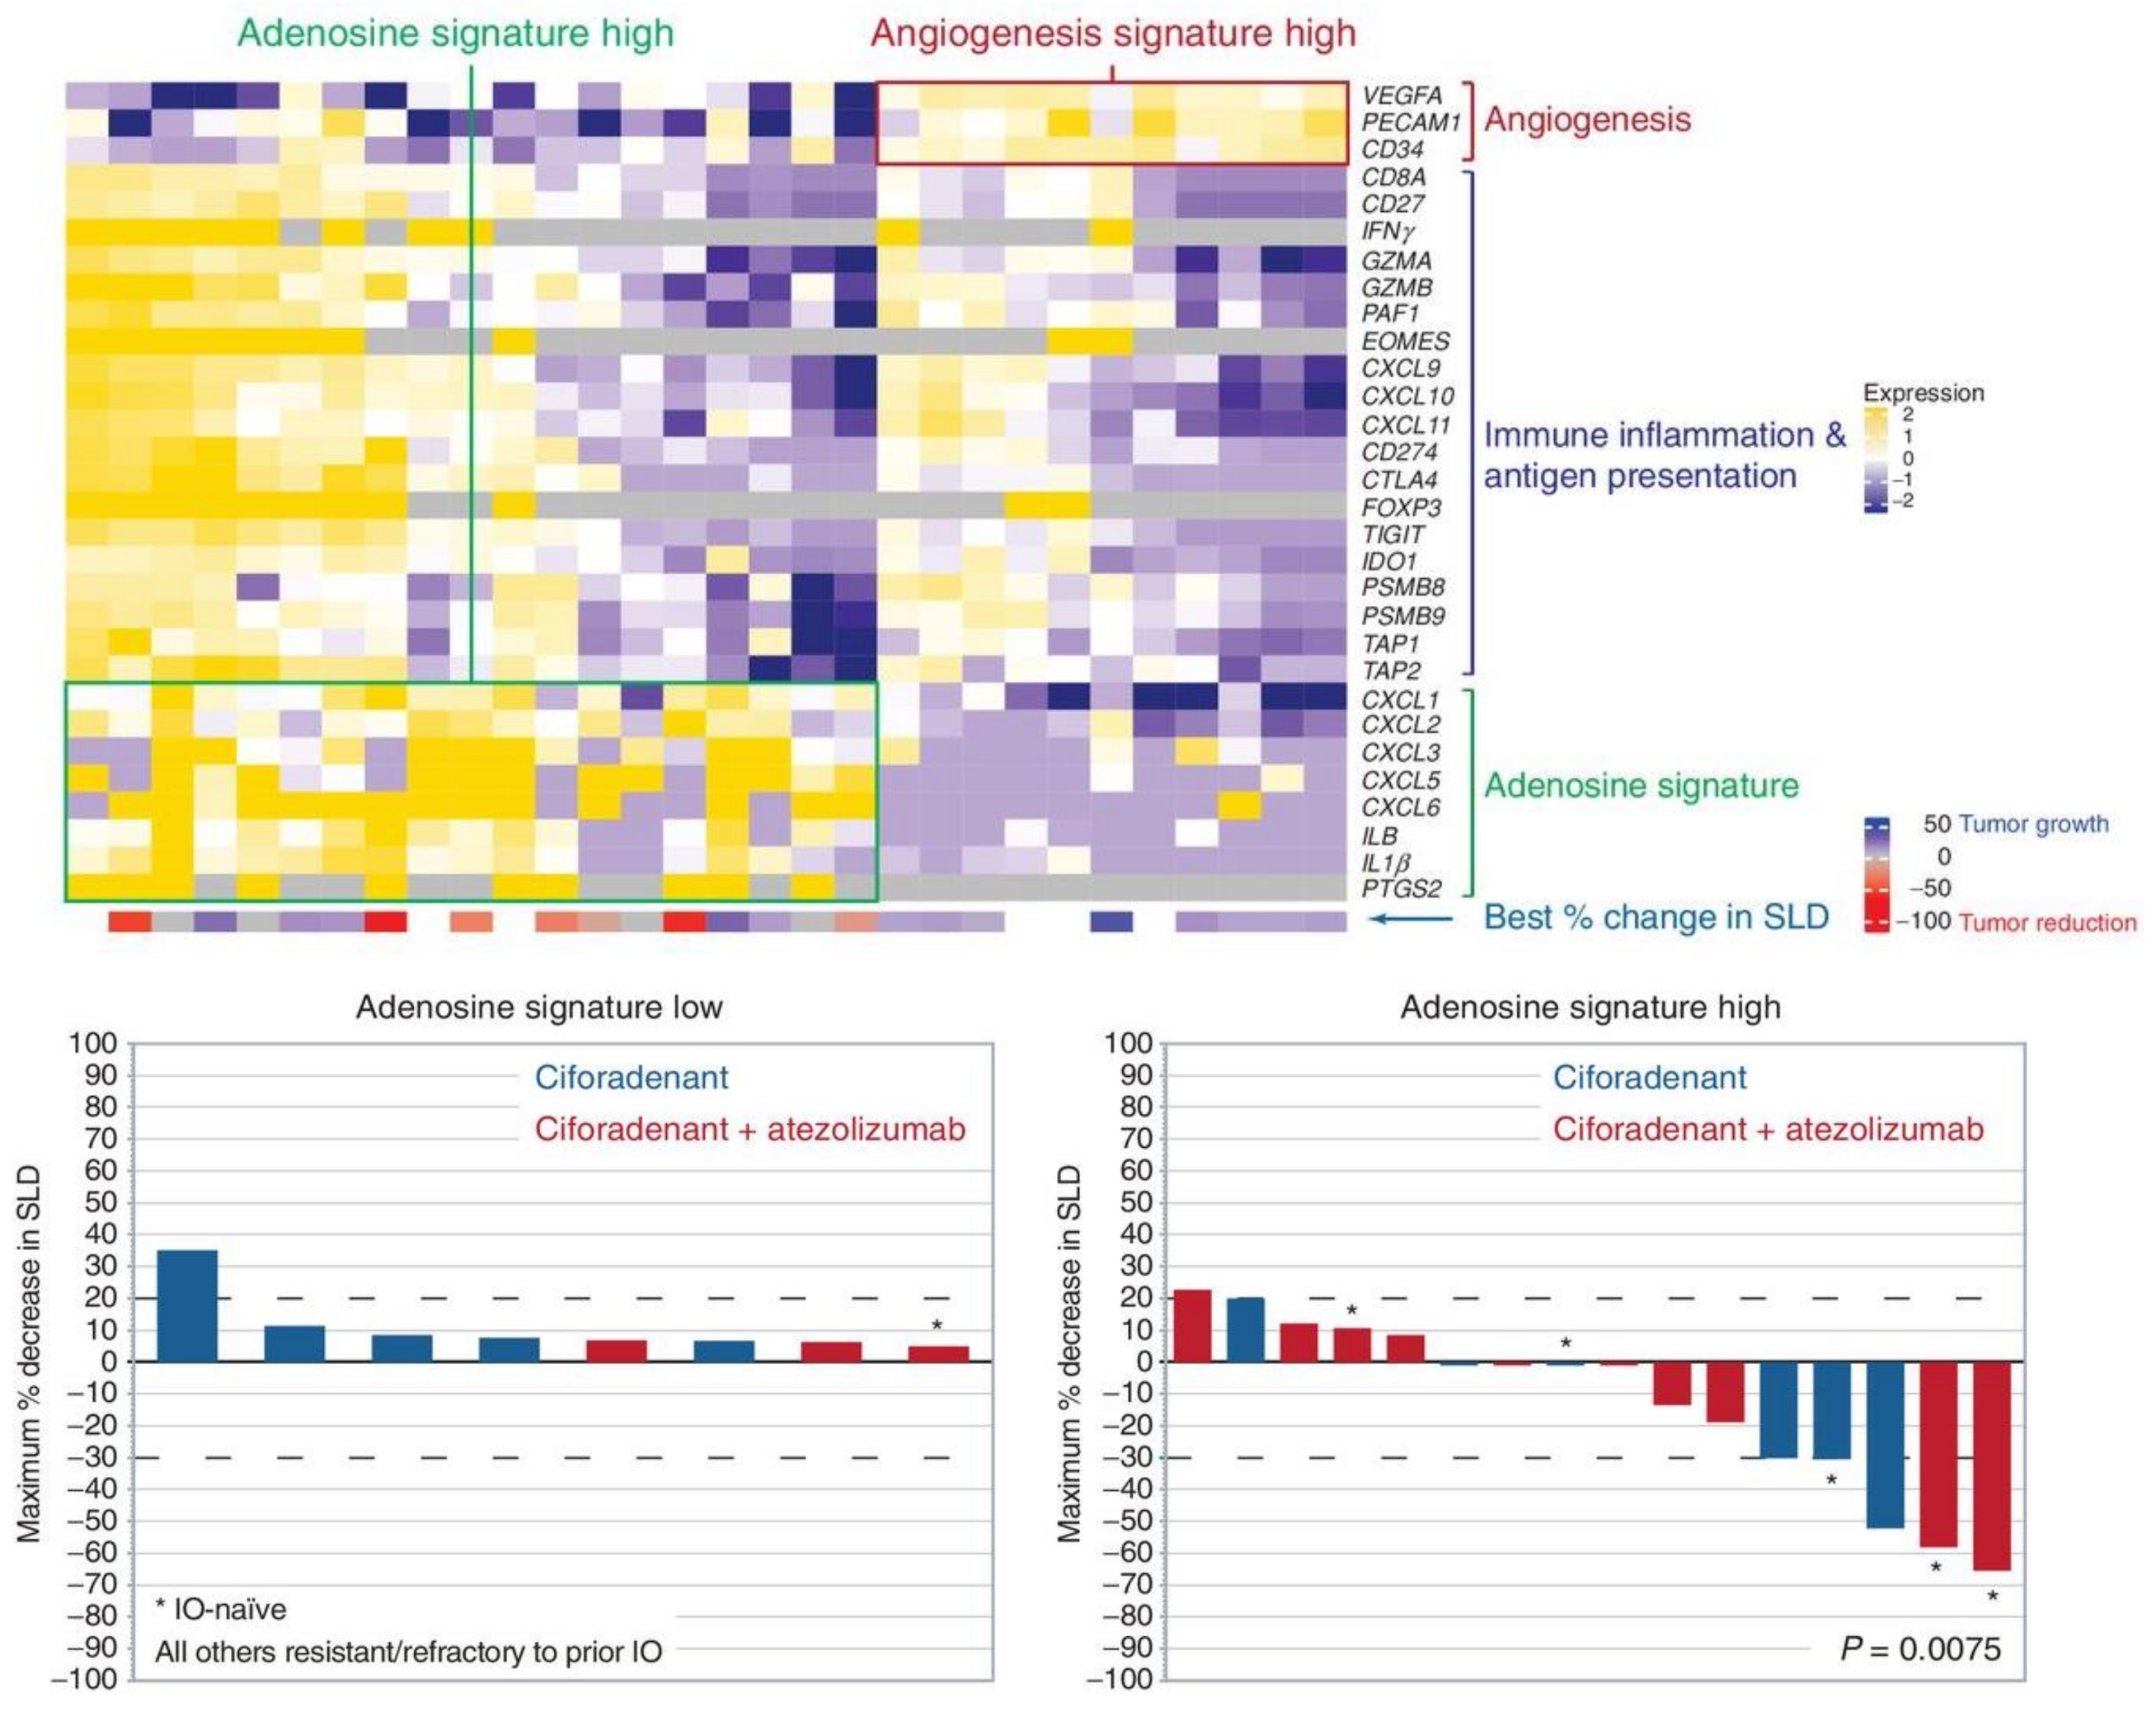
Adenosine signature high
Angiogenesis signature high
Angiogenesis
Immune inflammation & antigen presentation
Adenosine signature
Best % change in SLD
Expression
50 Tumor growth
−100 Tumor reduction
Adenosine signature low
Adenosine signature high
Ciforadenant
Ciforadenant + atezolizumab
Maximum % decrease in SLD
* IO-naïve
All others resistant/refractory to prior IO
P = 0.0075

Cancer Discov. 2020;10(1):40-53.

MATERIALS AND METHODS

Syngeneic murine tumor models: CT26 (colorectal carcinoma) and B16F10-OVA (melanoma) were subcutaneously implanted into Balb/c and C57BL/6 mice, respectively. Caliper measurement was performed by a digital caliper (Flwer-Sylvac). Tumor volume was calculated by the formula (length x width^2)/2. Mice with established tumors (~50 mm^3) were treated with anti-PD1 (clone RPM1-14, 25 µg/mouse) and anti-CTLA4 (clone 9H10, 25 µg/mouse) and Ciforadenant (30 mg/kg).
Gene profile analysis: Tumor treated with Ciforadenant alone by oral gavage or in combination with anti-PD1 and anti-CTLA4 were harvested and stored in RNAlater Solution (ThermoFisher) at 4°C. RNA was extracted using Rneasy Mini Kit (Qiagen). Nanostring mouse Myeloid Panel along with a custom codeset consisting of probes to genes were used on nCounter Sprint Profiler (NanoString). Data were analyzed by nSolver Analysis Software.
Analysis of surface and intracellular marker in tumor: Single-cell suspension of tumors were prepared by tumor dissociation kit and gentleMAC dissociator (Miltenyi Biotec). Cell surface markers were stained for 20-30 min in 4°C, and then fixed and permeabilized using the Foxp3 Fix/Perm buffer set (ThermoFisher). Intracellular proteins or cytokines were stained after permeabilization. Flow cytometry was performed on the Cytoflex cytometer (Beckman Coulter) and data were analyzed using FlowJo software (TreeStar).
Measurement of intratumoral cytokines/chemokines: 50 mg of tumors were resuspended with PBS and 1% Triton and 1% protease inhibitor cocktail (Sigma-Aldrich). The tissue was then homogenized with a Bullet Blender Blue (Next Advance). Intratumoral cytokines were measured by MSD (Meso Scale Diagnostics). The cytokine concentrations were normalized to protein concentration of the same tumor lysates.

RESULTS

Depletion of Myeloid Cells Abolishes Synergy of Ciforadenant & ICB in Murine B16 Ova Melanoma Model

Study Design

Day 0 — Tumor engraftment
Day 8 — Randomization
Day 9 — Depletion antibodies begin
Day 10 to Day 15 — Depletion antibodies, ICB & Ciforadenant
Day 17 — End Point

Tumor Growth Curve

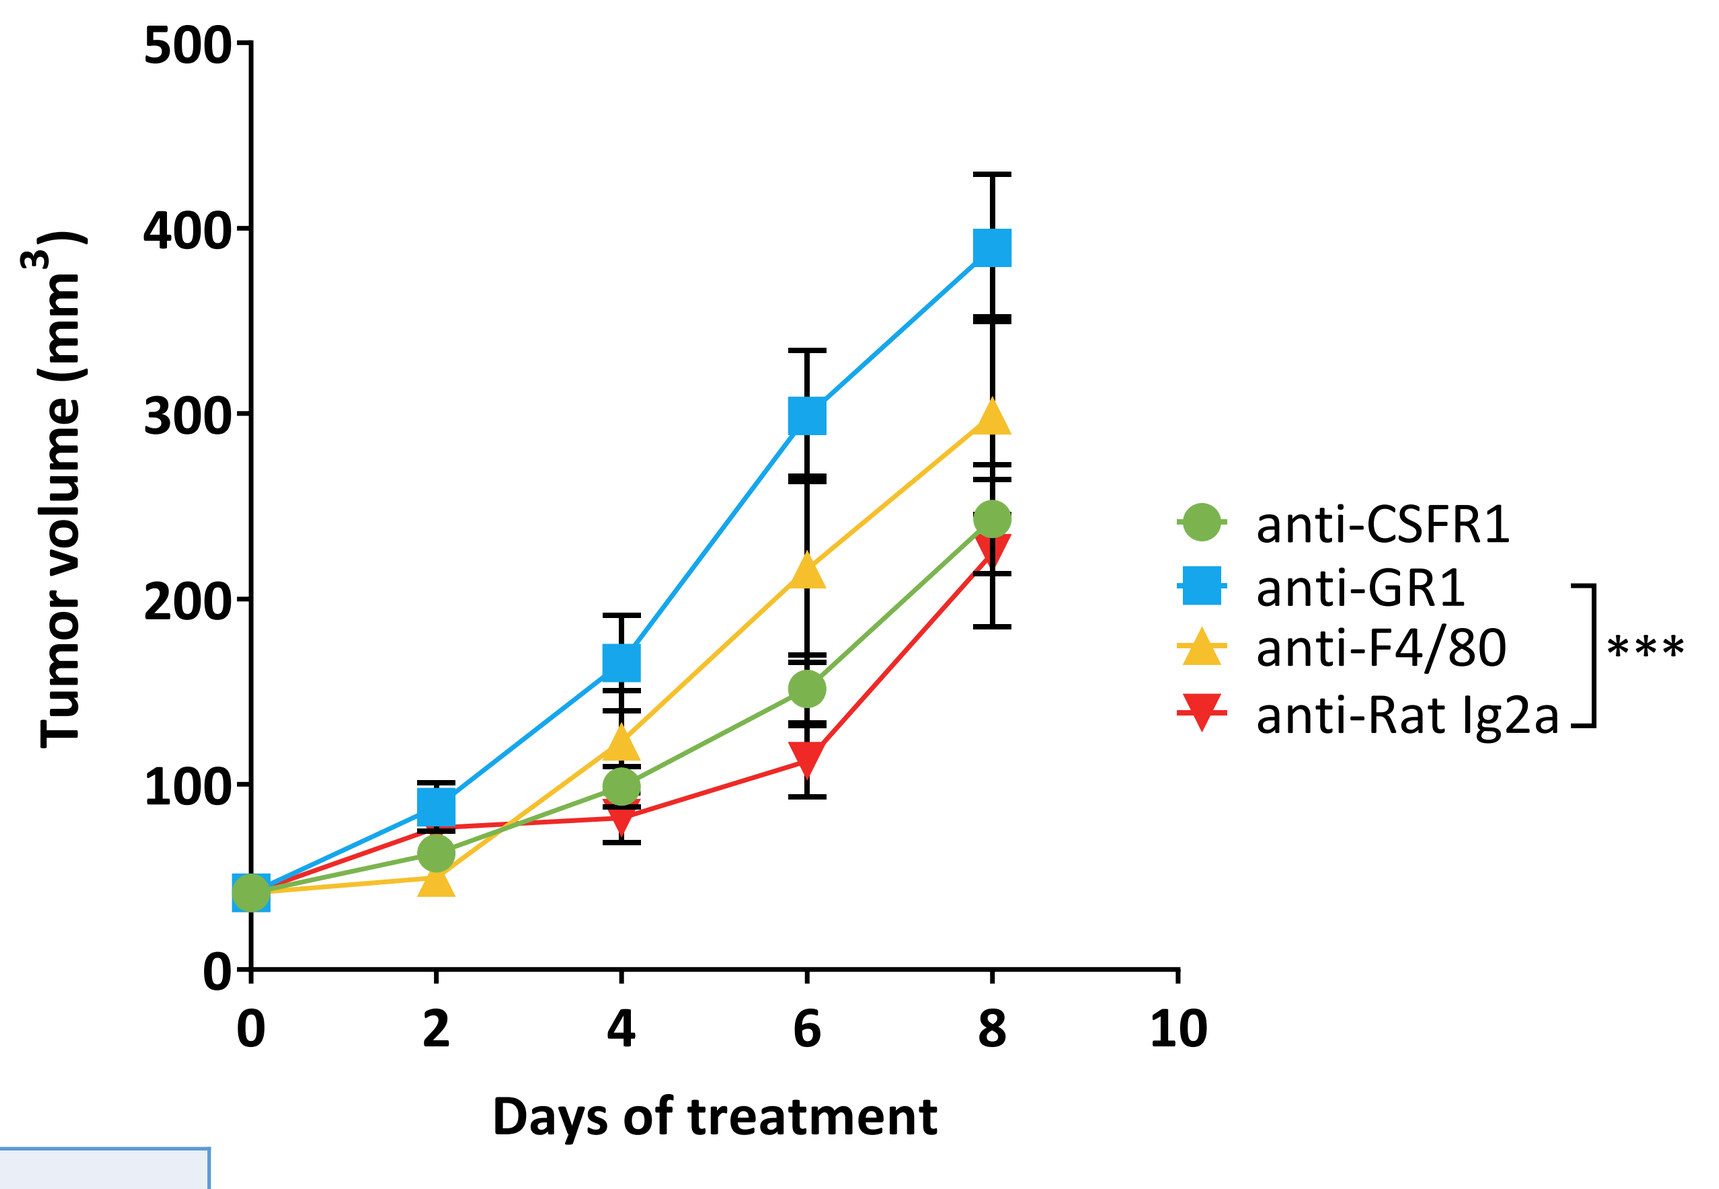
Tumor volume (mm3)
Days of treatment
anti-CSFR1
anti-GR1
anti-F4/80
anti-Rat Ig2a

Materials

ICB+Ciforadenant				
Depletion antibody	anti-CSFR1	anti-GR1	anti-F4/80	anti-Rat Ig2a
Clone	AFS98	RB6-8C5	CI: A3-1	2A3
Depleting Cell types	TAM & DC	MDSC	M2 TAM	Control Isotype

CSF1R: colony-stimulating factor 1 receptor
TAM: tumor-associated macrophage
DC: dendritic cell
GR1: gamma response 1
MDSC: myeloid-derived suppressor cell
F4/80: adhesion G protein-coupled receptor E1
M2 TAM: M2-polarized tumor-associated macrophage

Figure 1. ***Depletion of MDSC induces tumor progression in mice.*** 1 million of B16F10-Ova tumor cells were engrafted to B6 mouse. Anti-CSFR1 (400 µg/mouse) or anti-GR1 (400 µg/mouse) or anti-F4/80 (200 µg/mouse) or anti-Rat Ig2a (400 µg/mouse) plus ICB, anti-PD1 and anti-CTLA4 and Ciforadenant were injected when the tumor was around 50 mm^3. Data are expressed as mean±SEM, 2-way ANOVA, ***p<0.001.

Combination of Ciforadenant & ICB Upregulates Genes in IL-12 Signaling Axis and Proinflammatory Th1 Helper Cell Responses

Upregulated Genes in IL-12 Signaling Pathway

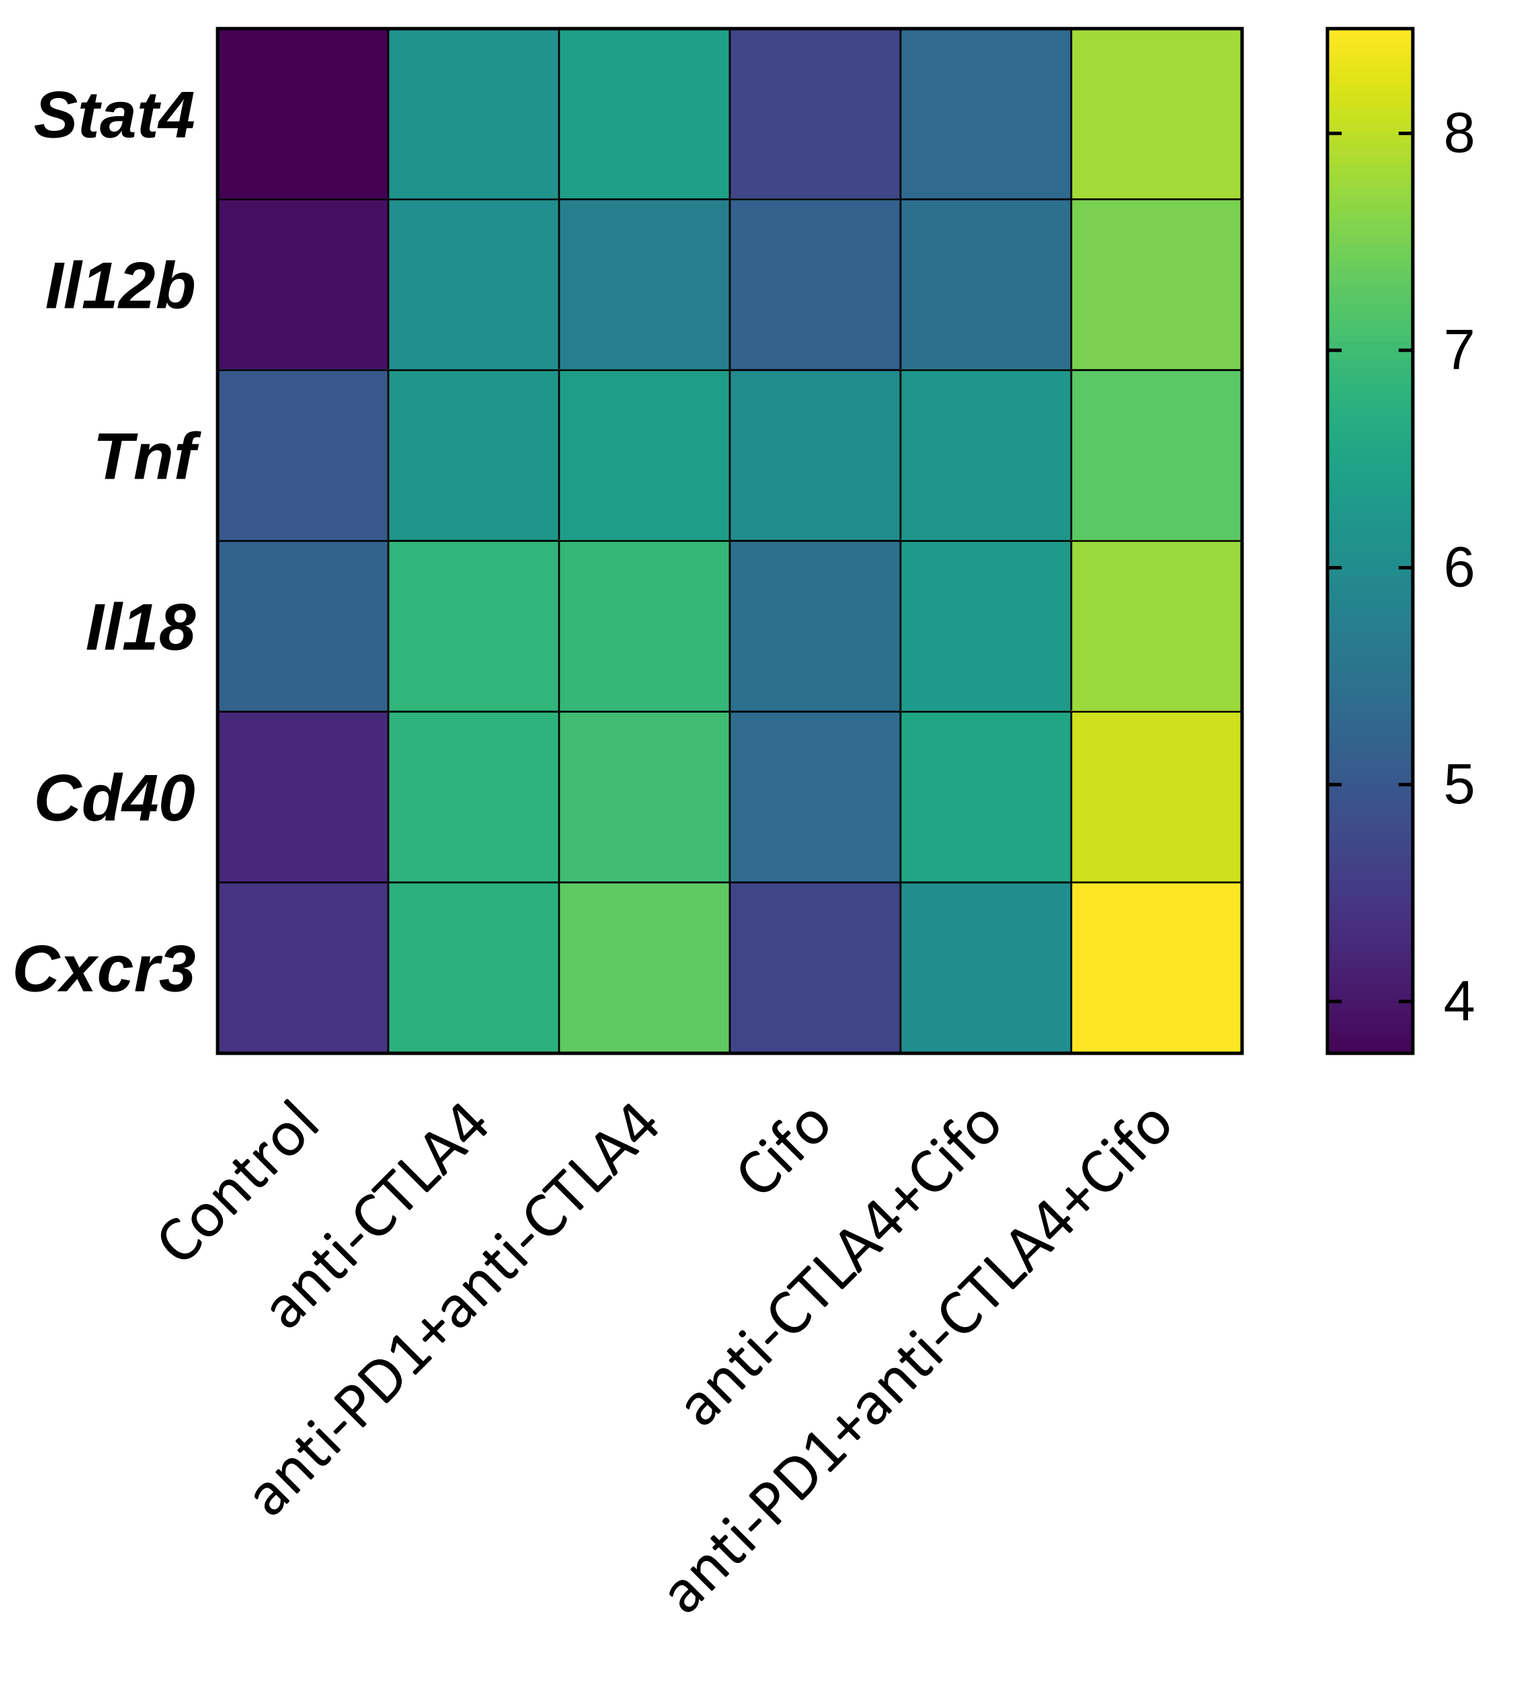
Stat4
Il12b
Tnf
Il18
Cd40
Cxcr3
Control
anti-CTLA4
anti-PD1+anti-CTLA4
Cifo
anti-CTLA4+Cifo
anti-PD1+anti-CTLA4+Cifo

Increased Th1 Helper CD4⁺ Cells in Tumor

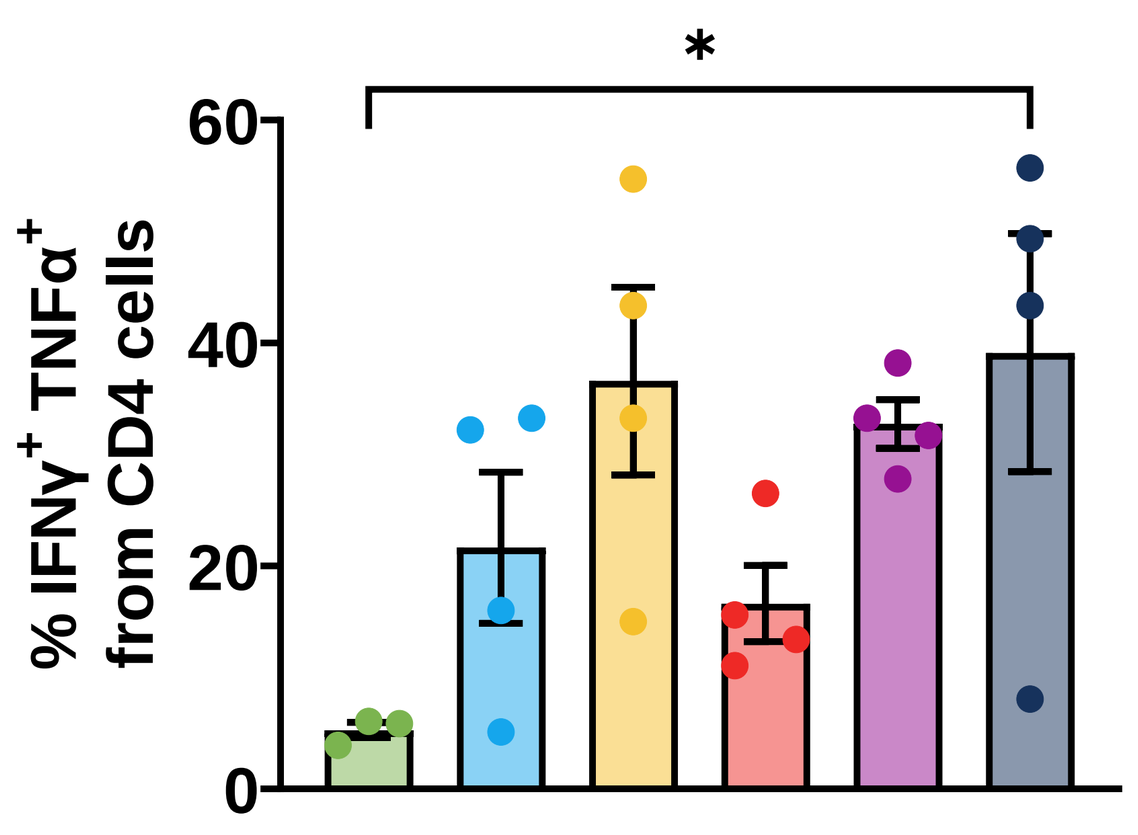
% IFNγ+ TNFα+ from CD4 cells
*

Decreased Exhausted CD8⁺ T Cells in Tumor

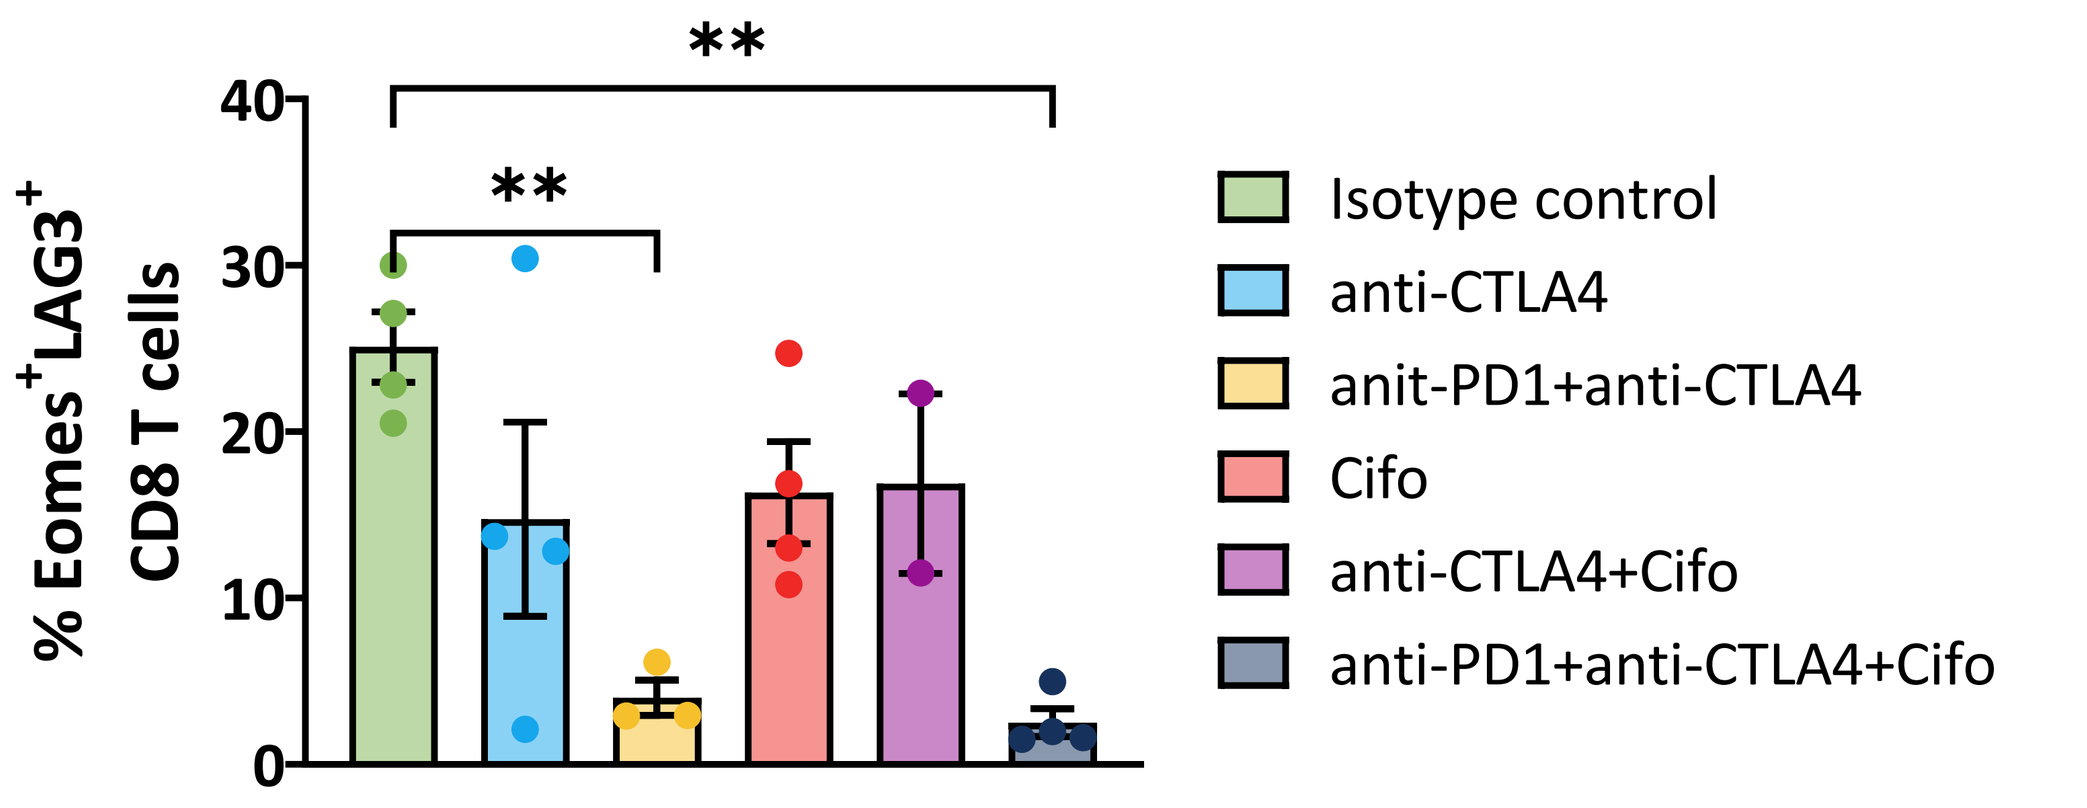
% Eomes+LAG3+ CD8 T cells
**
Isotype control
anti-CTLA4
anit-PD1+anti-CTLA4
Cifo
anti-CTLA4+Cifo
anti-PD1+anti-CTLA4+Cifo

Figure 2. ***Proinflammatory state in the TME of CT26 mouse tumor with Ciforadenant and ICB.*** Genes in IL-12/STAT4 signaling axis in combination of Ciforadenant with ICB in CT26 mouse tumor model were identified using the NanoString mouse myeloid panel and shown with Log2 fold changes. The production of Th1 effector cytokines from CD4⁺ T cells marked by IFNγ and TNFα and exhausted CD8⁺ T cells marked by Eomes (Eomesodermin) and LAG3 (Lymphocyte Activating Gene 3) from tumor were detected by Flow cytometry. Data are expressed as mean±SEM, 1-way ANOVA, *p<0.05, **p<0.01.

Combination of Ciforadenant & ICB Increases CXCL10 for Recruiting Proinflammatory Th1 Helper Cell Responses to Hot and Cold Tumors

CT26 Colorectal Carcinoma

B16F10 OVA-expressing Melanoma

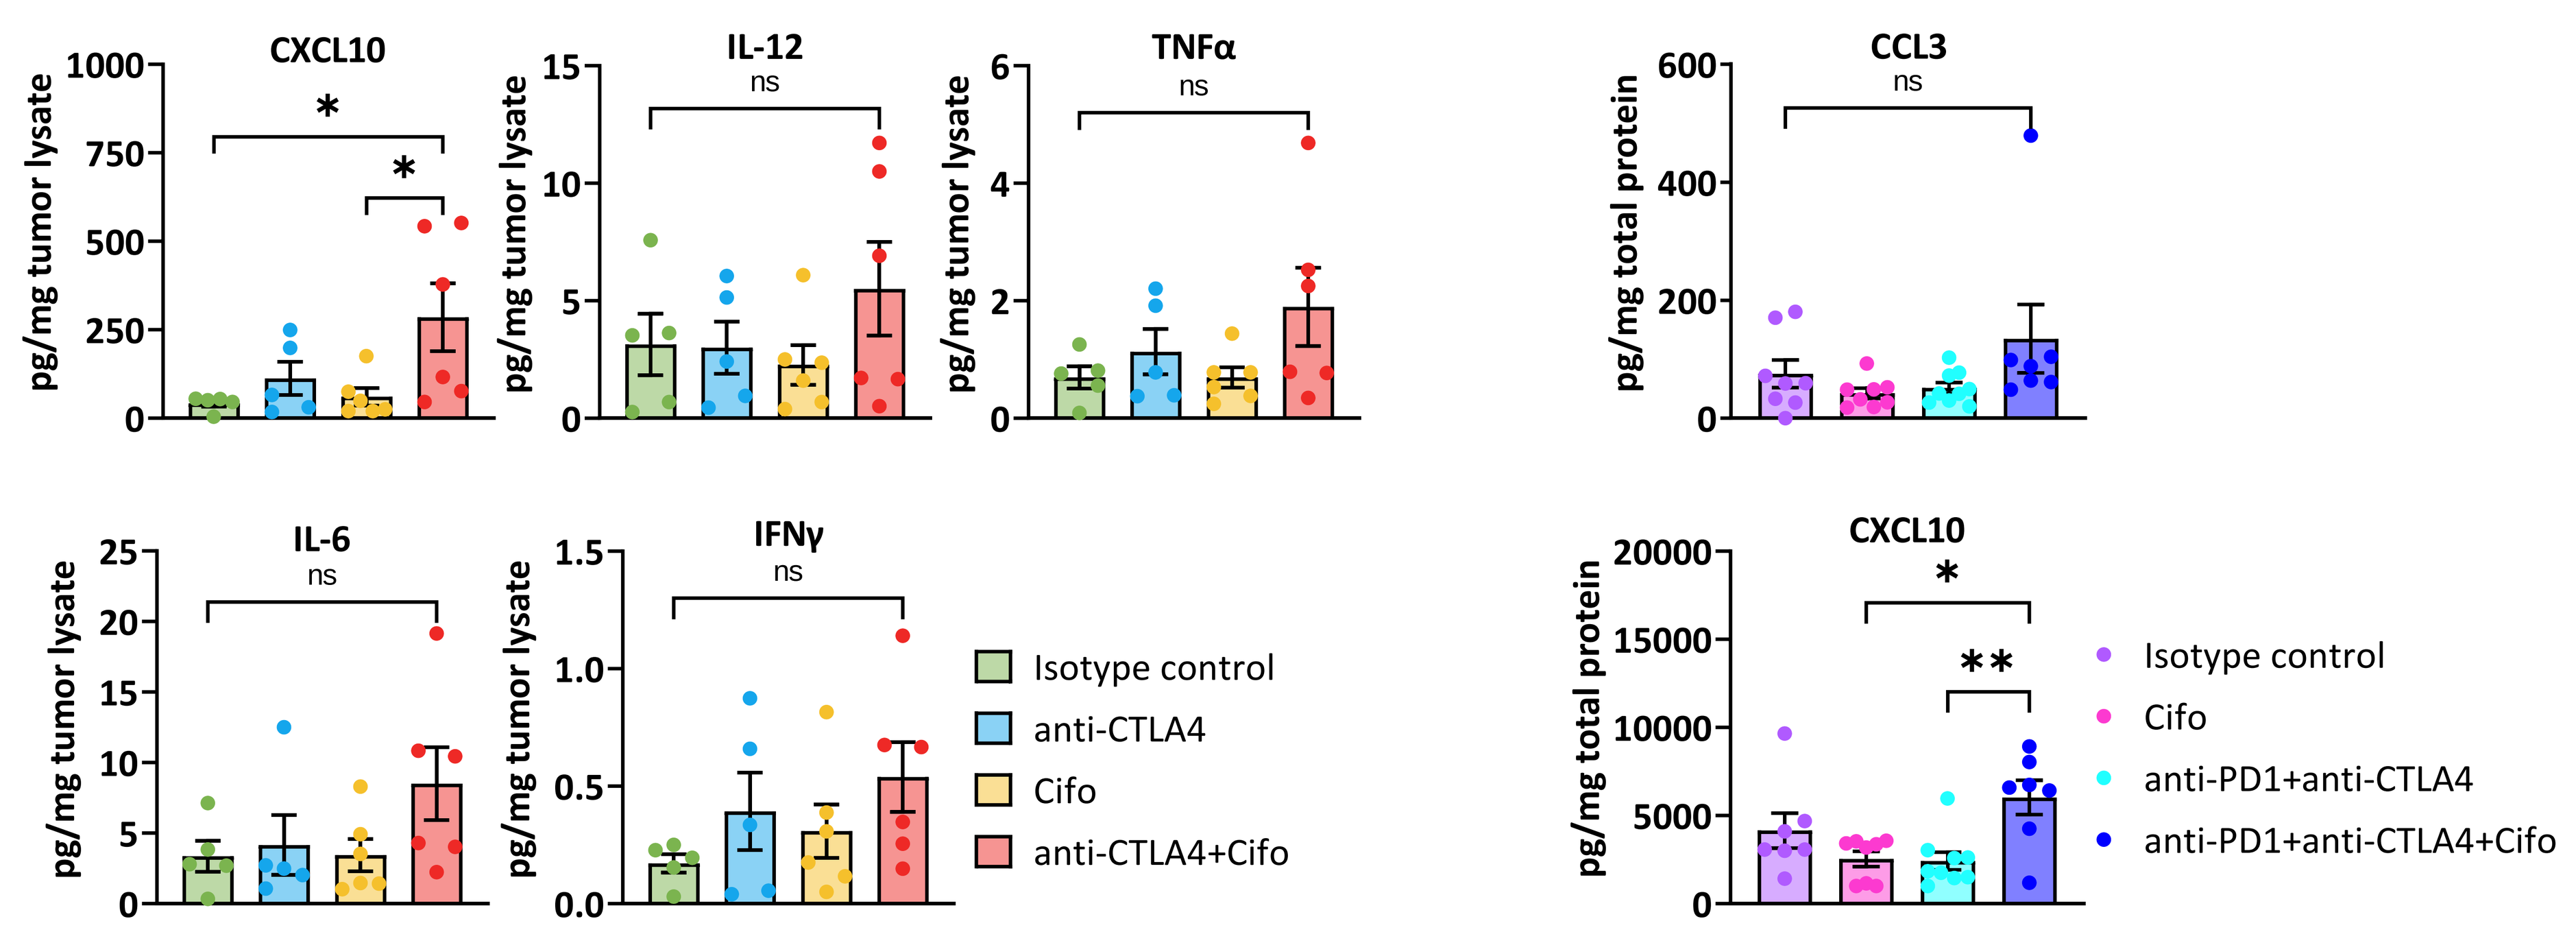
CXCL10
IL-12
TNFα
IL-6
IFNγ
CCL3
pg/mg tumor lysate
pg/mg total protein
ns
Isotype control
anti-CTLA4
Cifo
anti-CTLA4+Cifo
anti-PD1+anti-CTLA4
anti-PD1+anti-CTLA4+Cifo

Figure 3. ***Ciforadenant & ICB increase proinflammatory cytokine production in tumor.*** Ciforadenant with ICB treatment increased production of CXCL10, ligands for recruiting CXCR3+ T cells into the tumor from CT26 and B16F10 OVA detected by MSD. Data are expressed as mean±SEM, 1-way ANOVA, *p<0.05, **p<0.01.

Mechanism of Ciforadenant in Potentiating Proinflammatory Th1 Helper Cell Responses in Tumor

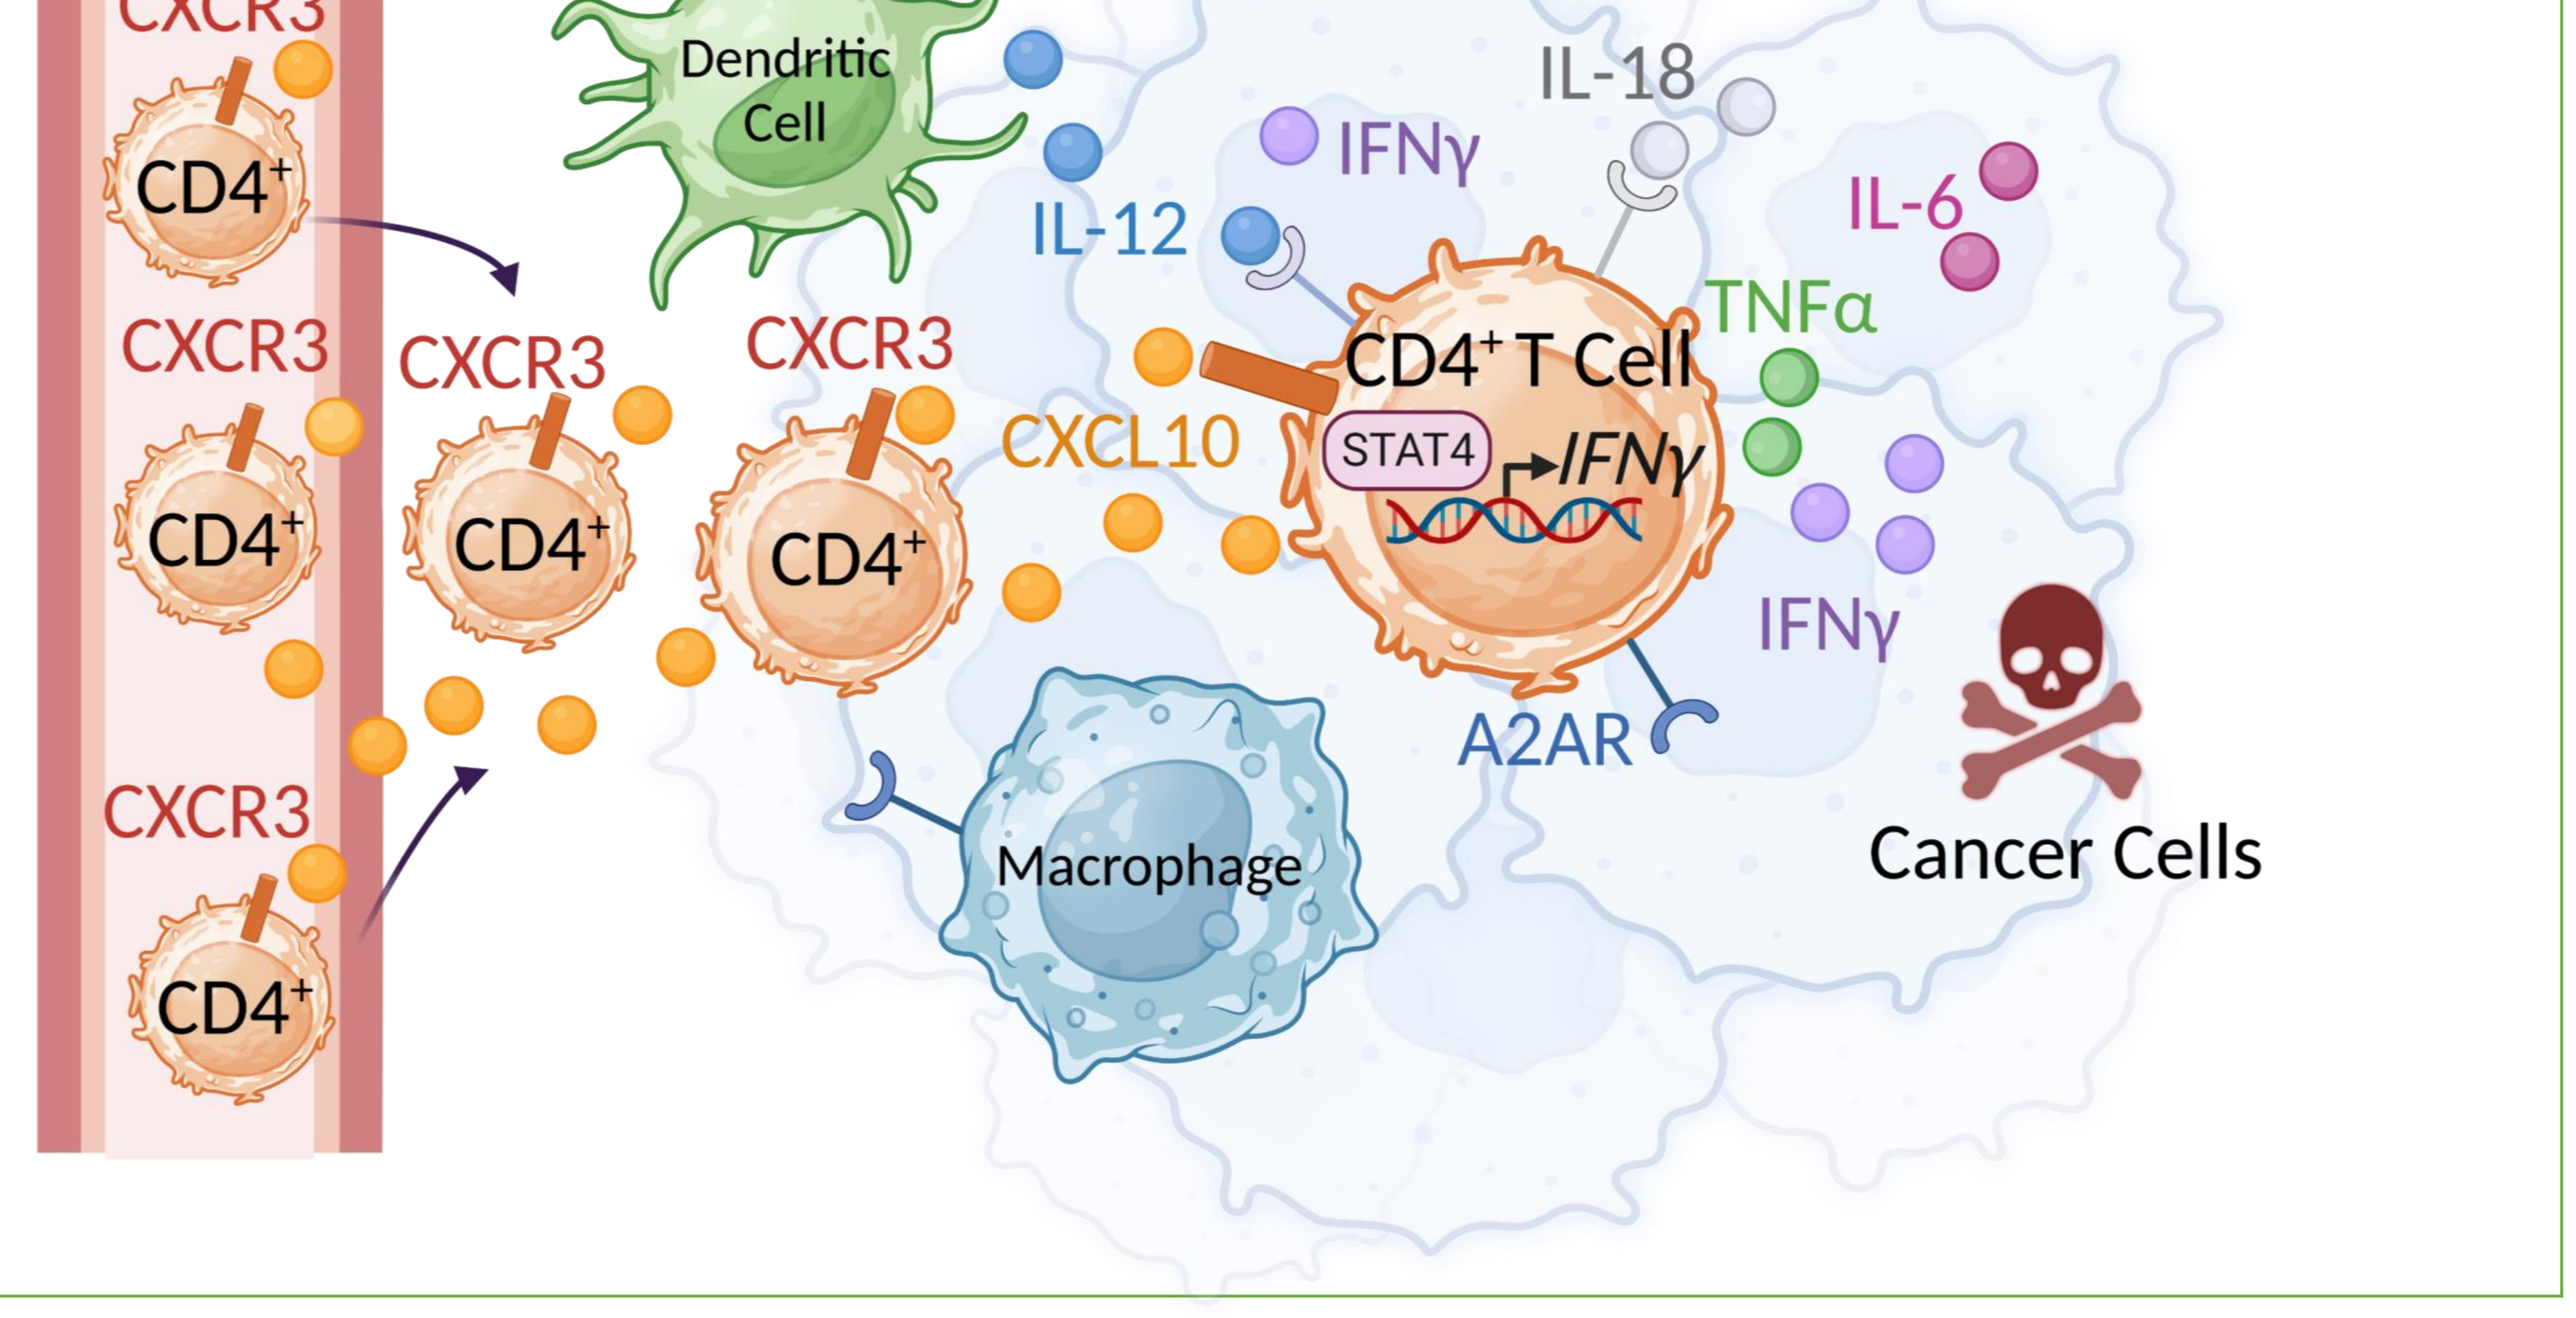
Ciforadenant
CXCR3
CD4+
Dendritic Cell
IL-12
IFNγ
IL-18
IL-6
TNFα
CXCL10
CD4+ T Cell
STAT4
IFNγ
A2AR
Macrophage
Cancer Cells

CONCLUSIONS

- Depletion of myeloid cells abolishes the synergy of Ciforadenant and ICB in the murine melanoma model.
- Combination of Ciforadenant with ICB upregulates the genes involved in IL-12/STAT4 signaling axis which leads to the development of CXCR3⁺ IFNγ-producing Th1 helper cells.
- Ciforadenant treatment increases production of chemokine CXCL10, a ligand for recruitment of CXCR3⁺ Th1 helper cells into the tumor.
- Ciforadenant modulates antitumor responses by turning the tumor microenvironment into the proinflammatory state.
- Combining Ciforadenant with ICB promotes production of proinflammatory cytokines such as IL-6, TNFα, and IFNγ.
- Ongoing Phase II clinical trial together with nivolumab plus ipilimumab (NIVO+IPI) is in collaboration with Kidney Cancer Research Consortium.

CORVUS PHARMACEUTICALS

Disclosure: Dan Li is a full-time employee at Corvus Pharmaceuticals, Inc. and holds stock options of Corvus.